Chemoablation of Metastatic Melanoma with Rose Bengal (PV-10)

S. S. Agarwala [1], J.F. Thompson [2], B.M. Smithers [3], M.I. Ross [4], B.J. Coventry [5], D.R. Minor [6], C.R. Scoggins [7] and E.A. Wachter [8]

[1] St Luke's Hospital and Health Network, Bethlehem, PA USA; [2] Melanoma Institute Australia, Sydney, NSW AUS; [3] Princess Alexandra Hospital, Woolloongabba, QLD AUS; [4] M. D. Anderson Cancer Center, Houston, TX USA; [5] Royal Adelaide Hospital, Adelaide, SA AUS; [6] California Pacific Medical Center, San Francisco, CA USA; [7] University of Louisville, Louisville, KY USA; [8] Provectus Pharmaceuticals, Inc., Knoxville, TN USA

Abstract No. 8534
ASCO Annual '10 Meeting
4 – 8 June 2010

For additional information:
Eric Wachter
wachter@pvct.com

Abstract

Background: Intralesional rose bengal (PV-10, 10% RB in saline) can elicit selective chemoablation of solid tumors and an apparent bystander response in untreated lesions. In phase I (20 subjects with AJCC stage III-IV melanoma), a single injection into a total of 114 lesions was well tolerated, yielding durable objective response (OR) at 12-24 weeks in 40% of subjects (20% CR + 20% PR by modified RECIST) and locoregional disease control (CR+PR+SD) in 75% of subjects; 15% of subjects achieved an OR in their bystander lesions (43 lesions).

Methods: Phase II testing (80 subjects with measurable Stage III-IV melanoma) commenced at 7 centers in Australia and the USA in October 2007 with enrollment completed in May 2009. After initial treatment of 1-20 cutaneous, subcutaneous or nodal lesions, new or incompletely responsive lesions could be retreated at weeks 8, 12 or 16, with follow-up to 52 weeks. Another 1-2 lesions, including designated visceral lesions, could remain untreated for assessment of bystander response. The primary end-point was OR of injected target lesions; secondary endpoints included OR of bystander lesions, PFS of target lesions, and plasma PK assessment.

Results: The first 40 subjects (median age 74.5, range 37-92) completing the study received PV-10 into 486 lesions (mean 1.9 treatment sessions per subject). Adverse events were predominantly mild to moderate, locoregional and transient, the most common being pain at the treatment site (82% of subjects), vesicles or edema (50% each) followed by swelling, pruritus, skin discoloration or headache (18% each), with no grade 4 or 5 AEs attributed to PV-10. Among all 40 subjects, 33% achieved CR, 28% PR and 20% SD of their target lesions; 33% of 21 subjects with evaluable bystander lesions achieved CR in these lesions, along with 10% PR and 14% SD. Mean PFS was 8.5 months (all subjects), while the OR cohort had significantly longer PFS (11.1 months) than SD or PD subjects (2.8 and 2.7 months, respectively). Plasma clearance was bi-exponential ($t_{1/2}$ of 6.3 hrs and 4.3 days) consistent with rapid uptake of extravasate and prolonged intralesional retention.

Conclusions: Overall, the safety and efficacy profile of PV-10 compares favorably with available and emerging options for this patient population.

Background

PV-10 is a sterile, non-pyrogenic solution of Rose Bengal disodium (10% RB)

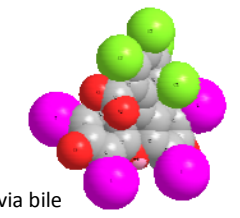

- ***RB is a Fluorescein derivative attributed to Gnehm in 1882***
- ***Prior Human Use of RB***
 - IV hepatic diagnostic, ^{131}I radiolabeled RB: Robengatope®
 - Topical ophthalmic diagnostic: Rosettes® and Minims®
- ***Established Safety History***
 - Not metabolized, short circulatory half-life (ca 30 min), excretion via bile
- ***In Non-Clinical Testing PV-10 Targeted Neoplastic Tissue***
 - Murine xenograft/ homograft and spontaneous tumors in companion animals
 - Prolonged retention in tumors
 - Lysosomal accumulation within cancer cells leading to acute necrosis
 - Minimal toxicity in normal tissue
 - Selective chemoablation of injected tumors
- ***PV-10 May Elicit Bystander Effect in Non-Injected Tumors***
 - Ablation reduces tumor burden and exposes tumor antigens to host
 - Apparent immune-mediated response
 - Possible systemic benefit
- ***Phase 1 Clinical Testing***
 - Single injection into 1–20 lesions in 20 subjects with AJCC Stage III/IV melanoma
 - Intralesional dosing at 50% of calculated lesion volume
 - 1–3 additional lesions untreated to assess bystander response
 - 12–24 weeks observation
 - ORR assessed by modified RECIST
 - AEs generally mild to moderate grade – predominantly locoregional
 - Preliminary efficacy
 - Injected lesions: ORR = 40% (locoregional disease control in 75% of subjects)
 - Bystander lesions: ORR = 15% (locoregional disease control in 55% of subjects)

Phase 2 Study Overview

Protocol PV-10-MM-02

- ***Study Design***
 - Open label, single-arm trial
 - 80 subjects with AJCC Stage III/IV melanoma
 - **Treatment of 1–10 Target Lesions and up to 10 Non-Target Lesions**
 - Target Lesions must be $\geq$ 0.2 cm diameter
 - Biopsy confirmation of at least one Target Lesion
 - Single intralesional dosing at 50% of calculated lesion volume
 - **Observe up to 1–2 untreated Bystander Lesions**
 - Typically small or difficult to access
 - Biopsy confirmation of each Bystander Lesion
 - **Retreatment (new or partially-responsive lesions) allowed at weeks 8, 12 or 16**
 - **Observe for 52 weeks**
- ***Outcome Assessment***
 - Modified RECIST assessed on Target and Bystander Lesions
 - Progression Free Survival
 - Modified Fleming design
 - Interim Safety Assessment 28 days after 20th and 40th subject treated
 - Interim Efficacy Assessment 24 weeks after 20th and 40th subject treated
- ***Study Status: Enrollment commenced Aug 2007, completed May 2009***
 - N = 40 Interim Safety and Efficacy Assessments complete
 - Final Safety and Efficacy Assessment (N=80) ongoing

Phase 2 Study Population

N = 40 Interim Safety and Efficacy Cohort

- AJCC Stage IIIB (20 subjects), IIIC (9), IV-M1a (1), IV-M1b (8) and IV-M1c (2)
- 26 males / 14 females (all subjects white)
- Median age 74.5 yrs (range 37–92 yrs)
- Disease history: median 33.6 months from 1⁰ diagnosis to study enrollment
- Treatment history: Sx (40 subjects), Nodal biopsy (24), ILI/ILP (7), XRT (7), Amputation (3), IFN (2), Vaccine (2), Systemic chemo (1), Investigational agents (1)
- Study lesions: extremity (27 subjects lower and 4 upper extremity), torso (6), H&N (5)
- Lesions treated per subject: median 8 (range 1–20)
- Number of treatment cycles per subject: median 2 (range 1–3)
- PV-10 dose per treatment: mean 2.5 mL, median 1.4 mL (range 0.2–15.0 mL)
- Cumulative PV-10 dose per subject: mean 4.6 mL, median 2.9 mL (range 0.4–25.6 mL)

Example Clinical Response

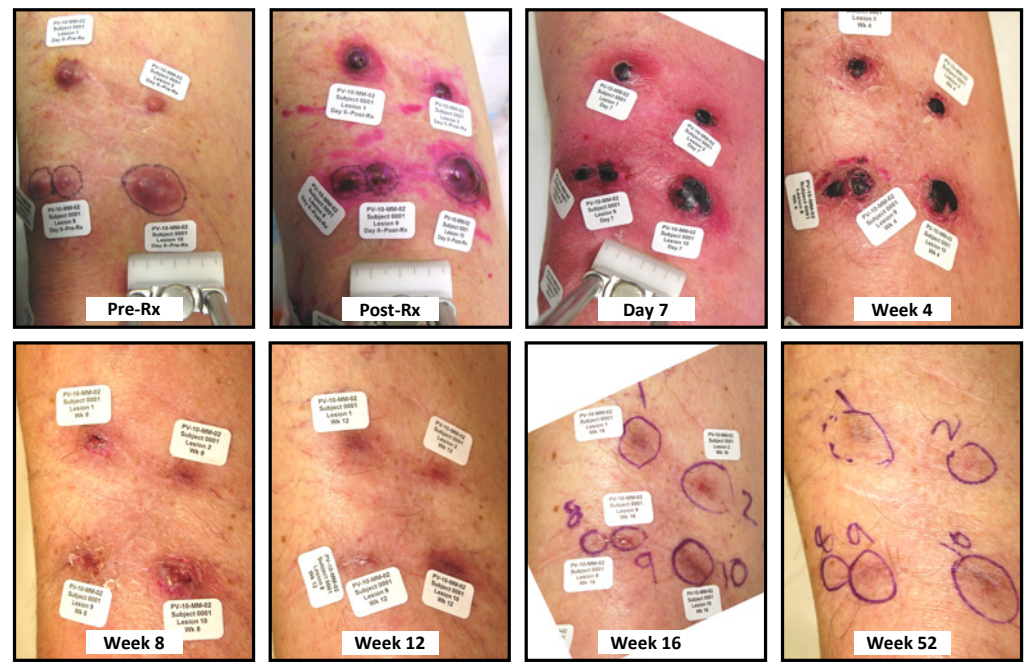

Pre-Rx
Post-Rx
Day 7
Week 4
Week 8
Week 12
Week 16
Week 52

Subject 0001: Male, age 78, Stage IIIB (N2c), onset 1992, multiple Sx of mets since 2005.
Single treatment with 2.0 mL PV-10 to 10 lesions; 1 untreated bystander lesion.

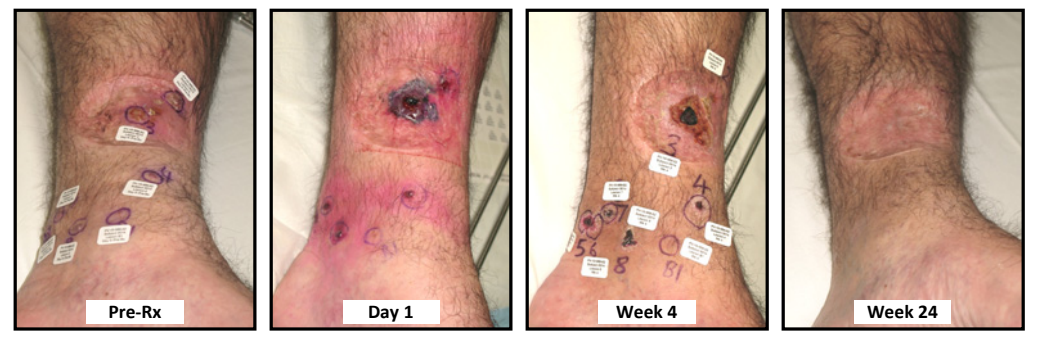

Pre-Rx
Day 1
Week 4
Week 24

Subject 0014: Male, 48, Stage IIIB (N2c) since 2008, Sx of 1⁰ and mets.
Single treatment with 1.3 mL PV-10 to 10 lesions; 1 untreated bystander lesion.

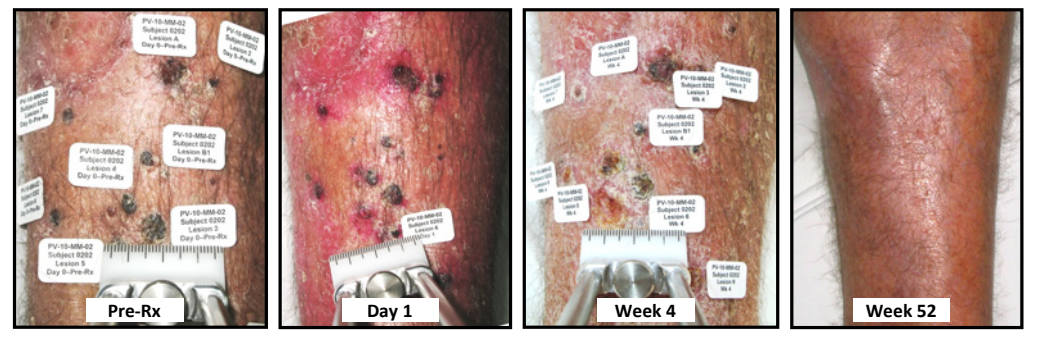

Pre-Rx
Day 1
Week 4
Week 52

Subject 0202: Male, 73, Stage IIIB (N2c) since 2008, Sx of 1⁰ and mets.
Three treatments (Day 0, Wk 8 and Wk 16) with PV-10 to 11 lesions; 1 untreated bystander lesion.

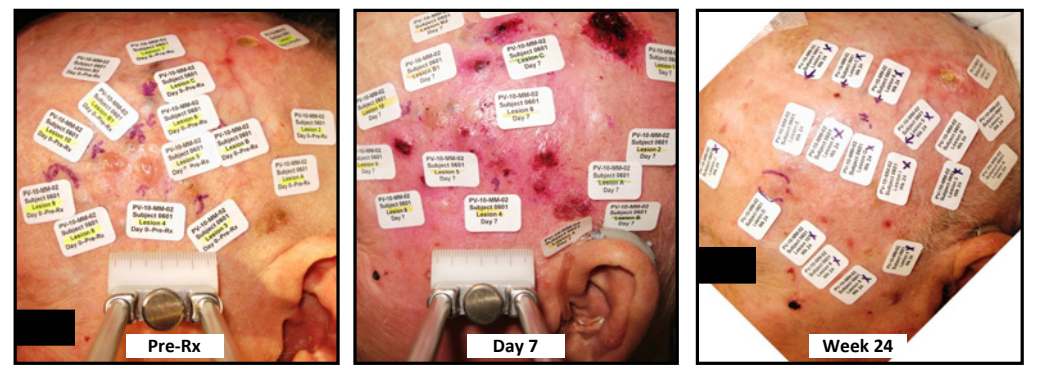

Pre-Rx
Day 7
Week 24

Subject 0601: Male, 79, Stage IV (M1b) since 2004, multiple Sx, CLND, 30 Gy XRT.
Three treatments (Day 0, Wk 10 and Wk 19) with PV-10 to 15 lesions; 2 untreated bystander lesions.

Phase 2 Interim Efficacy

Objective Response of Study Lesions
All Subjects (N=40)

Best Response (RECIST, N = 40 Subjects through Week 52)	Target Lesions N		Bystander Lesions N	
N (Subjects)	40		21	
CR	13	33%	7	33%
PR	11	28%	2	10%
SD	8	20%	3	14%
PD	8	20%	9	43%
ND	--	--	19	
CR + PR	24	60%	9	43%
CR + PR + SD (*Locoregional Disease Control*)	32	80%	12	57%

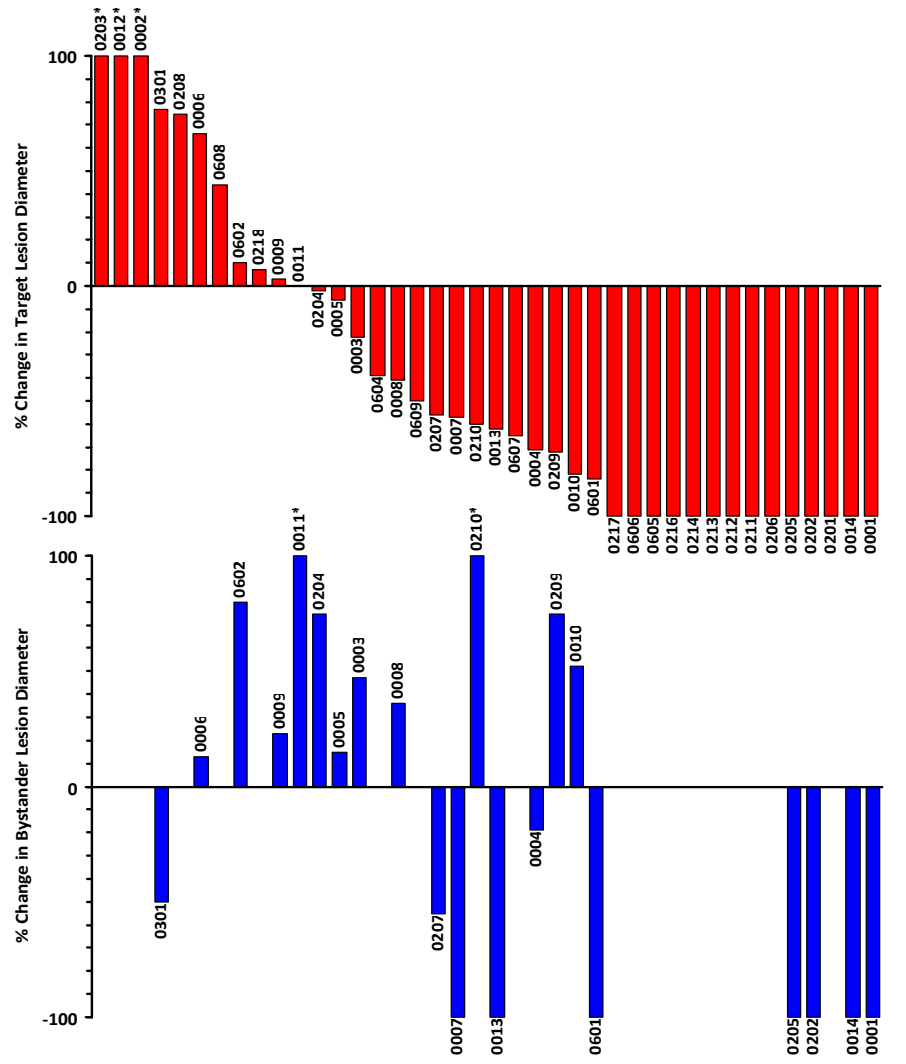

% Change in Target Lesion Diameter
100
0
-100
0203* 0012* 0002* 0301 0208 0006 0608 0602 0218 0009 0011 0204 0005 0003 0604 0008 0609 0207 0007 0210 0013 0607 0004 0209 0010 0601 0217 0606 0605 0216 0214 0213 0212 0211 0206 0205 0202 0201 0014 0001
% Change in Bystander Lesion Diameter
0301 0006 0602 0009 0011* 0204 0005 0003 0008 0207 0007 0210* 0013 0004 0209 0010 0601 0205 0202 0014 0001

Objective Response of Bystander Lesions
Grouped According to Subject Objective Response of Target Lesions
All Subjects (N=40)

Bystander Lesion Response	Subjects with POSITIVE Objective Response of Target Lesions		Subjects with NEGATIVE Objective Response of Target Lesions		χ^2	Fisher Exact
N (Subjects)	13		8			
CR	7	54%	0	0%		
PR	1	8%	1	12%		
SD	1	8%	2	25%		
PD	4	31%	5	62%		
ND	(11)		(8)			
CR + PR	8	62%	1	12%	χ^2 = 3.067	P = 0.067
SD + PD	5	38%	7	88%	1 df	
					P = 0.080	

Response of each subject's bystander lesions (overall subject response) as a function of the subject's objective response of target lesions (POSITIVE Objective Response = CR + PR subjects; NEGATIVE Objective Response = SD + PD subjects). Statistical significance of response rates tested using the Chi-Square and Fisher Exact tests. Nineteen subjects had no designated bystander lesion (or no assessable lesion) to assess (ND) and were censored.

Objective Response of Target Lesions (by AJCC Stage)
All Subjects (N=40)

Best Response (RECIST, N=40, through Week 52)	Stage IIIB N		Stage IIIC N		Stage IV M1a N		Stage IV M1b N		Stage IV M1c N	
N (Subjects)	20		9		1		8		2	
CR	7	35%	2	22%	1	100%	3	38%	0	0%
PR	6	30%	2	22%	0	0%	2	25%	1	50%
SD	5	25%	1	11%	0	0%	1	13%	1	50%
PD	2	10%	4	44%	0	0%	2	25%	0	0%
CR + PR	13	65%	4	44%	1	100%	5	63%	1	50%
CR + PR + SD	18	90%	5	56%	1	100%	6	75%	2	100%

Survival

Progression Free Survival
All Subjects (N=40)

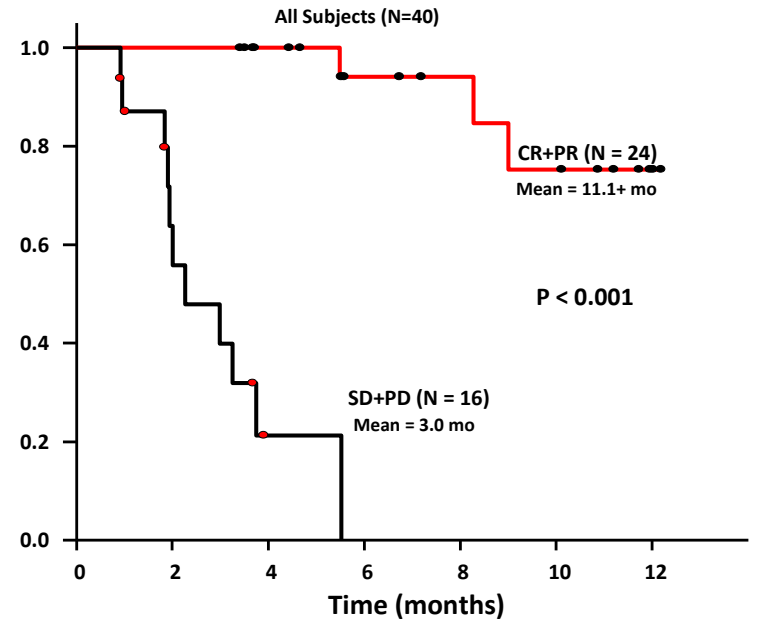

1.0
0.8
0.6
0.4
0.2
0.0
0 2 4 6 8 10 12
Time (months)
CR+PR (N = 24)
Mean = 11.1+ mo
P < 0.001
SD+PD (N = 16)
Mean = 3.0 mo

Overall Survival
All Subjects (N=40)

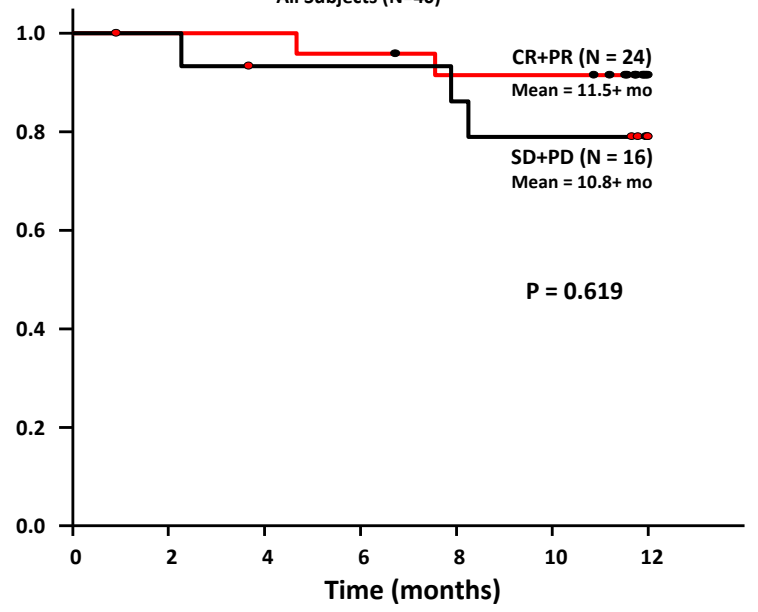

1.0
0.8
0.6
0.4
0.2
0.0
0 2 4 6 8 10 12
Time (months)
CR+PR (N = 24)
Mean = 11.5+ mo
SD+PD (N = 16)
Mean = 10.8+ mo
P = 0.619

CR + PR Cohort (24 subjects)
- 3 subjects (12%) had progression of injected lesions
- 2 subjects (8%) died from melanoma

SD + SP Cohort (16 subjects)
- 11 subjects (69%) had progression of injected lesions
- 3 subjects (19%) died from melanoma

Adverse Events

Adverse Events At Least Possibly Related to Treatment
Protocol PV10-MM-02 – 52 Weeks Follow-up, All Subjects (N = 40)
Events occurring in less than two subjects and with severity < 3 not shown

System Organ Class[1] / Preferred Term[1]	Adverse Events (by CTCAE Grade) 1	2	3	4	Total	%
General disorders and administration site conditions						
Injection site pain	15	15	**3**	0	33	82%
Injection site vesicles	12	7	**1**	0	20	50%
Injection site oedema	11	9	0	0	20	50%
Injection site swelling	4	3	0	0	7	18%
Injection site pruritus	5	2	0	0	7	18%
Injection site erythema	3	2	0	0	5	12%
Injection site infection	2	1	0	0	3	8%
Lethargy	3	0	0	0	3	8%
Injection site warmth	1	1	0	0	2	5%
Injection site photosensitivity reaction	2	0	0	0	2	5%
Injection site cellulitis	0	0	**2**	0	2	5%
Injection site ulcer	2	0	0	0	2	5%
Skin and subcutaneous tissue disorders						
Skin discolouration	5	2	0	0	7	18%
Injury, poisoning and procedural complications						
Skin flap necrosis	0	0	**1**	0	1	3%
Gastrointestinal disorders						
Diarrhoea	3	0	0	0	3	8%
Metabolism and nutrition disorders						
Decreased appetite	2	0	0	0	2	5%
Nervous system disorders						
Headache	6	1	0	0	7	18%
Renal and urinary disorders						
Polyuria	2	0	0	0	2	5%

[1] System Organ Class and Preferred Term are based on the MedDRA version 12.1 dictionary

Visceral Lesion Response

Changes in Untreated Nodal and Visceral Lesions
Grouped According to Objective Response of Target Lesions

Subject	Stage	TL	Nodal or Visceral Disease: Status at Screening	Outcome	Scoring
0205	IIIC (N3)	CR	4 +nodes right axilla, 16-23 mm (per U/S)	Stable at Wk 24	SD
0206	IV (M1b)	CR	Multiple pulmonary lesions, 4-13 mm	Slight reduction in size at Wk 12	SD
0216	IV (M1b)	CR	Multiple bilateral pulmonary lesions (largest 11 x 11 mm)	Generalized improvement at Wk 12; "no focal parenchymal pathology indicative of metastatic disease" at Wk 52	CR
0606	IV (M1b)	CR	Multiple small, stable pulmonary lesions	Stable through Wk 36	SD
0007	IIIC (N3)	PR	Axillary node met (17 mm)	14 mm at Wk 12	SD
0010	IV (M1c)	PR	Liver (multiple 7-13 mm) + Lung (9 mm)	Stable through Wk 36	SD
0207	IV (M1b)	PR	Probable multiple pulmonary, axillary lesions	NR	--
0217	IIIB (N2c)	PR	Possible 1.5 mm cerebral lesion at screening	NR	--
0601	IV (M1b)	PR	Pulmonary nodule right upper lobe (6 mm)	Possible increased size, new nodules at Wk 12	PD
0005	IV (M1c)	SD	Axilla, Chest Wall and Liver	NR	--
0009	IIIB (N2c)	SD	Possible numerous 5-6 mm lesions both lungs	NR	--
0011	IV (M1b)	SD	Possible pulmonary met	4 pulmonary mets at Wk 12	PD
0218	IIIB (N2c)	SD	Possible soft tissue mass posterior to spleen	NR	--
0002	IIIC (N3)	PD	Groin (nodal lesion)	NR	--
0006	IV (M1b)	PD	Pulmonary lesion upper lobe (6 mm)	Unchanged through Wk 24	SD
0208	IV (M1b)	PD	Bilateral small pulmonary lesions	NR	--
0220*	IV (M1c)	--	Several 6-8 mm liver mets	Markedly progressive widely disseminated liver metastases at Wk 12	PD
0222*	IV(M1b)	--	Possible small pulmonary met	Unchanged at Wk 12	--
0907*	IV (M1c)	PR	Multiple pulmonary and brain nodules	"Near complete resolution" of pulmonary nodules at Wk 12, incremental improvement of brain nodules at Wk 19, alive at Wk 52	PR
1006*	IV (M1b)	--	Probable bone, pulmonary and nodal mets	NR (alive at Wk 52)	--

Sixteen subjects in the N=40 interim assessment cohort presented evidence of nodal or visceral disease that was documented at screening. Response of these lesions over the study interval is grouped according to the subject's objective response of their target lesions. Overall 7 of 16 (44%) exhibited regression or stasis of these lesions (similar to the rate of response in bystander lesions). TL = target lesions, NR = not reported.
*Subjects in second study cohort (N = 41–80).

Subject 0907: Male, 40, Stage IV (M1c) since 2006
Multiple Sx, CLND, whole brain XRT, stereotactic radiosurgery, DTIC, IV- and SQ-IFN.
Three treatments (Day 0, Wk 8 and Wk 12) with PV-10 to 10 cutaneous lesions.

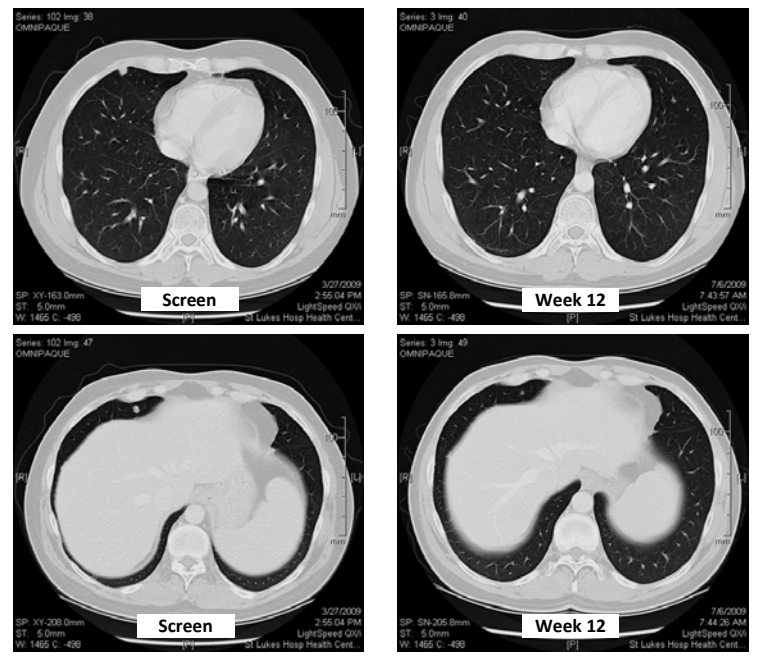

Screen
Week 12
Screen
Week 12

Interval progression of widespread 6–9 mm pulmonary mets at screening.
"Near complete resolution" of pulmonary nodules observed at Wk 12.

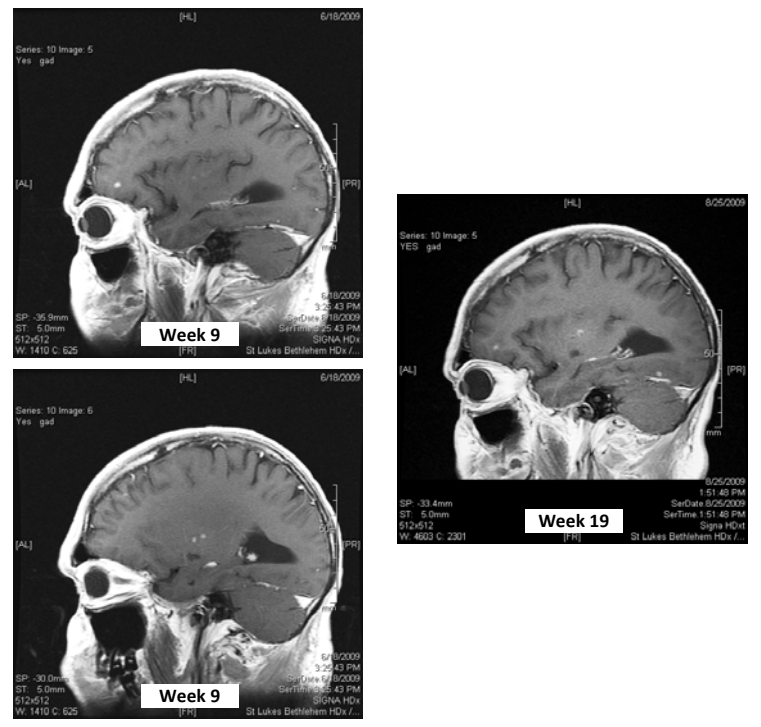

Week 9
Week 19
Week 9

Stereotactic radiosurgery (18 Dec 2008), whole brain XRT (17 Feb 2009), PV-10 (13 Apr, 08 Jun and 06 Jul 2009)
New lesions left frontal and right superior temporal lobes at Wk 9 (18 Jun 2009 MRI vs 6 Jan 2009 baseline)
"The majority of lesions which were previously identified appear improved" at Wk 19 vs Wk 9.

PV-10 Pharmacokinetics

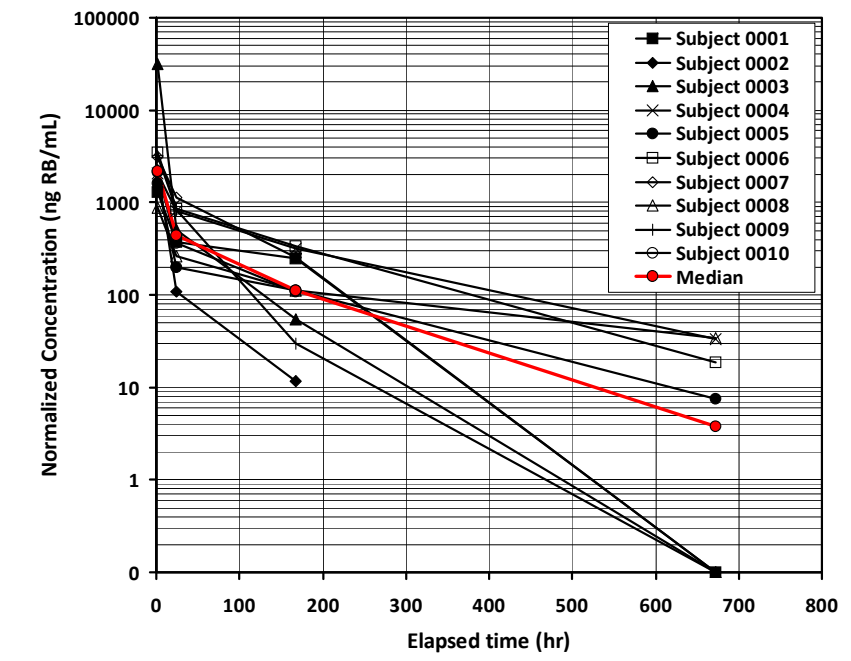

Normalized Concentration (ng RB/mL)
100000
10000
1000
100
10
1
0
0 100 200 300 400 500 600 700 800
Elapsed time (hr)
Subject 0001
Subject 0002
Subject 0003
Subject 0004
Subject 0005
Subject 0006
Subject 0007
Subject 0008
Subject 0009
Subject 0010
Median

RB extravasate is rapidly cleared with prolonged retention in tumor

- PK samples collected from 10 phase 2 participants over 28 days following 1st injection
- Data normalized to uniform dose (15 mL dose in a 70 kg subject)
- Bi-exponential clearance (extravasate followed by tumor depot):
 - $C_{initial}$ 2,400 ng RB/mL
 - k_A 0.0018 min^{-1}; kE 0.00011 min^{-1}
 - $t_{1/2, A}$ 376 min (6.3 hours); $t_{1/2, E}$ 6,200 min (4.3 days)
- Rapid clearance consistent with observed safety of PV-10
- Systemic uptake and clearance compare favorably with Robengatope® bolus ($C_{initial} \approx$ 20,000 ng/mL and k_E = 0.0039 min^{-1})
- Plasma levels decrease to < 5% of $C_{initial}$ within 7 days, making cumulative toxicity unlikely upon repeat treatment at intervals of 1 week or greater

Current Activities in Cutaneous Tumors

PV-10-MM-02X – Continuation study available to Phase 2 participants

- Subjects with evidence of response to PV-10 during Phase 2 where disease was not completely controlled under Phase 2 dosing schedule
- Allows multiple treatments as indicated NLT 28 days apart for up to 24 months
- 8 subjects have been enrolled

PV-10-EA-02 – Expanded access for solid cutaneous or subcutaneous tumors

- Open to PV-10 naïve patients with cutaneous / SQ tumors
- Dosing and follow-up identical to protocol PV-10-MM-02X
- 25 subjects enrolled to date at existing Phase 2 centers in USA and AUS

PV-10-XRT-01 – IL PV-10 followed by radiotherapy in the treatment of melanoma

- Single center investigator-initiated study of PV-10 chemoablation followed by XRT
- Single intralesional PV-10 dosing; if CR not achieved 6 fractions x 5 Gy at 6-10 weeks post-PV-10
- Up to 25 subjects anticipated

Future Activities

Phase 3 and Accelerated Approval

- Initial End-of-Phase-2 meeting conducted with U.S. FDA in April 2010
 - Agreed to finalize proposed Phase 3 RCT study design incorporating Agency input to qualify submission for Special Protocol Assessment (SPA)
 - Anticipate durable response as primary endpoint
 - Approximately 300 subjects, expected to commence in 2011
 - Currently recruiting investigators in USA, AUS and EU
- Continuing to assess potential paths to accelerated approval in the USA and abroad

Mechanism of Action

- Phase 2B study planned to fully validate bystander effect
 - Response in untreated proximal and visceral lesions consistent with immunologic process
 - PV-10 chemoablation yields immediate reduction in tumor burden
 - Ablation may recruit immune cells to exposed tumor antigens
 - Assess immune markers in peripheral blood and tumor tissue

Conclusions

PV-10 is well tolerated, eliciting a robust response in a majority of patients

- Response of injected lesions appears to be unrelated to disease stage or prior treatment
- The safety and efficacy profile compare favorably with existing and emerging therapies
- Suitable for repeat treatment to maximize OR, ablate new lesions and enhance long-term outcome
- Non-responsive patients are quickly evident, avoiding delay in transition to alternate therapy

Locoregional treatment may yield systemic benefit via the bystander effect

- Bystander effect in untreated cutaneous lesions correlates closely with response of injected lesions
- Stasis or regression of visceral lesions evident in several subjects
- PV-10 offers potential locoregional control of metastatic disease