Subgroup Efficacy in Patients Receiving Intralesional Rose Bengal to All Existing Melanoma in Phase II Study PV-10-MM-02

S.S. Agarwala [1], J.F. Thompson [2], B.M. Smithers [3], M.I. Ross [4], C.R. Scoggins [5], B.J. Coventry [6], S.J. Neuhaus [6], D.R. Minor [7], J.M. Singer [8] and E.A. Wachter [8]

[1] St Luke's Hospital and Health Network and Temple University, Bethlehem, PA USA; [2] Melanoma Institute Australia and the University of Sydney, Sydney, NSW AUS; [3] Queensland Melanoma Project, Princess Alexandra Hospital and the University of Queensland, Brisbane, QLD AUS; [4] The University of Texas M. D. Anderson Cancer Center, Houston, TX USA; [5] University of Louisville, Louisville, KY USA; [6] University of Adelaide and Royal Adelaide Hospital, Adelaide, SA AUS; [7] California Pacific Medical Center, San Francisco, CA USA; [8] Provectus Biopharmaceuticals, Inc., Knoxville, TN USA

For additional information: conference@pvct.com

Abstract No. 1120P
39th ESMO Congress
Madrid, Spain 26-30 September 2014

Abstract

BACKGROUND: The safety and efficacy of intralesional (IL) treatment of refractory cutaneous melanoma with rose bengal disodium (PV-10) was evaluated in an 80 patient (pt) international, multicenter, single arm phase II trial (NCT00521053).

METHODS: Patients refractory to a median of 6 previous interventions with 6.3 cm median sum lesion diameter in biopsy-confirmed melanoma received PV-10 into up to 20 cutaneous and subcutaneous lesions up to 4 times over a 16-week period and were followed for 52 weeks. Best overall response rate (BORR) was assessed in up to 10 injected target lesions. Secondary endpoints included assessment of duration of response, BORR of untreated bystander lesions, overall survival and adverse events. Confidence intervals for response were based on the exact cumulative probabilities of the binomial distribution (95% confidence intervals).

RESULTS: Overall, PV-10 was well tolerated and 41 of 80 ITT pts met the primary endpoint of objective response (CR+PR) in their injected target lesions (51% ORR CI 40-63%, 26% CR). In the subgroup of 28 pts who received PV-10 into all existing melanoma lesions, ORR was 71% (CI 51-87%) with 50% CR (CI 31-69%). In these pts plus 26 pts with uninjected disease limited to bystanders (i.e., 54 pts) CR was achieved in 232 of 363 injected lesions (64% CR): 121 lesions required a single injection for CR, 84 required 2 injections, 22 required 3 injections and 5 required 4 injections. Additionally, 10 of 28 uninjected bystander lesions achieved CR.

CONCLUSIONS: Recurrent locoregional melanoma can be a source of persistent morbidity, including disfigurement frequently accompanied with pain, ulceration, bleeding and infection. The high rate of symptom control in refractory patients, manifest in CR of injected lesions after minimal intervention, was the basis for a breakthrough therapy designation application to the US FDA based on the 28 patient "all treated" subgroup, and implications of the Agency's ruling on this application will be presented. Although the primary ablative effect is responsible for CR in injected tumors, durability of response and bystander response implicate an immunologic mechanism of action secondary to ablation.

Background

Primary Ablative Mechanism

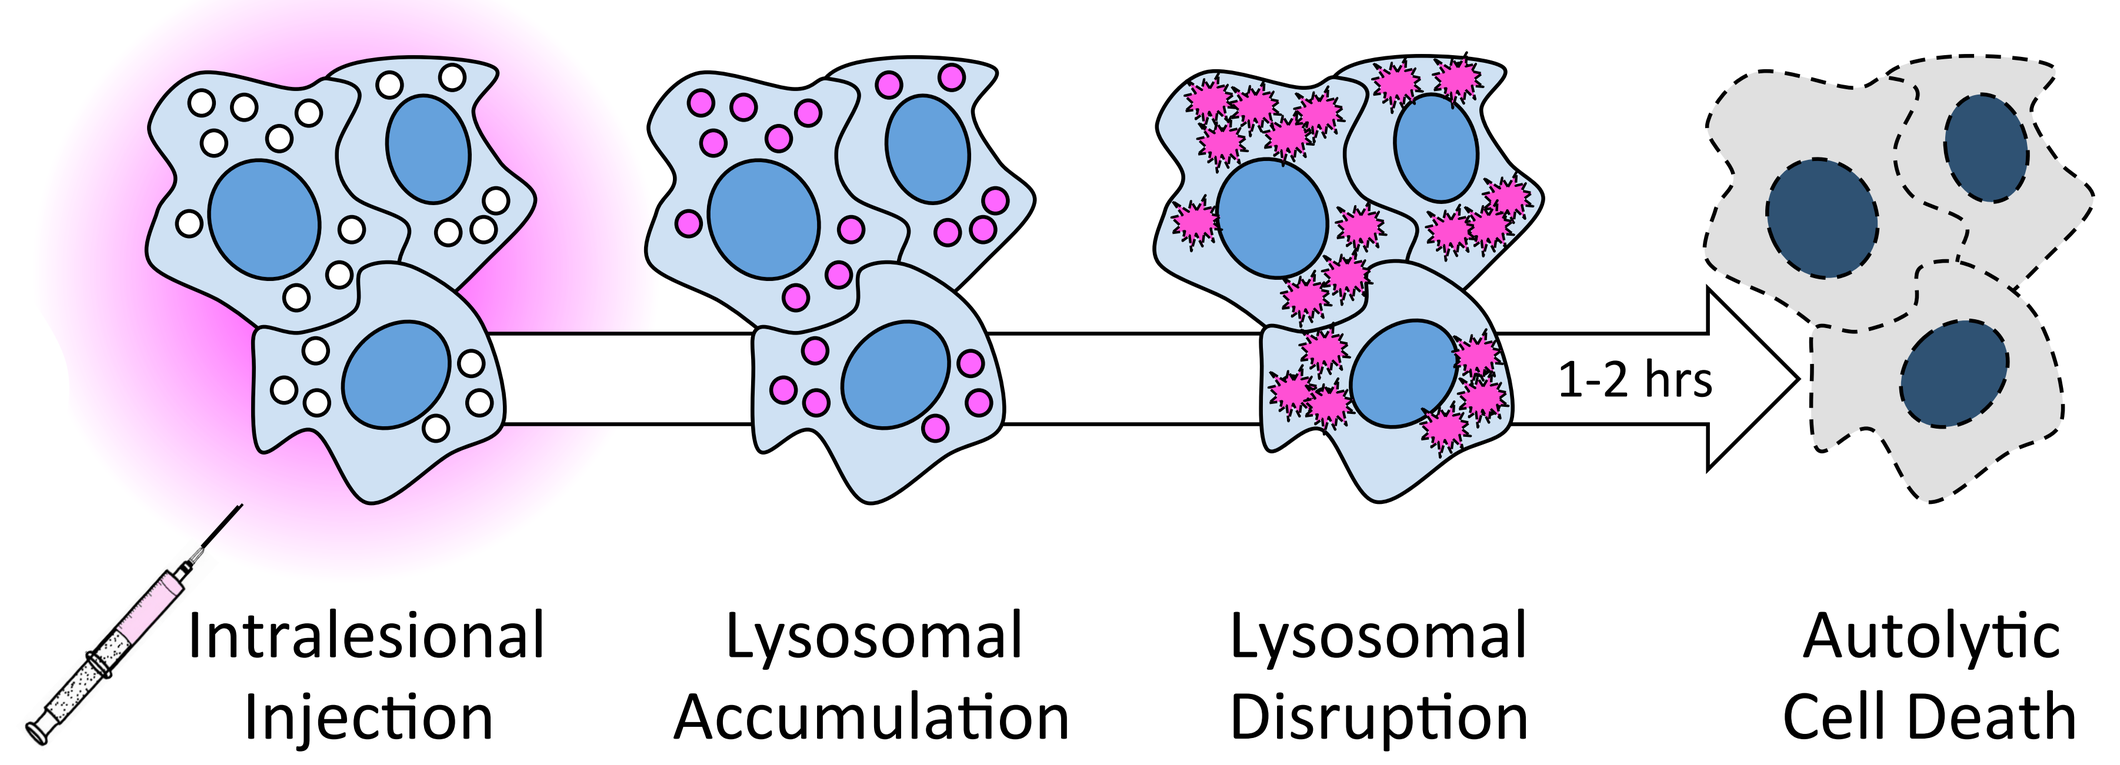
1-2 hrs
Intralesional Injection
Lysosomal Accumulation
Lysosomal Disruption
Autolytic Cell Death

Secondary Immunomodulatory Mechanism

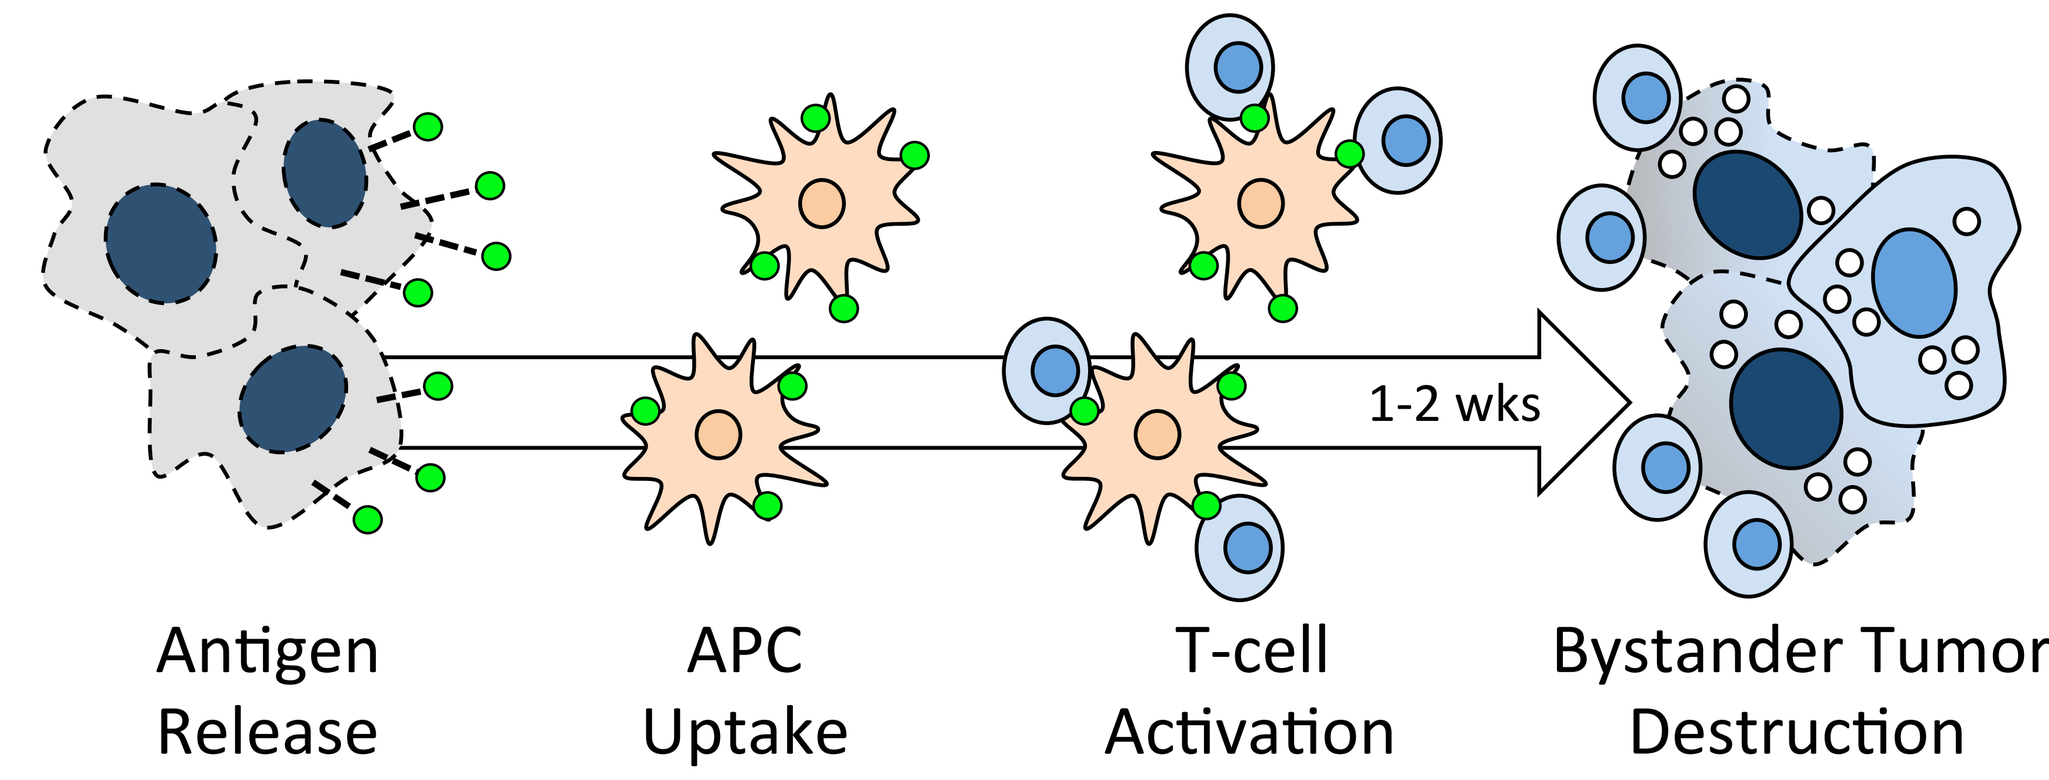
1-2 wks
Antigen Release
APC Uptake
T-cell Activation
Bystander Tumor Destruction

- Wachter et al., SPIE 4630, 147, 2002 (lysosomal accumulation and rupture in tissue culture)
- Thompson et al., Mel Res 18, 405, 2008 (ablation of injected tumors and bystander regression in recurrent patients)
- Toomey et al., PLoS One 8, e68561, 2013 (tumor-specific immune response in mice)
- Liu et al., AACR 2014 Abstract 0630 (DC recruitment/activation and T-cell activation in mice)
- Sarnaik et al., ASCO 2014 Abstract 9028 (pCR and T-cell activation in refractory patients)

Study Design

Up to **10 target** and **10 non-target lesions** were injected with PV-10 up to 4 times during first 16 weeks of study. One or 2 additional lesions could be designated as **bystander lesions** and followed but not injected. Patients followed up to 52 weeks.

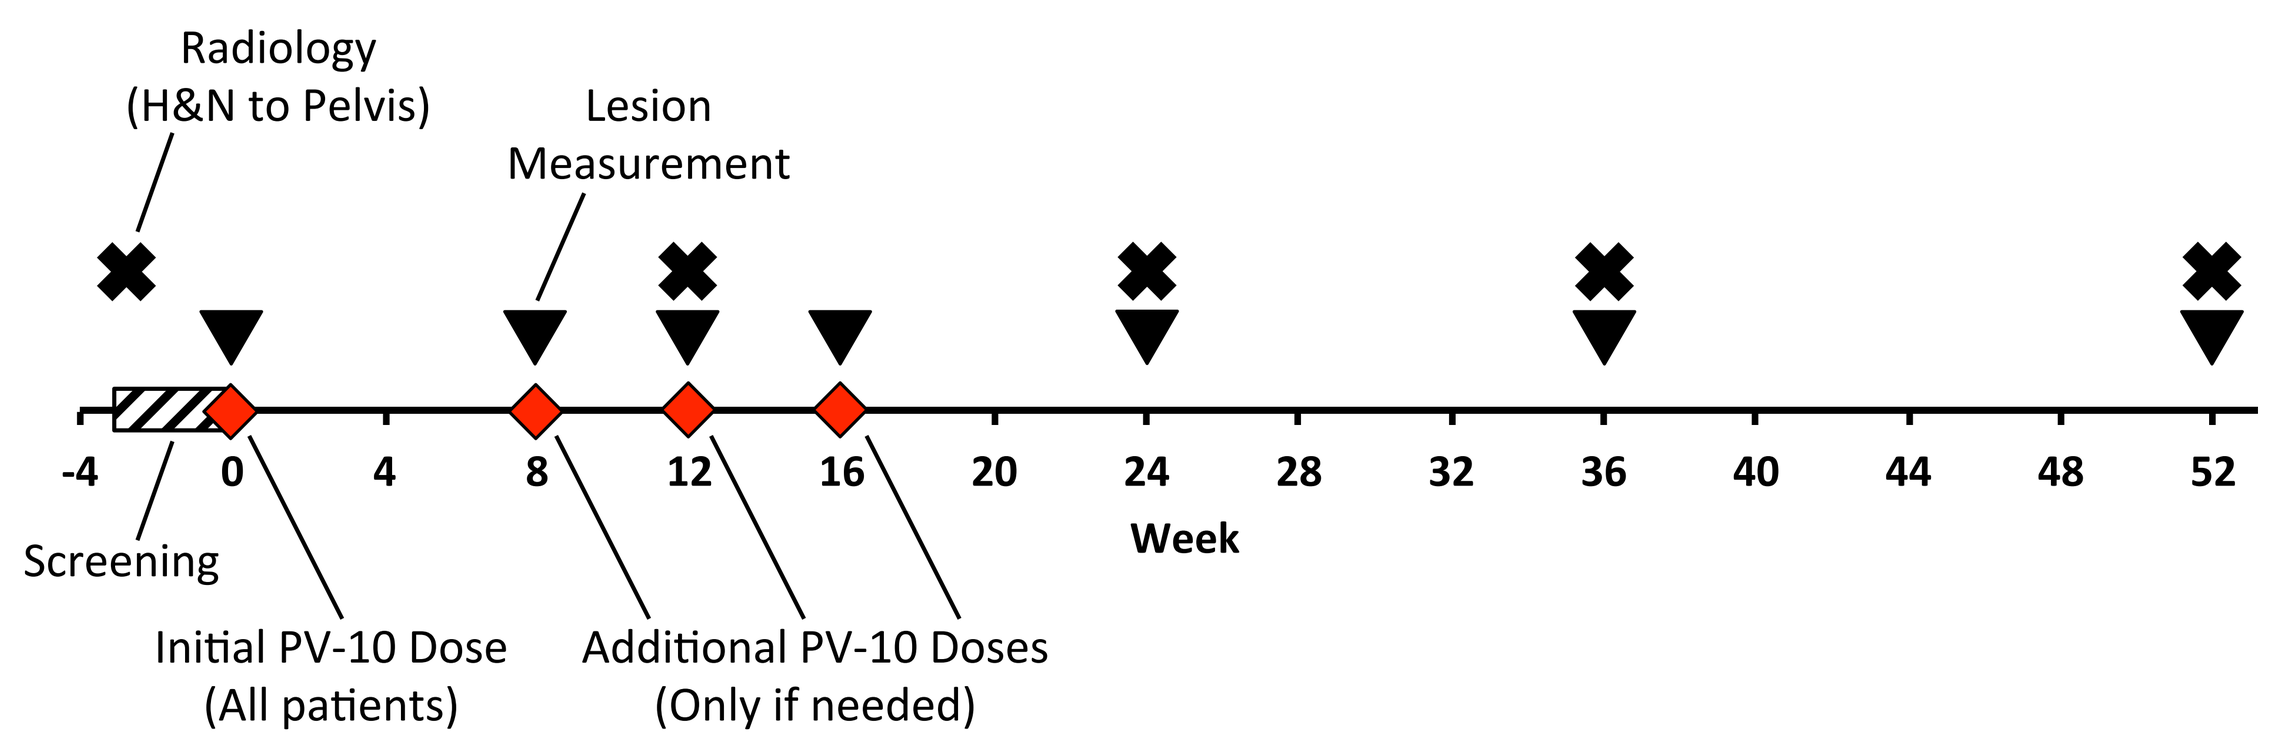
Radiology
(H&N to Pelvis)
Lesion
Measurement
-4
0
4
8
12
16
20
24
28
32
36
40
44
48
52
Week
Screening
Initial PV-10 Dose
(All patients)
Additional PV-10 Doses
(Only if needed)

Response of Patients by Sub-Group

ITT Population
N = 80 Patients
26% CR (CI 17-37%)
51% ORR (CI 40-63%)

Stable Visceral Disease with Cutaneous Lesions
N = 18 Patients

Cutaneous Disease Only
N = 62 Patients

Some Disease Not Followed (TNTC or > 1 Lesion Not Measured)
N = 8 Patients

All Melanoma Followed
N = 54 Patients
37% CR (CI 24-51%)
63% ORR (CI 49-76%)

Bystanders Untreated
N = 26 Patients
23% CR (CI 9-44%)
54% ORR (CI33-73%)

All Lesions Treated
N = 28 Patients
50% CR (CI 31-69%)
71% ORR (CI 51-87%)

- Patients in the All Melanoma Followed sub-group had a median of 8 study lesions (range 1-22) with a median SLD of 4.9 cm (range 0.9-16.1 cm) and had undergone a median of 6 prior interventions (range 1-19).
- Regression of bystander lesions was highly correlated with objective response of target lesions (p = 0.023 by Chi-Square test).
- Progression Free Survival (PFS) calculations consistent with RECIST 1.1.
- Radiologic or other evidence of visceral progression deemed progression.

Objective Response of Target Lesions (ITT Population by Disease Burden)

Response of Target Lesions	Patients Categorized by Disease Burden at Baseline: All Lesions Treated		Bystanders Untreated		Up to 10 Skin Lesions Untreated [1]		TNTC or Stage IV	
N (Patients)	28	%	26	%	7	%	19	%
CR	14	50%	6	23%	1	14%	0	0%
PR	6	21%	8	31%	1	14%	5	26%
SD	3	11%	8	31%	1	14%	2	11%
PD (PD + NEV) [2]	5	18%	4	15%	4	57%	12	63%
[NEV: Progression Prior to Week 8]	[2]	[7%]	[1]	[4%]	[3]	[43%]	[7]	[37%]
CR + PR	20	71%	14	54%	2	29%	5	26%
CR + PR + SD (Locoregional Disease Control)	23	82%	22	85%	3	43%	7	37%
Mean PFS (months) [3]	9.8		8.9		6.0		2.6	

Abbreviations: TNTC, too numerous to count; NEV, non-evaluable. [1] Median number of untreated lesions: 5. [2] Non-evaluable patients were tracked separately but combined with PD for tabulation of outcome. [3] PFS by mRECIST, maximum follow-up duration 12 months.

Response by Patient

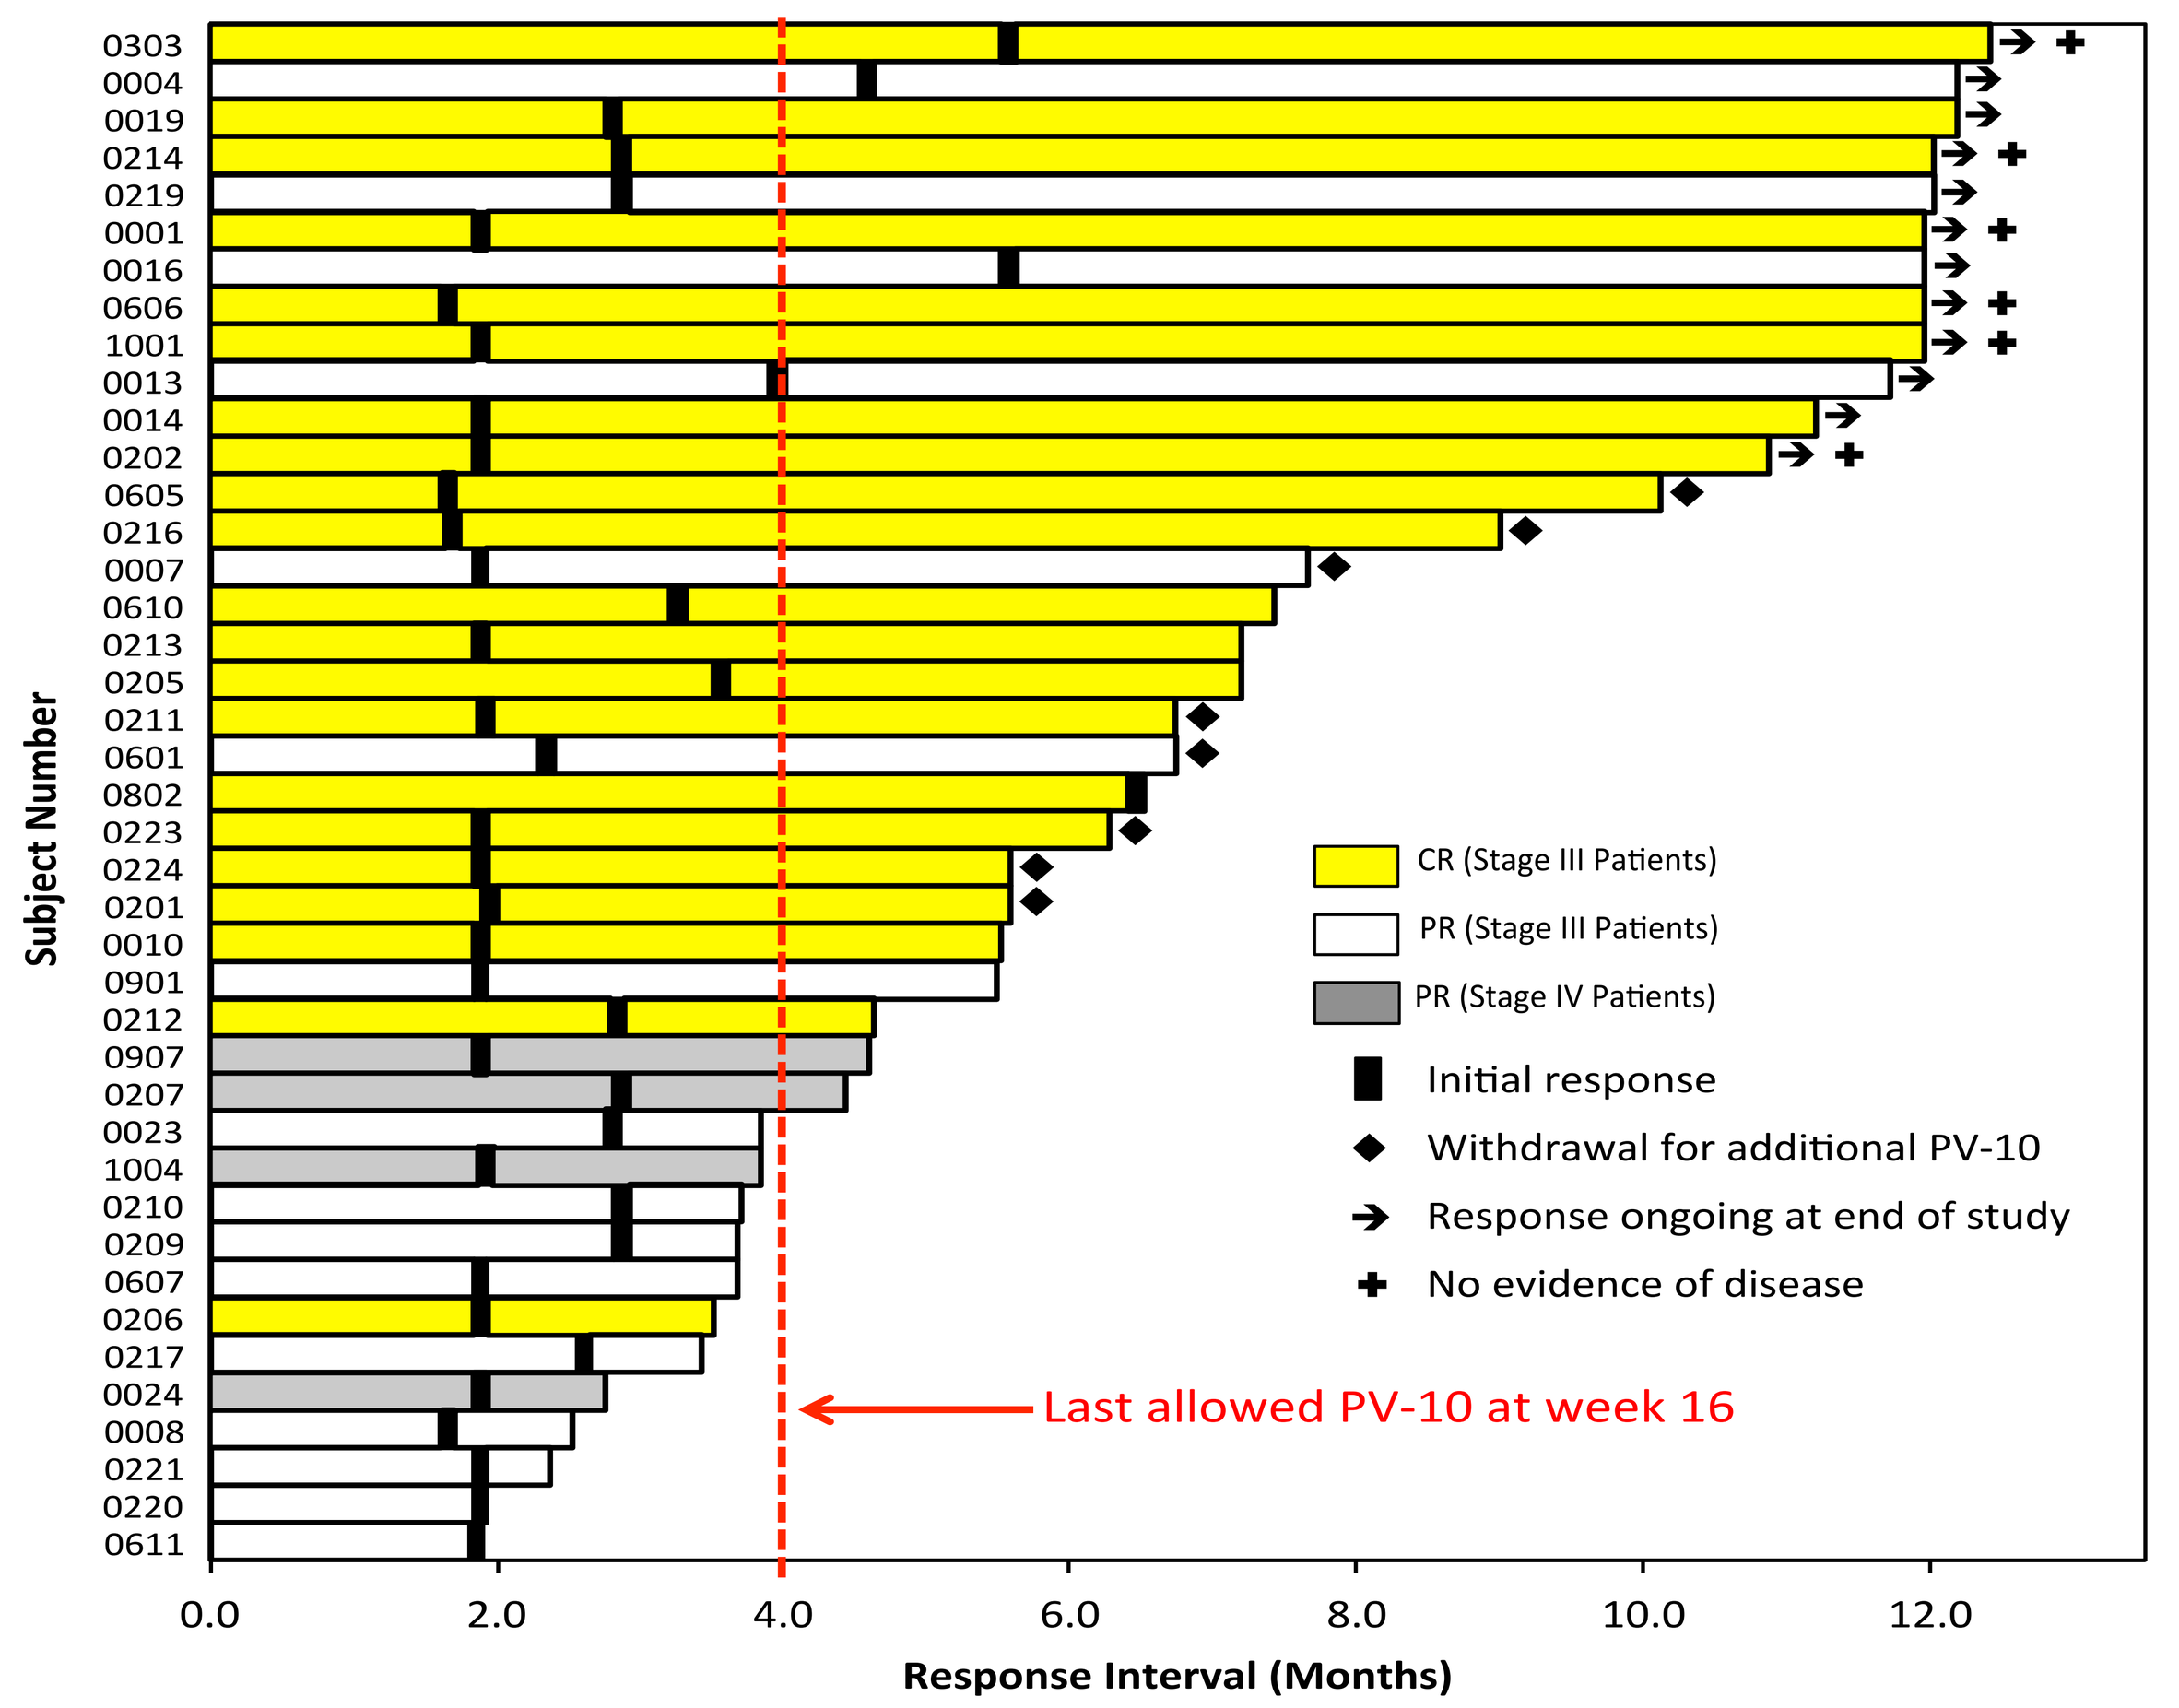
Subject Number
0303
0004
0019
0214
0219
0001
0016
0606
1001
0013
0014
0202
0605
0216
0007
0610
0213
0205
0211
0601
0802
0223
0224
0201
0010
0901
0212
0907
0207
0023
1004
0210
0209
0607
0206
0217
0024
0008
0221
0220
0611
CR (Stage III Patients)
PR (Stage III Patients)
PR (Stage IV Patients)
Initial response
Withdrawal for additional PV-10
Response ongoing at end of study
No evidence of disease
Last allowed PV-10 at week 16
0.0
2.0
4.0
6.0
8.0
10.0
12.0
Response Interval (Months)

Summary of Patients in the All Melanoma Followed Sub-Group

Patient ID	Age	Gender	Stage	Dz Loc	Previous Interventions N	Previous Interventions Sys?	Injected Lesions N	Injected SLD (cm)	Injected BORR	Injected PFS (mons)	Bystander N	Bystander SLD (cm)	Bystander BORR
0201	66	M	IIIB	LE	5	N	2	0.9	CR	5.6	--	--	--
0206	59	M	IIIB	HN	1	N	4	2.6	CR	3.5	--	--	--
0211	82	M	IIIB	LE	5	N	8	4.0	CR	6.8	--	--	--
0212	69	M	IIIC	LE	6	N	6	2.5	CR	4.7	--	--	--
0213	83	F	IIIB	LE	7	N	8	4.8	CR	7.2	--	--	--
0214	57	M	IIIB	HN	3	N	6	5.0	CR	12.1	--	--	--
0216	57	M	IIIC	LE	8	Y	4	2.4	CR	9.0	--	--	--
0223	58	M	IIIC	LE	5	N	20	13.0	CR	6.3	--	--	--
0224	54	M	IIIB	LE	10	N	8	3.0	CR	5.6	--	--	--
0605	55	F	IIIB	LE	6	Y	6	3.8	CR	10.2	--	--	--
0606	79	F	IIIB	LE	4	Y	19	10.3	CR	12.0	--	--	--
0610	63	M	IIIB	LE	3	N	10	2.8	CR	7.5	--	--	--
0802	73	M	IIIB	LE	4	Y	5	3.5	CR	6.4	--	--	--
1001	68	F	IIIB	LE	3	Y	2	2.9	CR	12.0	--	--	--
0217	79	M	IIIB	LE	9	N	15	7.4	PR	3.4	--	--	--
0219	83	F	IIIB	LE	14	N	8	5.1	PR	12.1	--	--	--
0220	61	M	IIIB	LE	5	Y	12	5.8	PR	1.9	--	--	--
0607	58	M	IIIB	T	8	Y	13	6.0	PR	3.7	--	--	--
0611	81	F	IIIB	LE	4	N	20	13.7	PR	1.8	--	--	--
0901	51	F	IIIB	LE	8	Y	6	3.6	PR	5.5	--	--	--
0218	92	M	IIIB	UE	6	N	15	10.7	SD	3.1	--	--	--
0222	70	M	IIIC	LE	5	Y	2	3.3	SD	2.9	--	--	--
1002	50	F	IIIB	LE	6	Y	7	3.6	SD	4.9	--	--	--
0608	92	F	IIIC	LE	7	N	9	3.0	PD	2.0	--	--	--
0903	57	M	IIIC	LE	12	Y	2	1.9	PD	1.9	--	--	--
1003	36	M	IIIB	T	5	Y	1	2.2	PD	1.9	--	--	--
0604	70	F	IIIB	LE	12	Y	15	9.5	NEV	1.0	--	--	--
0609	48	F	IIIC	LE	8	Y	15	5.4	NEV	1.1	--	--	--
0001	78	M	IIIB	LE	9	N	10	7.5	CR	12.0	1	0.4	CR
0010	71	M	IIIB	UE	8	N	4	2.5	CR	5.6	1	0.3	PD
0014	48	M	IIIB	LE	3	N	10	8.0	CR	11.2	1	0.3	CR
0019	82	F	IIIB	LE	10	N	3	1.7	CR	12.2	1	0.7	CR
0202	73	M	IIIB	LE	2	N	11	6.0	CR	10.9	1	0.5	CR
0205	82	F	IIIC	LE	4	Y	4	1.7	CR	7.2	1	0.3	CR
0004	82	M	IIIC	UE	10	N	8	6.6	PR	12.2	1	0.5	SD
0008	72	F	IIIC	LE	5	N	19	15.5	PR	3.5	1	0.5	PD
0013	62	M	IIIB	LE	4	N	6	4.2	PR	11.8	1	0.4	CR
0016	65	M	IIIB	LE	5	N	6	3.3	PR	12.0	1	0.4	CR
0209	62	F	IIIB	LE	2	N	6	3.6	PR	3.7	1	0.4	SD
0210	73	M	IIIC	LE	6	N	7	4.0	PR	3.7	1	0.2	PD
0221	58	M	IIIC	T	4	N	20	15.2	PR	2.4	2	0.6	PD
0601	79	M	IIIC	HN	9	N	13	7.0	PR	6.8	2	1.5	CR
0003	80	F	IIIB	UE	3	Y	2	5.3	SD	3.0	1	1.4	PD
0009	59	M	IIIB	LE	2	N	7	12.2	SD	1.9	1	0.3	PD
0011	89	F	IIIC	LE	4	N	12	9.1	SD	2.8	1	0.7	PD
0020	64	M	IIIB	LE	10	N	4	7.3	SD	3.7	1	0.8	SD
0025	57	F	IIIB	LE	9	N	4	3.1	SD	5.6	1	0.6	SD
0204	82	F	IIIB	LE	4	N	8	3.0	SD	1.9	1	0.3	PD
0302	53	M	IIIC	LE	19	Y	2	4.3	SD	1.9	1	3.6	PD
0304	69	F	IIIC	LE	8	Y	8	7.1	SD	3.9	1	0.7	PR
0602	43	F	IIIC	LE	9	Y	9	3.9	PD	2.0	1	0.5	PD
0801	57	M	IIIB	LE	11	Y	10	8.4	PD	1.9	1	0.6	PD
0803	57	F	IIIC	LE	12	Y	20	6.7	PD	1.7	1	0.3	PD
0021	87	F	IIIC	LE	3	N	9	7.8	NEV	0.5	1	0.3	NEV

Dz Loc (Disease Location): LE, lower extremity; UE, upper extremity; HN, head/neck; T, trunk. Sys? (systemic intervention), including regional chemotherapy. Injected lesions: N, number lesions injected; SLD, sum lesion diameter. Bystander lesions: N, number uninjected bystander lesions. NEV, withdrew before first formal response assessment, tabulated as PD.

Response by Lesion

A majority of lesions in patients in the All Melanoma Followed sub-group achieved a complete response after 1 or 2 injections with PV-10 as illustrated below.

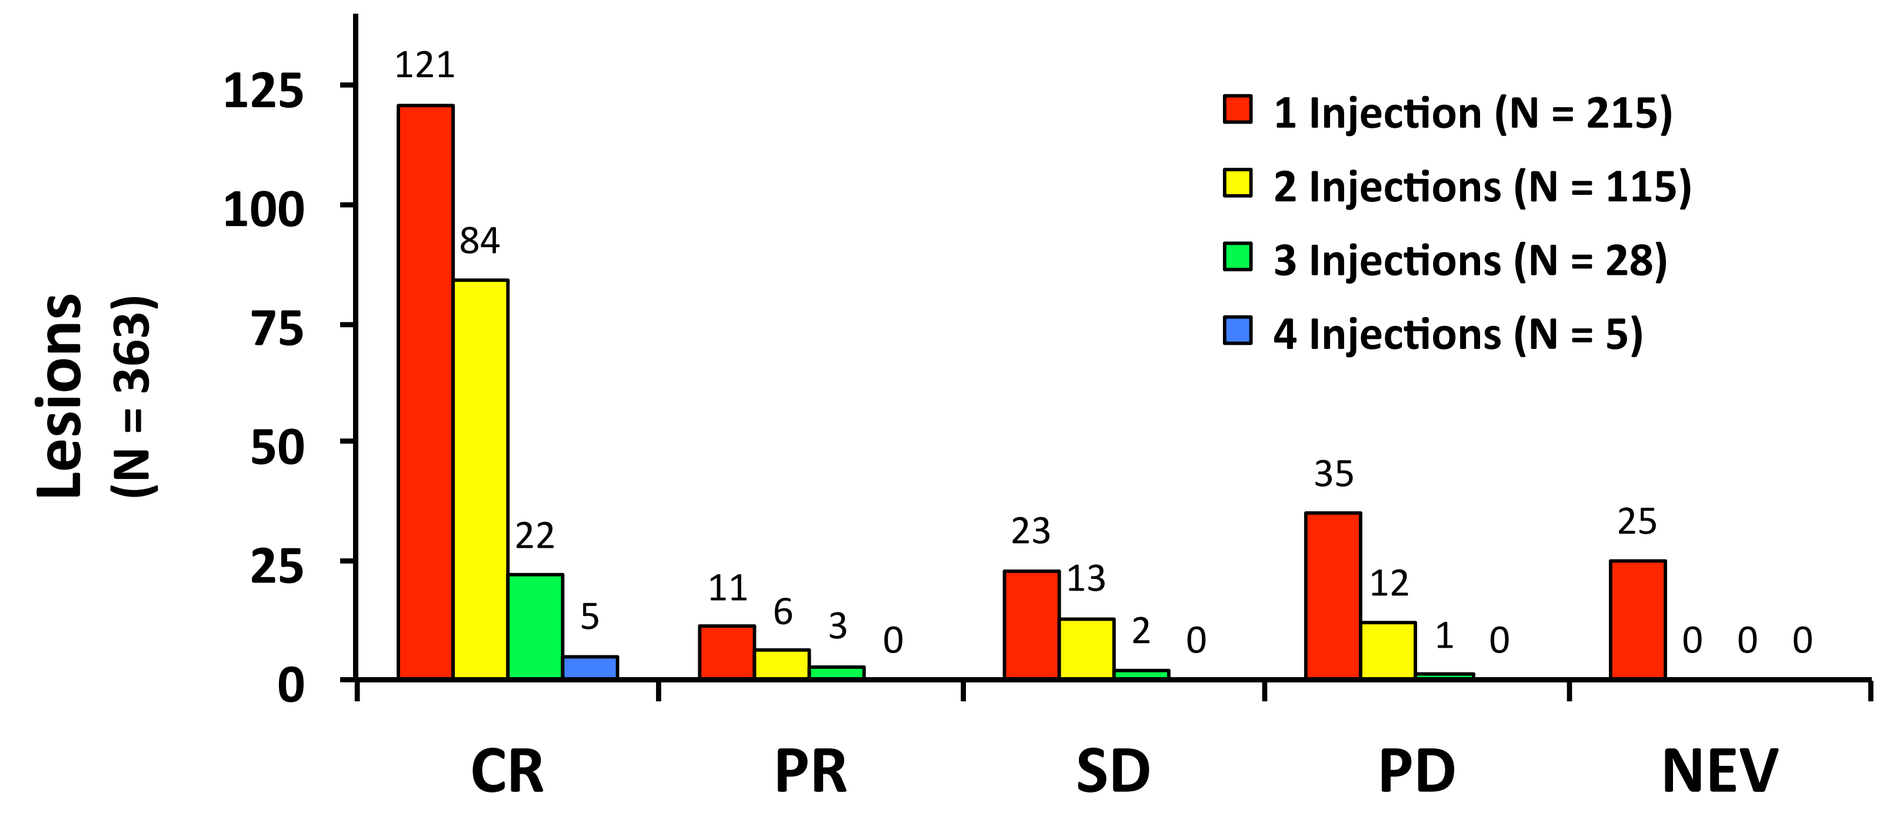
Lesions
(N = 363)
1 Injection (N = 215)
2 Injections (N = 115)
3 Injections (N = 28)
4 Injections (N = 5)
121
84
22
5
11
6
3
0
23
13
2
0
35
12
1
0
25
0
0
0
CR
PR
SD
PD
NEV

Objective Response of Target Lesions (By Disease Burden)	All Lesions Treated		Bystanders Untreated		All Melanoma Followed	
N (Lesions)	181		182		363	
CR	134	74%	98	54%	232	64%
PR	11	6%	9	5%	20	6%
SD	8	4%	30	16%	38	10%
PD + non-evaluable lesions	28	15%	45	25%	73	20%
ORR (CR + PR)	145	80%	107	59%	252	69%

Patient Reported Outcome

There were no requirements for lesion pain, bleeding or ulceration for enrollment, but 28 of 80 ITT patients reported mild or greater lesion pain (defined as a score of 5 mm or greater on a 100 mm Visual Analog Scale) at baseline. Of these, 26 had follow-up data at or after week 4 allowing change in symptomatic status to be tracked.

- Median lesion pain scores for patients reporting pain at baseline analyzed by sub-group.
- Patients in the All Melanoma Followed cohort exhibited substantial improvement after initial PV-10 treatment that coincided with reduction in tumor burden.
- Lesion pain was the most frequently reported AE immediately following PV-10 injection.
- Concomitant pain medication use not evaluated.

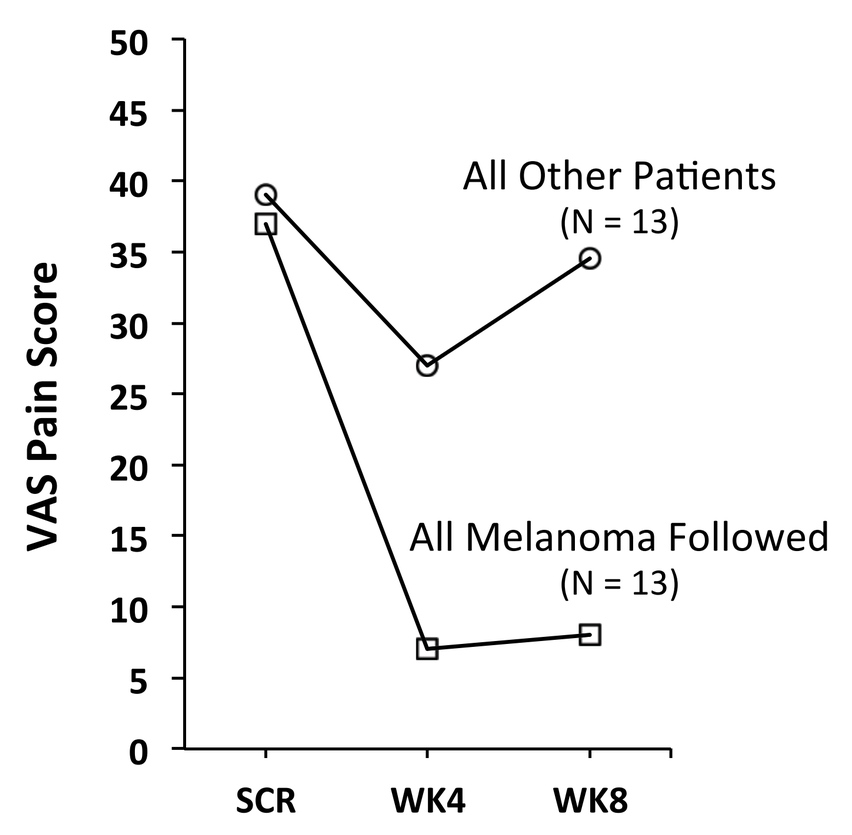
VAS Pain Score
All Other Patients
(N = 13)
All Melanoma Followed
(N = 13)
SCR
WK4
WK8

Example Clinical Response

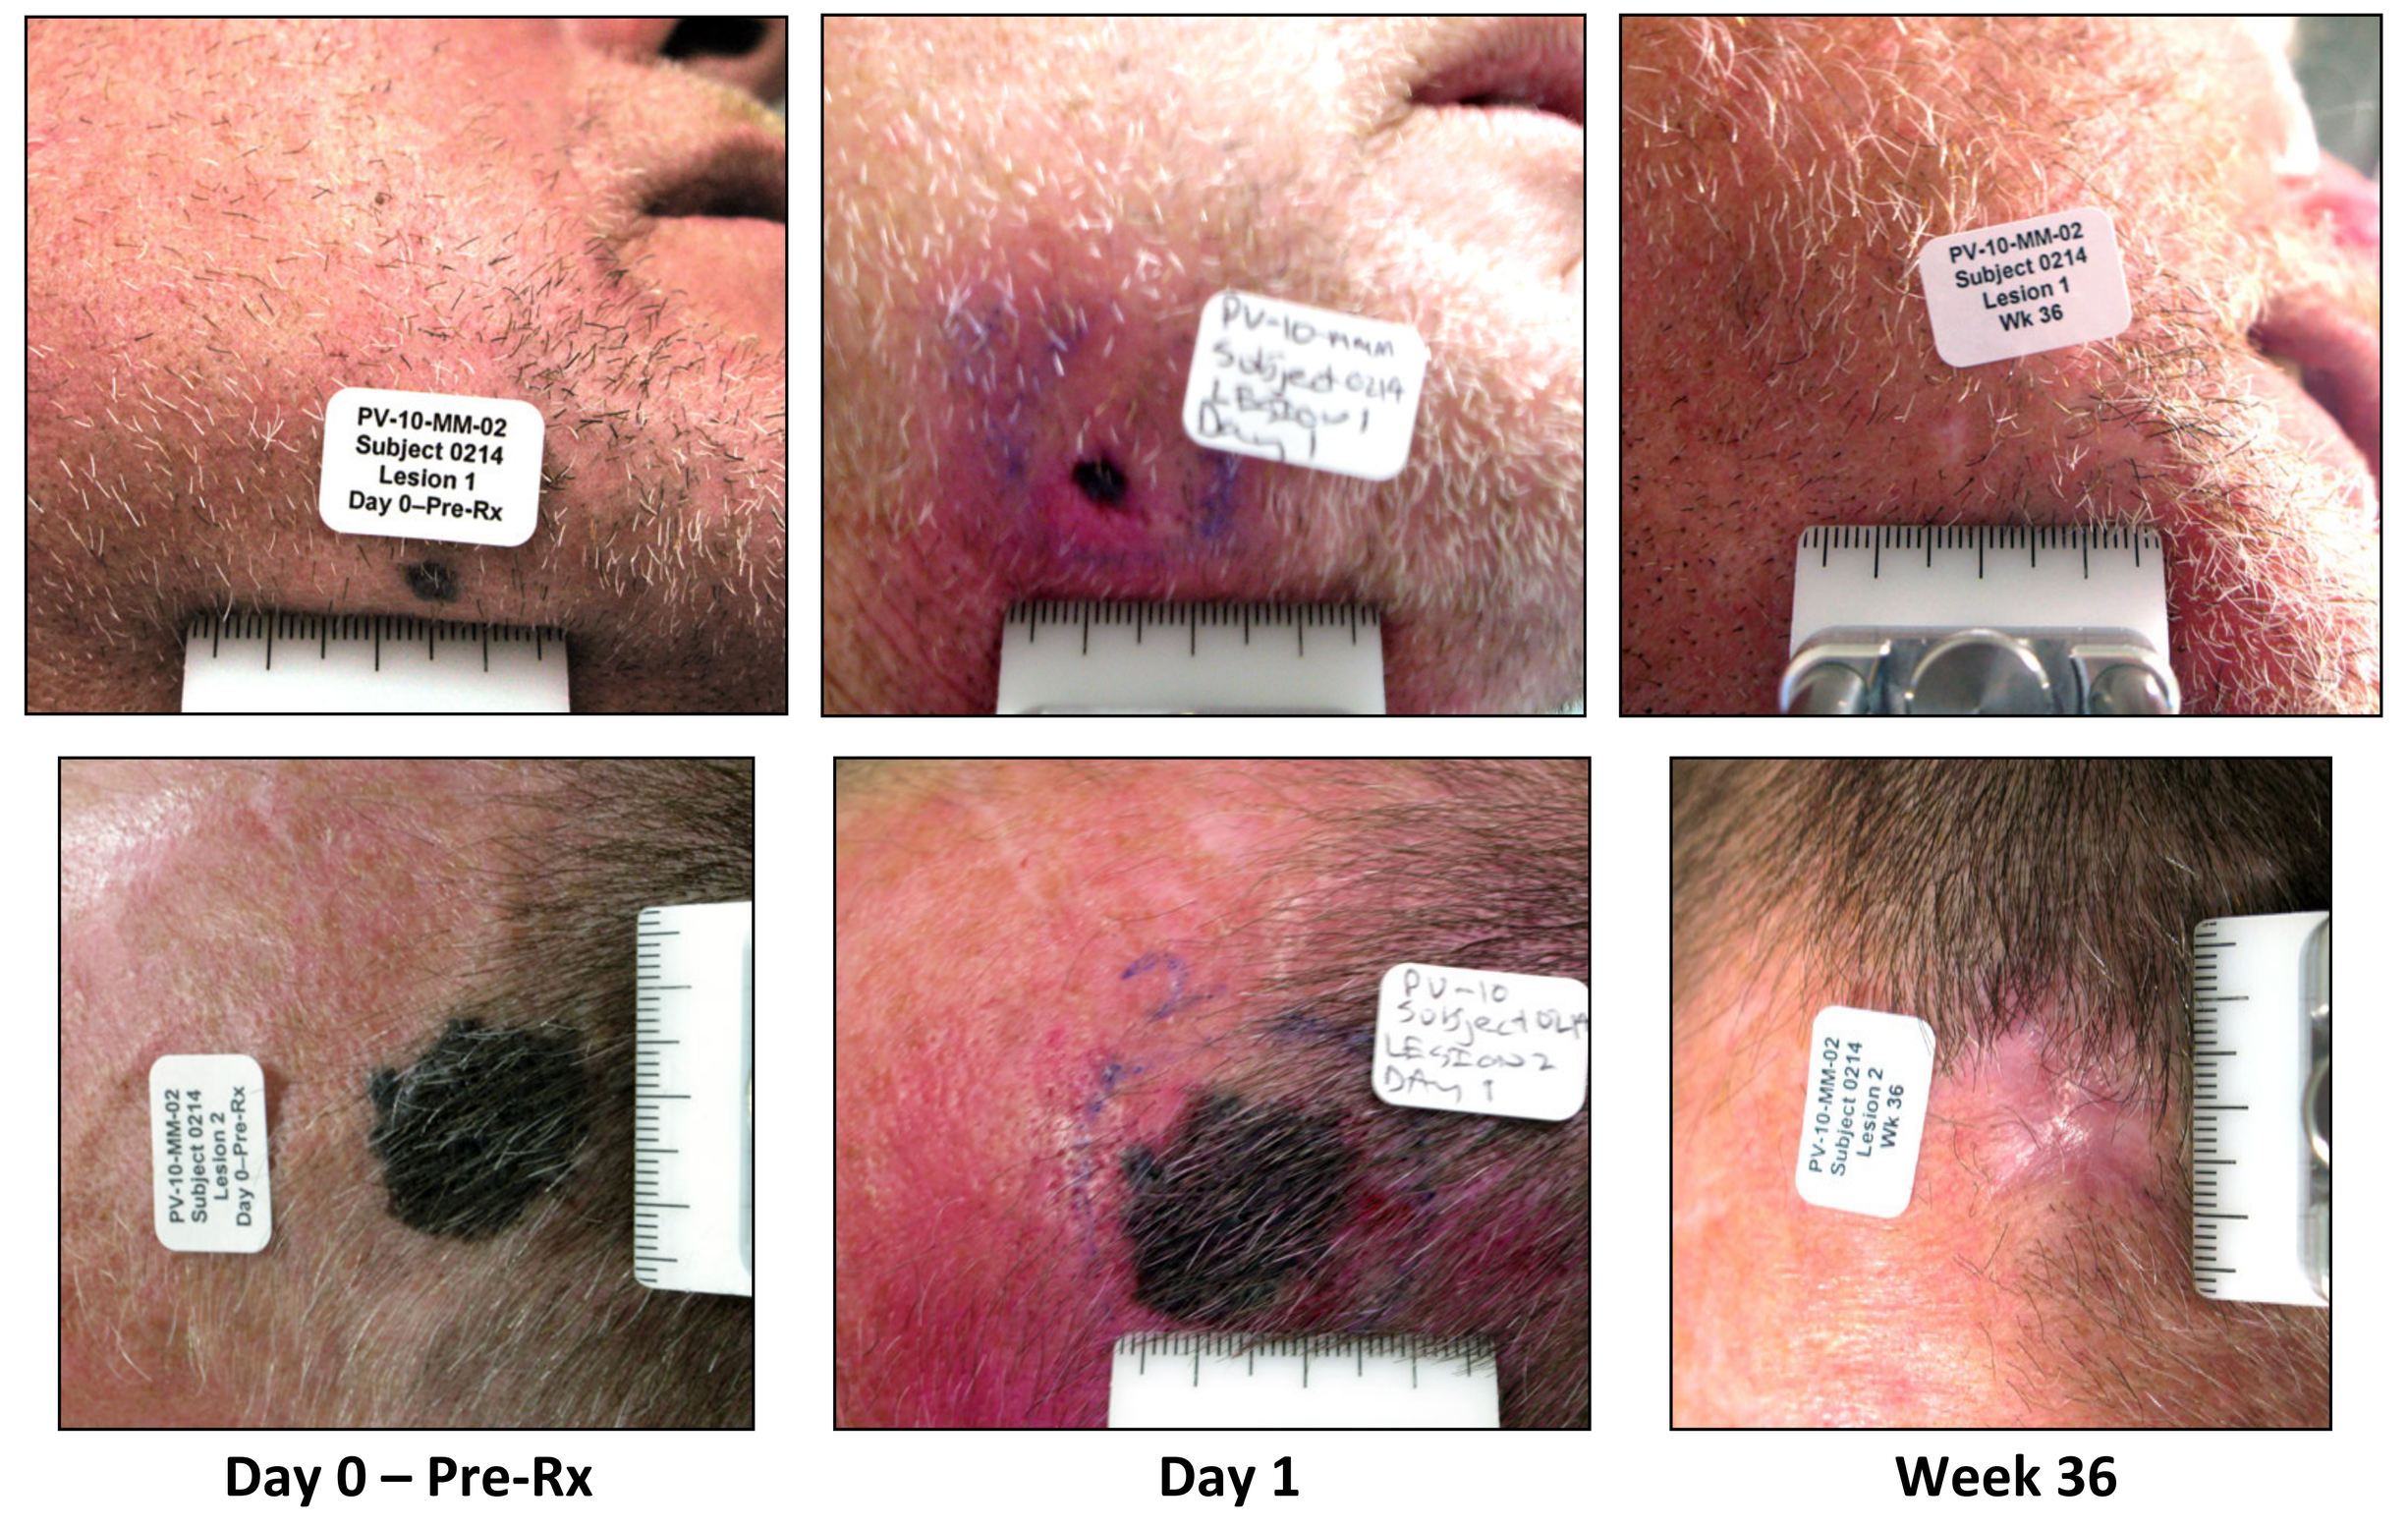
PV-10-MM-02
Subject 0214
Lesion 1
Day 0–Pre-Rx
PV-10-MM-02
Subject 0214
Lesion 1
Wk 36
PV-10-MM-02
Subject 0214
Lesion 2
Wk 36

Day 0 – Pre-Rx | Day 1 | Week 36

Patient 0214, male age 57, Stage IIIB melanoma of the scalp recurrent after 3 surgical interventions. Injection of 3.2 mL PV-10 into all lesions (6 lesions) at day 0, 4.5 mL into 3 lesions at week 8 and 0.3 mL PV-10 into 3 lesions at week 16. Durable CR from Week 24 with NED at end of study (Week 52).

Adverse Events

Most Frequent Adverse Events at Least Possibly Related to PV-10 Treatment

System Organ Class [1] / Preferred Term [1]	Adverse Events [2] (ITT Population, N = 80) CTCAE Grade 1	2	3	Total	%
General Disorders and Administration Site Conditions					
Injection Site Pain	29	25	10	64	80%
Injection Site Edema	19	14	0	33	41%
Injection Site Vesicles	17	13	1	31	39%
Injection Site Discoloration	13	12	0	25	31%
Injection Site Swelling	14	7	1	22	28%
Injection Site Pruritus	14	3	0	17	21%
Injection Site Erythema	6	4	1	11	14%
Injection Site Infection	3	2	1	6	8%
Peripheral Edema	3	0	1	4	5%
Injection Site Cellulitis	0	2	1	3	4%
Gastrointestinal Disorders					
Dysphagia	0	0	1	1	1%
Nervous System Disorders					
Headache	11	2	0	13	16%
Skin and Subcutaneous Tissue Disorders					
Photosensitivity Reaction	0	0	1	1	1%

[1] System Organ Class and Preferred Term based on MedDRA® version 13.0 dictionary. [2] Includes all Grade 3 AEs; no Grade 4 or 5 AEs were reported.

Assessing Symptoms: Path Forward

Measuring skin symptoms of locally advanced cutaneous melanoma is an important parameter in evaluating patient benefit from a therapy. Provectus is working with ERT, Inc., to implement a patient-reported outcome (PRO) instrument to capture changes in symptoms over time. Per FDA Guidance on PROs, this includes:

- Conducting interviews with patients and experts and reviewing the literature to identify the key symptoms important in this population
- Examining widely-used measures to identify the PRO instrument best suited to capturing these symptoms in this patient population

Conclusions

Phase 3 RCT of PV-10 vs Systemic Therapy in Locally Advanced Cutaneous Melanoma

- Response data from the **All Melanoma Followed Sub-group** enabled study design by:
 - Facilitating dialog with regulatory authorities on patient population and endpoints
 - Suggesting a substantial PFS advantage for PV-10 over historical data on comparator
 - Enabling optimization of PV-10 dose schedule
- Key study design elements
 - Patients failed or ineligible for systemic immunotherapy and are BRAF wild-type
 - Disease limited to cutaneous and subcutaneous sites
 - All disease to be treated until complete response or disease progression
 - Four week treatment cycles (PV-10 or DTIC/TMZ) during initial 12 week treatment phase
 - Comprehensive disease assessment starting at week 12 then every 12 weeks for up to 18 months, then every 6 months until disease progression, death or study closure
 - Many design elements adapted from recent pivotal trials in oncology
- Endpoints
 - Progression-Free Survival (PFS) by RECIST 1.1
 - Complete Response Rate (CRR) by RECIST 1.1
 - Change in Patient Reported Outcomes (to assess impact on lesion symptoms)
 - Overall Survival
- Sample size estimated at 219 patients (2 : 1 randomization, $\alpha = 0.05$ (2-sided), 90% power)
- Start Q4-2014, accrual ca. 18 months with 12 months follow-up of last patient enrolled
- Interim assessment by an independent Clinical Trial Data Monitoring Committee when 50% of the required events (i.e., disease progressions) have occurred

PV-10 with Immune Checkpoint Blockade for Widely Metastatic Disease

- Emerging immunology data supports T-cell mediated tumor-specific immune response secondary to PV-10 ablation
- Data to be presented in November on IL PV-10 in combination with immune checkpoint blockade in melanoma models may indicate clinical studies are warranted [1]
- Design of a Phase 1b/2 combination study is underway

[1] Pilon-Thomas et al., Efficacy of Intralesional Injection with PV-10 in Combination with Co-Inhibitory Blockade in a Murine Model of Melanoma, SITC 2014.