Combination of PV-10 Immuno-chemoablation and Systemic anti-CTLA-4 Antibody Therapy in Murine Models of Melanoma

Abstract No. 4755
American Association for Cancer Research Annual Meeting – Washington, DC
6 - 10 April 2013

Eric A. Wachter , Savannah O. Blair, Jamie M. Singer and H. Craig Dees
Provectus Pharmaceuticals, Inc., Knoxville, TN USA

For additional information:
Eric Wachter
wachter@pvct.com

Abstract

Rose bengal disodium (PV-10) is an investigational small molecule ablative agent currently entering pivotal phase 3 clinical testing as a monotherapy for locoregional control of cutaneous metastatic melanoma. Upon intralesional (IL) administration, PV-10 localizes to the injected tumor tissues while clearing rapidly from healthy tissue. Tumor infiltration with PV-10 leads to rapid necrosis of the injected lesion, with complete resolution common within 2-8 weeks. In phase 2 testing in 80 patients with Stage IIIB-IV (M1c) melanoma, IL PV-10 elicited an objective response in injected tumors in 51% of patients (CR:26%, PR:25%) after 1-4 treatment cycles. In addition to this direct ablative effect on injected tumors, some patients achieved an objective response in their monitored untreated tumors (CR:26%, PR:7% in 42 subjects with monitored untreated lesions) in an apparent immune-mediated bystander response that highly correlated with successful ablation of their injected tumors. Treatment was generally well tolerated, with adverse events confined mainly to the injection site and no grade 4 or 5 adverse events associated with use of PV-10. Recent nonclinical testing in the B16-F10 murine melanoma tumor line has confirmed that PV-10 ablation induces tumor-specific immunity, resulting in marked suppression of synchronous lung metastases upon ablation of a flank tumor and tumor-specific IFN-γ production. In this study we assess potential benefit of combination of PV-10 immuno-chemoablation with the hamster anti-murine CTLA-4 antibody 9H10 in bilateral flank and lung metastasis models (B16-F10 melanoma in C57BL/6 mice). Results from these models are reported and support clinical development of combination therapy in advanced melanoma patients, such as stage IV patients with substantial tumor burden in locations inaccessible to PV-10 injection. The rapid reduction in tumor burden and tumor specific immunologic stimulation provided by PV-10 appears to complement the immune stimulation of anti-CTLA-4 antibodies such as ipilimumab without increased toxicity.

PV-10 Immuno-Chemoablation [1],[2]

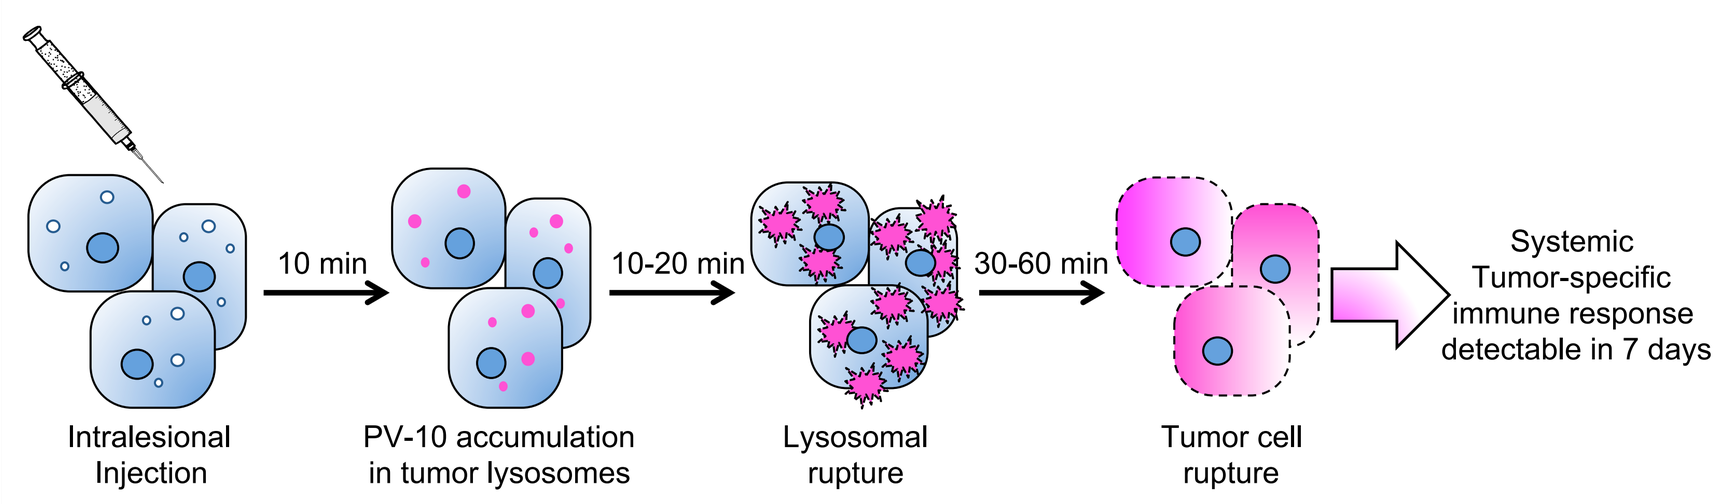
10 min
10-20 min
30-60 min
Systemic Tumor-specific immune response detectable in 7 days
Intralesional Injection
PV-10 accumulation in tumor lysosomes
Lysosomal rupture
Tumor cell rupture

Clinical Background

Rose bengal disodium (PV-10) is a small molecule immuno-chemoablative agent directly injected into tumors that is rapidly cleared from systemic circulation. Intermittent intralesional exposure to PV-10 in Phase 2 testing (limited to 4 doses over 16 weeks) resulted in locoregional disease control (CR+PR+SD) in 69% of patients. Complete response was observed in 54% of patients who had all of their tumor burden treated whereas those patients who entered the study with stage IV disease exhibited a substantially less robust response, typically due to progression prior to repeat dosing with PV-10.

Untreated Tumor Burden vs. Objective Response
Phase 2 Melanoma Study PV-10-MM-02

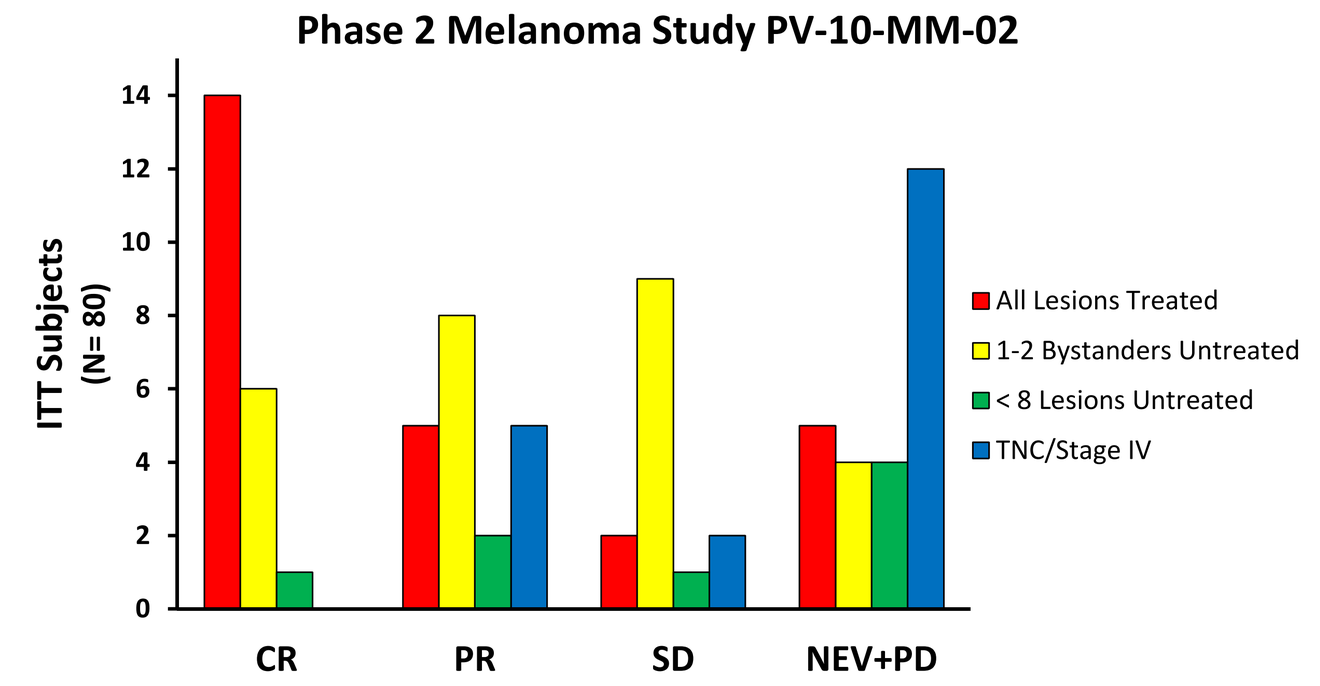
ITT Subjects (N= 80)
All Lesions Treated
1-2 Bystanders Untreated
< 8 Lesions Untreated
TNC/Stage IV
CR
PR
SD
NEV+PD

Outcome would potentially improve with more frequent dosing, however for those patients with extensive tumor burden inaccessible to PV-10 injection, a combination therapy strategy should be considered. Intralesional injection focuses PV-10 ablative impact within injected tumors and minimizes potential for systemic side effects, making PV-10 an attractive candidate for combination with systemic agents.

Most Frequent AEs at Least Possibly Related to PV-10 Treatment
All ITT Subjects (N = 80)

System Organ Class[1] Preferred Term[1]	Adverse Events[2] All ITT Subjects (N=80) Mild	Mod	Sev	Life	Total	%
General disorders and administration site conditions						
Injection Site Discoloration	13	12	0	0	25	31%
Injection Site Erythema	6	4	1	0	11	14%
Injection Site Oedema	19	14	0	0	33	41%
Injection Site Pain	29	25	10	0	64	80%
Injection Site Pruritus	14	3	0	0	17	21%
Injection Site Swelling	14	7	1	0	22	28%
Injection Site Vesicles	17	13	1	0	31	39%
Nervous System Disorders						
Headache	11	2	0	0	13	16%

[1] System Organ Class and Preferred Term are based on the MedDRA® terminology dictionary.
[2] If a subject experienced an adverse event more than once during the study period, the greatest severity is presented. No grade 5 (death) AEs were reported at least possibly related to treatment.

Potential for Complementary Therapy

- **Temporarily Arrest Tumor Growth in Disease Inaccessible to IL PV-10 and/or**
- **Amplify Tumor-Specific Immune Response upon PV-10 Ablation**

Prior to clinical investigation of a sorafenib + PV-10 combination in hepatocellular carcinoma (HCC) under protocol PV-10-LC-01, a similar combination was modeled (PV-10 + 5-fluorouracil) in mice. This combination showed a synergistic effect in a HCC bilateral flank model, including a longer time to progression in untreated tumors [3]. In this work systemic immune stimulation was modeled using the murine analog of anti-CTLA-4 therapy, 9H10, in combination with PV-10. Successful anti-CTLA-4 combinations in murine models have been reported in prostate (9H10 + cryoablation) [4] and breast cancer cell lines (9H10 + radiation) [5], and B16-OVA melanoma (9H10 + radiofrequency ablation) [6]. None of these models left untreated flank tumor burden to model visceral disease, rather induced lung metastases were counted or mice were re-challenged with tumor. Elsas and coworkers reported reduction in single flank tumor burden compared to controls for a B16-BL6 vaccine with 9H10 upon initiation of treatment either 8 or 12 days after tumor implantation [7]. At earlier time points (not shown), the mice never manifested palpable flank tumors (only 4 of the 5 mice treated with the combination at day 8 had a measureable tumor).

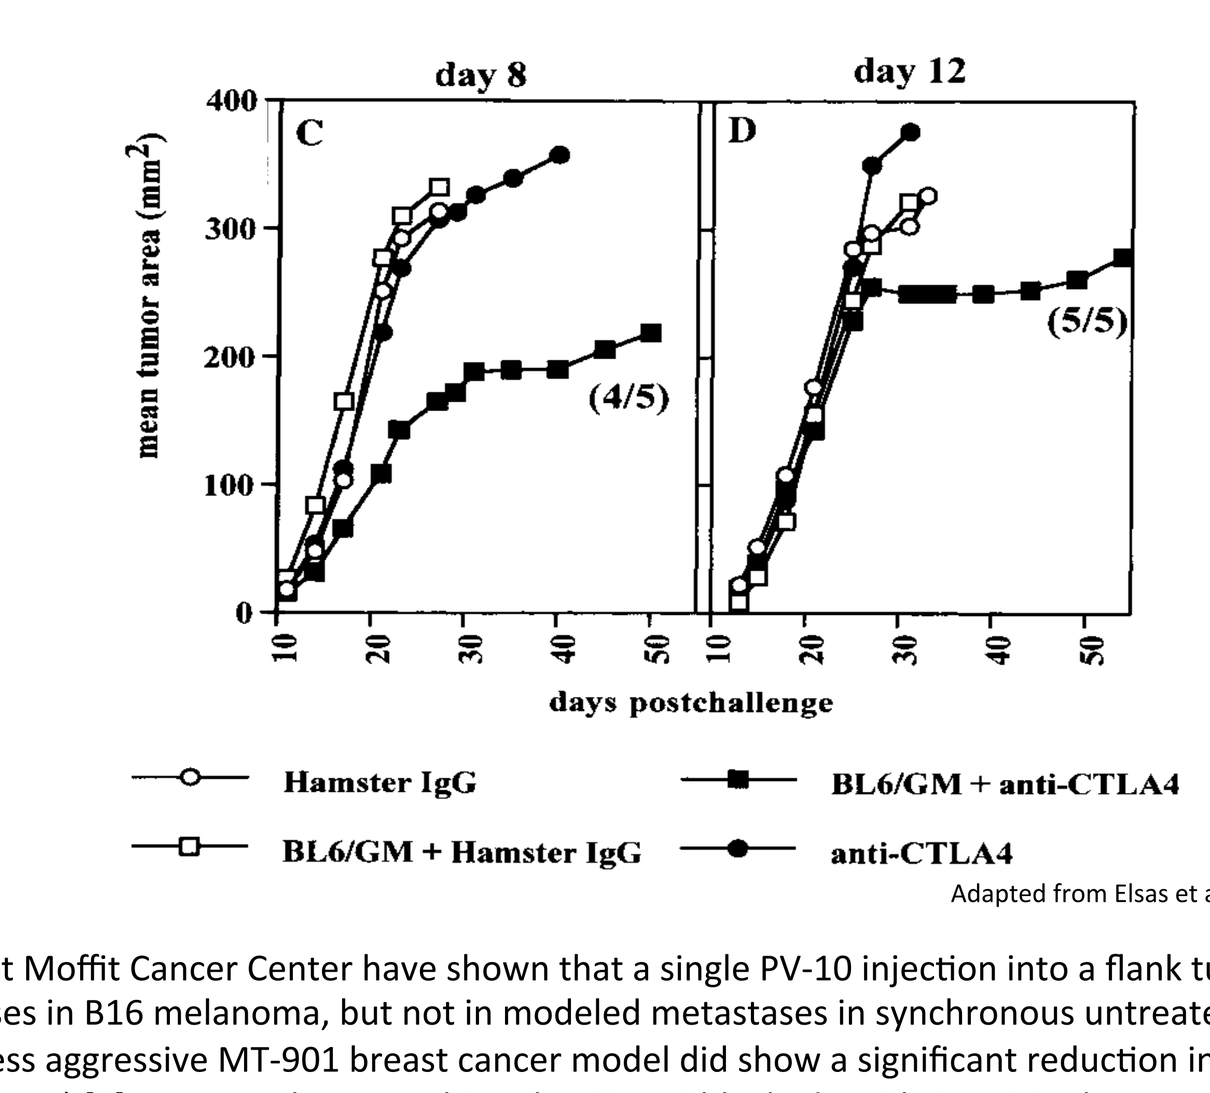
day 8
day 12
C
D
mean tumor area (mm²)
(4/5)
(5/5)
days postchallenge
Hamster IgG
BL6/GM + Hamster IgG
BL6/GM + anti-CTLA4
anti-CTLA4

Adapted from Elsas et al.

Researchers at Moffit Cancer Center have shown that a single PV-10 injection into a flank tumor can reduce lung metastases in B16 melanoma, but not in modeled metastases in synchronous untreated B16 flank tumors (the less aggressive MT-901 breast cancer model did show a significant reduction in untreated bystander tumors) [2]. We sought to combine the CTLA-4 blockade with PV-10 in the most aggressive models of existing tumor burden to ensure that any systemic treatment was potentially safe in combination with PV-10 and efficacy signals from the combination could be discriminated from those of PV-10 alone.

Lung Metastasis + Flank Tumor Model (B16-F10)

Background: Elsas et al. reported CTLA-4 blockade in combination with F10/GM vaccine reduced lung metastases (no flank tumor). Both treatments commenced 1 day after tail vein injections of 5 x 10^4 B16-F10 cells (9H10 IP dose: 100 µg, 50 µg, 50 µg, days 1, 4 and 7). CTLA-4 blockade alone resulted in > 200 lung mets in each animal, but hamster IgG control showed some lung met reduction. Since hamster IgG was not reported to have an effect in any of the other literature reviewed, we did not contemplate using that control in these studies. We first wanted to ensure that 9H10 or some IgG component therein did not reduce lung metastases in our model.

Murine Model: C57BL/6 (6 females per group)

Tumor Induction: Solitary SQ tumors were induced in the flank by injection of B16-F10 tumor cell line (2 x 10^5 cells). Lung metastases were produced by synchronous injection of tumor cells into the tail vein (2.5 x 10^5 B16-F10 cells).

Treatment: 8 days post tumor induction, a single IL injection of 50-100 µL of PV-10 (dose based on tumor size) or a control injection of 100 µL IL saline was administered into the flank lesion. 100 µg of 9H10 (BioXcell) was administered intraperitoneally (IP) on day 0, and 50 µg each on days 3 and 6.

Follow up: 19 days post tumor induction, lungs were removed and metastases were counted on the anterior and posterior sides using a 1 mm^2 grid. Each 1 mm^2 of tumor tissue was counted as 1 lung metastasis.

Lung Metastasis Results

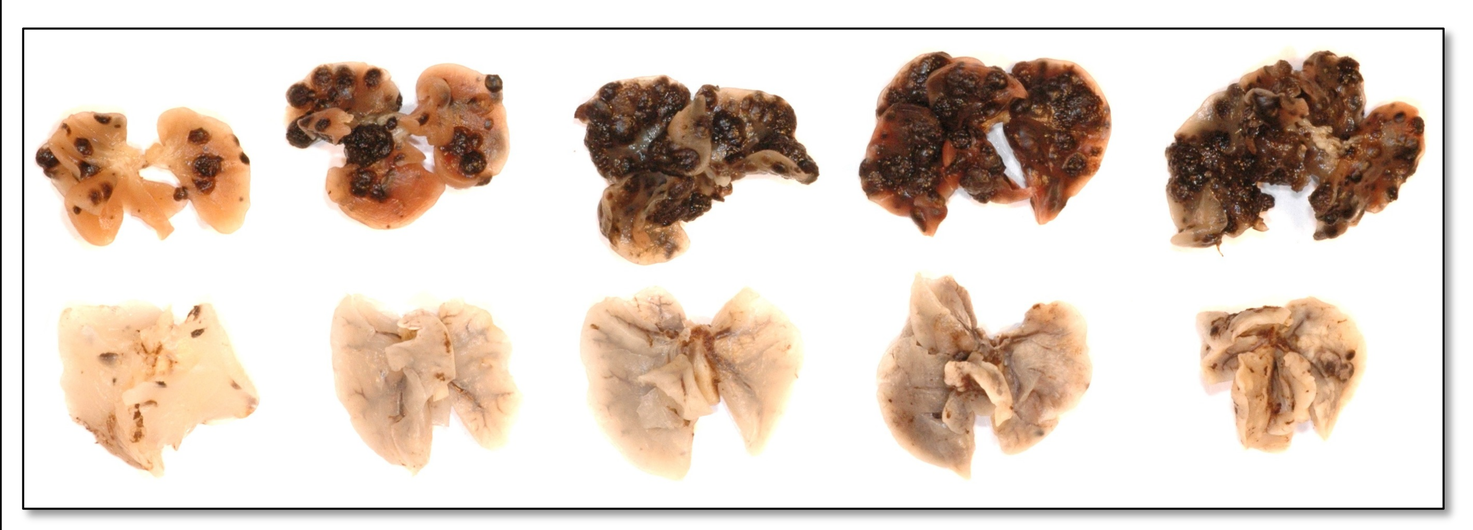
Treatment:
IP 9H10 (x3)
IP 9H10 (x3) + IL PV-10 (x1)

	Number of Lung Metastases
IL Saline + IP 9H10	37
	85
	>100
	>100
	>100
	*death prior to day 19
IL PV-10 + IP 9H10	0
	0
	3
	4
	6
	6

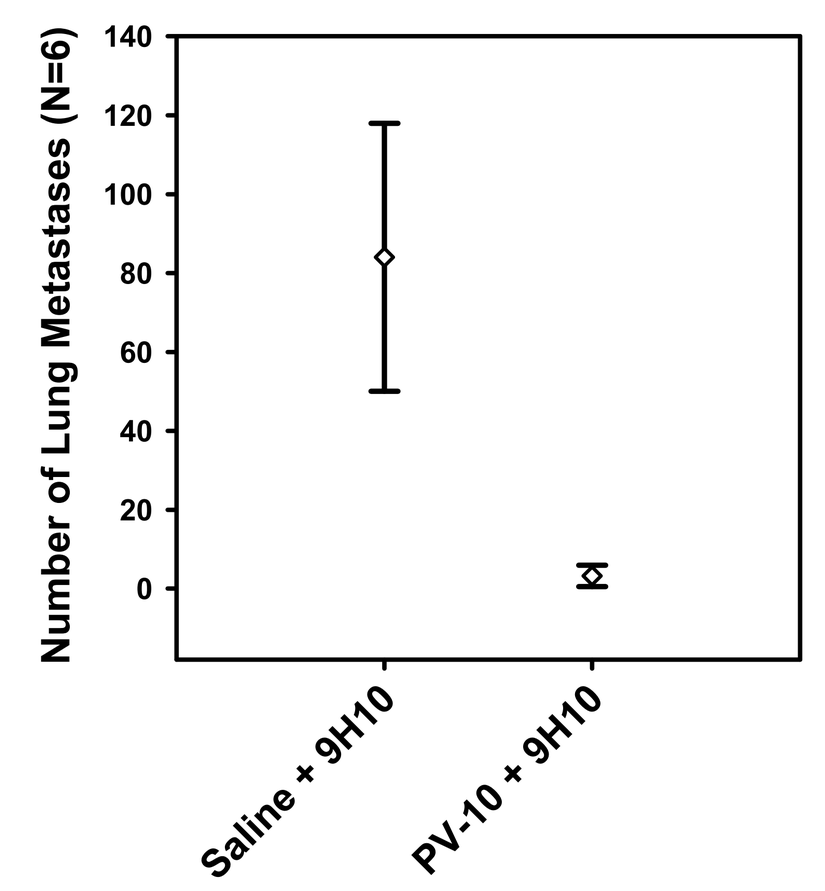
Number of Lung Metastases (N=6)
Saline + 9H10
PV-10 + 9H10

Lung Metastasis Results: Mice that received IL PV-10 + IP 9H10 exhibited a dramatic reduction in the number of synchronous lung metastases (p = 0.004). Mice treated with IL PV-10 + IP 9H10 had a mean of 3.2 lung metastases (range 0 to 6). Control mice treated with IL Saline + IP 9H10 had a mean of 84.0 lung metastases (range 37 to >100). It is unknown whether the combination is superior to PV-10 alone in this model since lung met results are comparable to previous tests of PV-10 alone [3].

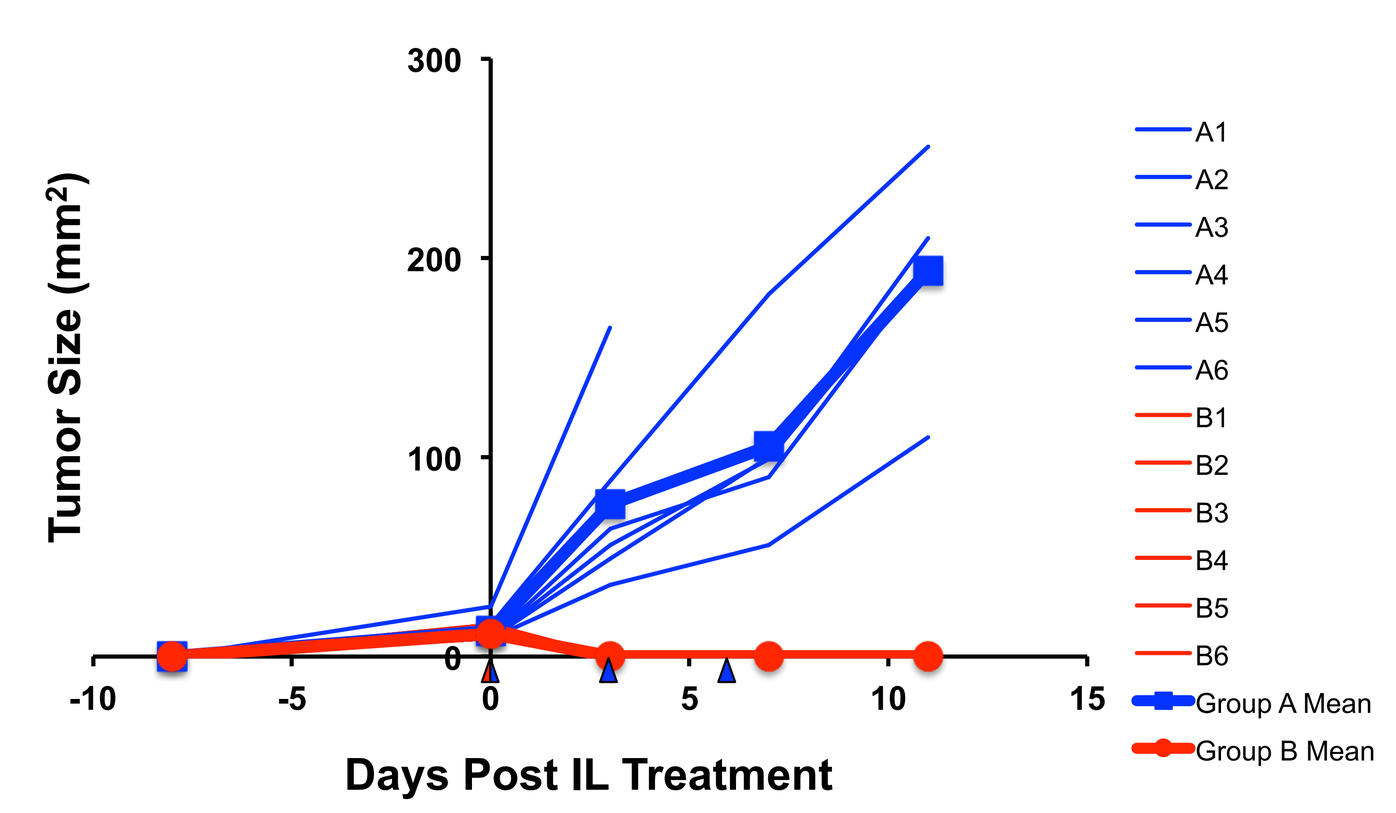
IL Saline + IP 9H10 vs. IL PV-10 + IP 9H10
Flank Tumor
Tumor Size (mm²)
Days Post IL Treatment
A1
A2
A3
A4
A5
A6
B1
B2
B3
B4
B5
B6
Group A Mean
Group B Mean

Synchronous Flank Tumor Results: The IL saline + IP 9H10 group tumor size means were 13 mm^2 at onset of treatment and 194 mm^2 19 days after cell implantation, individual animals are shown in thin lines above. The tumors in the PV-10 combo group were not measurable after ablation. Consistent with previous studies, the reduction in flank tumor burden in the PV-10 group is statistically significant.

Statistical Methods

Lung Metastases: Number of lung metastases on the anterior and posterior sides of each lung were enumerated from standardized photographs using a 1 mm^2 grid, with each 1 mm^2 of tumor tissue counted as 1 lung metastasis. The Mann-Whitney rank sum test was used to analyze for differences in group medians.

Tumor Area: Treated, untreated and total tumor burden (length x width) were analyzed using an Analysis of Covariance (ANCOVA) unequal slopes or parallel lines model, with pre-treatment Study Day 0 values used as the covariate, to adjust for baseline differences. Normality was assessed by various plots of residuals and by the Shapiro-Wilk test, while homogenous group variance was assessed using Levene's test at Day 0 (assessments at the 1% level of significance and visual inspection of residual plots). Consistent violations of either assumption across time points would have resulted in an appropriate data transformation (e.g. log, rank, etc.) to correct the problem.

Survival: Survival was analyzed using the Kaplan-Meier product-limit log-rank test to compute nonparametric survivor function estimates.

Group Comparisons: Pairwise group differences for all statistical analyses were assessed using the Tukey-Kramer multiple comparisons adjustment, which is based on Tukey's Studentized Range Test. All statistical tests of pairwise treatments differences were conducted two-sided, at the 5% level of significance.

Group Comparisons between Experiments: Pairwise group differences for evaluations of the PV-10 + 9H10 combinations across the three dose levels were validated using the Kruskal-Wallis one way Analysis of Variance (ANOVA) on Ranks with pairwise multiple comparison (Tukey Test) on baseline tumor burden (treated, untreated and total tumor burden). All were conducted two-sided, at the 5% level of significance.

Arbor Analytics

Bilateral Flank Model: Low Dose 9H10 (B16-F10)

Bilateral Flank Model: In order to discriminate the systemic effect of PV-10 alone from that in combination with CTLA-4 blockade, 3 dose levels of 9H10 were assessed in an aggressive melanoma model with four treatment groups (C57BL/6, 10 per group) randomized by tumor burden immediately prior to treatment. Tumors were induced in each flank by subcutaneous injection of B16-F10 tumor cells (2 x 10^5 cells per tumor). Mice were required to have palpable tumor burden in both flanks prior to randomization on treatment day 0. Only the larger of the flank tumors was infiltrated with saline (control) or PV-10 in each animal. The untreated flank tumor was followed as a surrogate for visceral disease burden inaccessible to PV-10 injection. Both tumors were measured in two dimensions with calipers in intervals of 2-5 days after IL-treatment (tumors that were eschar or no longer palpable were recorded as "0"). Animals were euthanized if on a measurement day the total tumor burden exceeded 300 mm^2, with last measurement captured. Statistically significant p-values were only reported in tumor burden measurement comparisons between groups if neither groups' survival medians had been reached.

Low Dose 9H10 Experiment: 9 days after bilateral tumor induction (treatment day 0), a single IL injection of 50-100 µL of PV-10 (dose based on tumor size) or 100 µL IL saline control was administered into the larger of the two lesions. For 9H10 treatment groups, 100 µg of 9H10 was administered IP on day 0, and 50 µg each on days 3 and 6 [7]. Differences in tumor burden at day 0 (shown below) were not significant.

Treatment	Mean Total Tumor Burden (mm^2)	Standard Deviation
IL-PV-10	33.3	14.8
IL-PV-10 + 9H10	30.7	12.8
IL-Saline	33.7	16.3
IL-Saline + 9H10	31.1	11.4

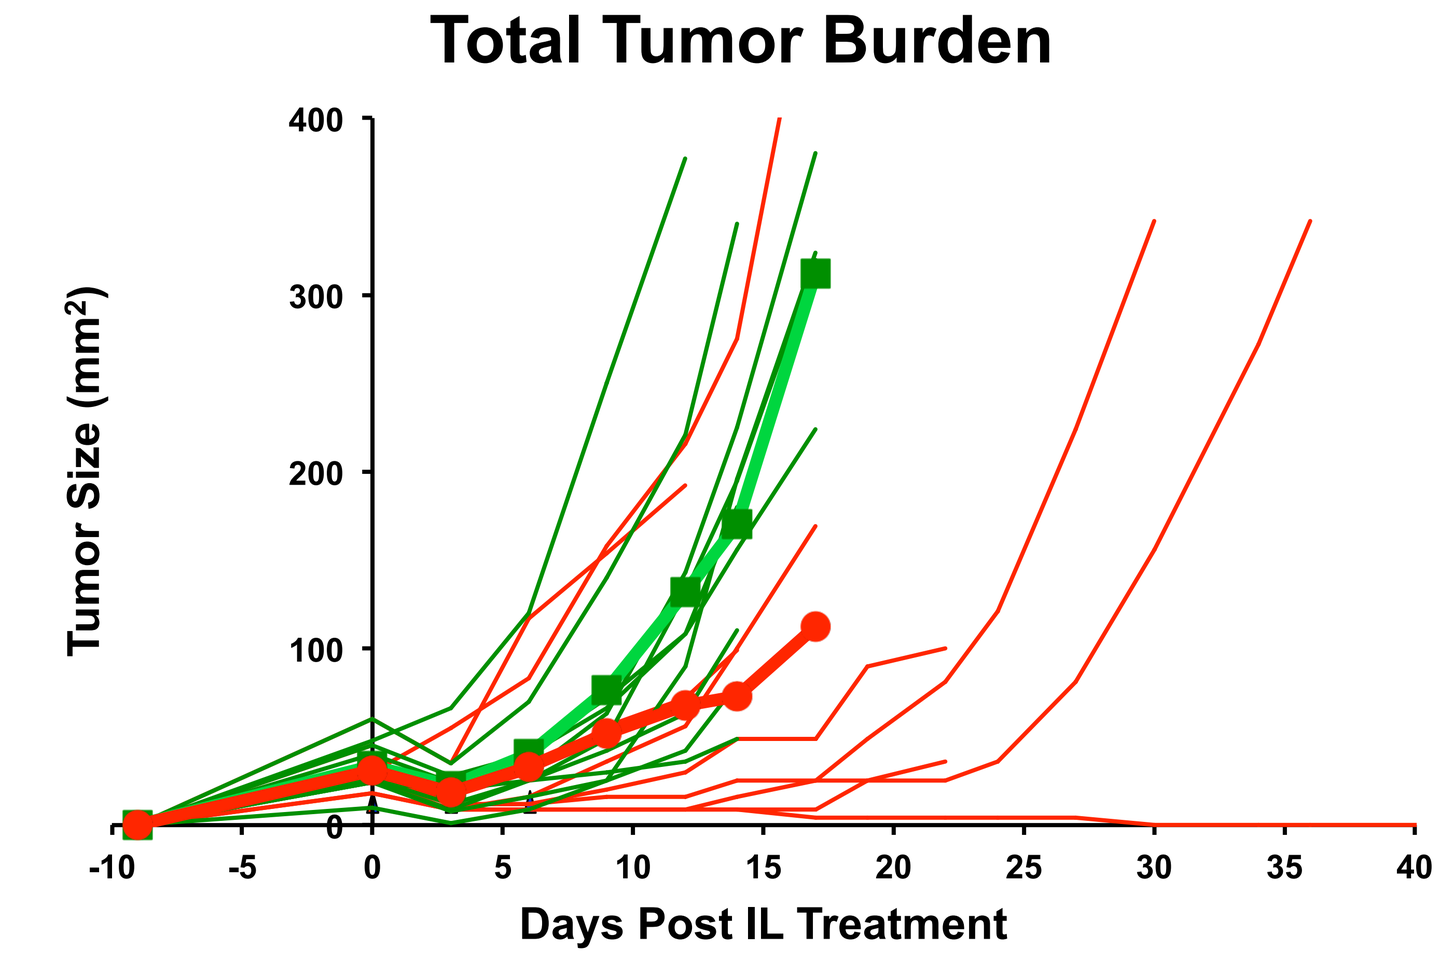
IL PV-10 vs IL PV-10 + IP 9H10
Total Tumor Burden
Tumor Size (mm²)
Days Post IL Treatment

Total Tumor Burden: A representation of PV-10 alone (green) and the combination of PV-10 + 9H10 (red) total tumor burden is shown above. The blue triangles represent 9H10 doses (PV-10 or saline injected IL only on day 0). Both treatment groups are significantly smaller than total tumor burden in the non-ablated saline and 9H10 controls. While the PV-10 + 9H10 group appears favorable to PV-10 alone, the groups aren't statistically different from one another in total tumor burden.

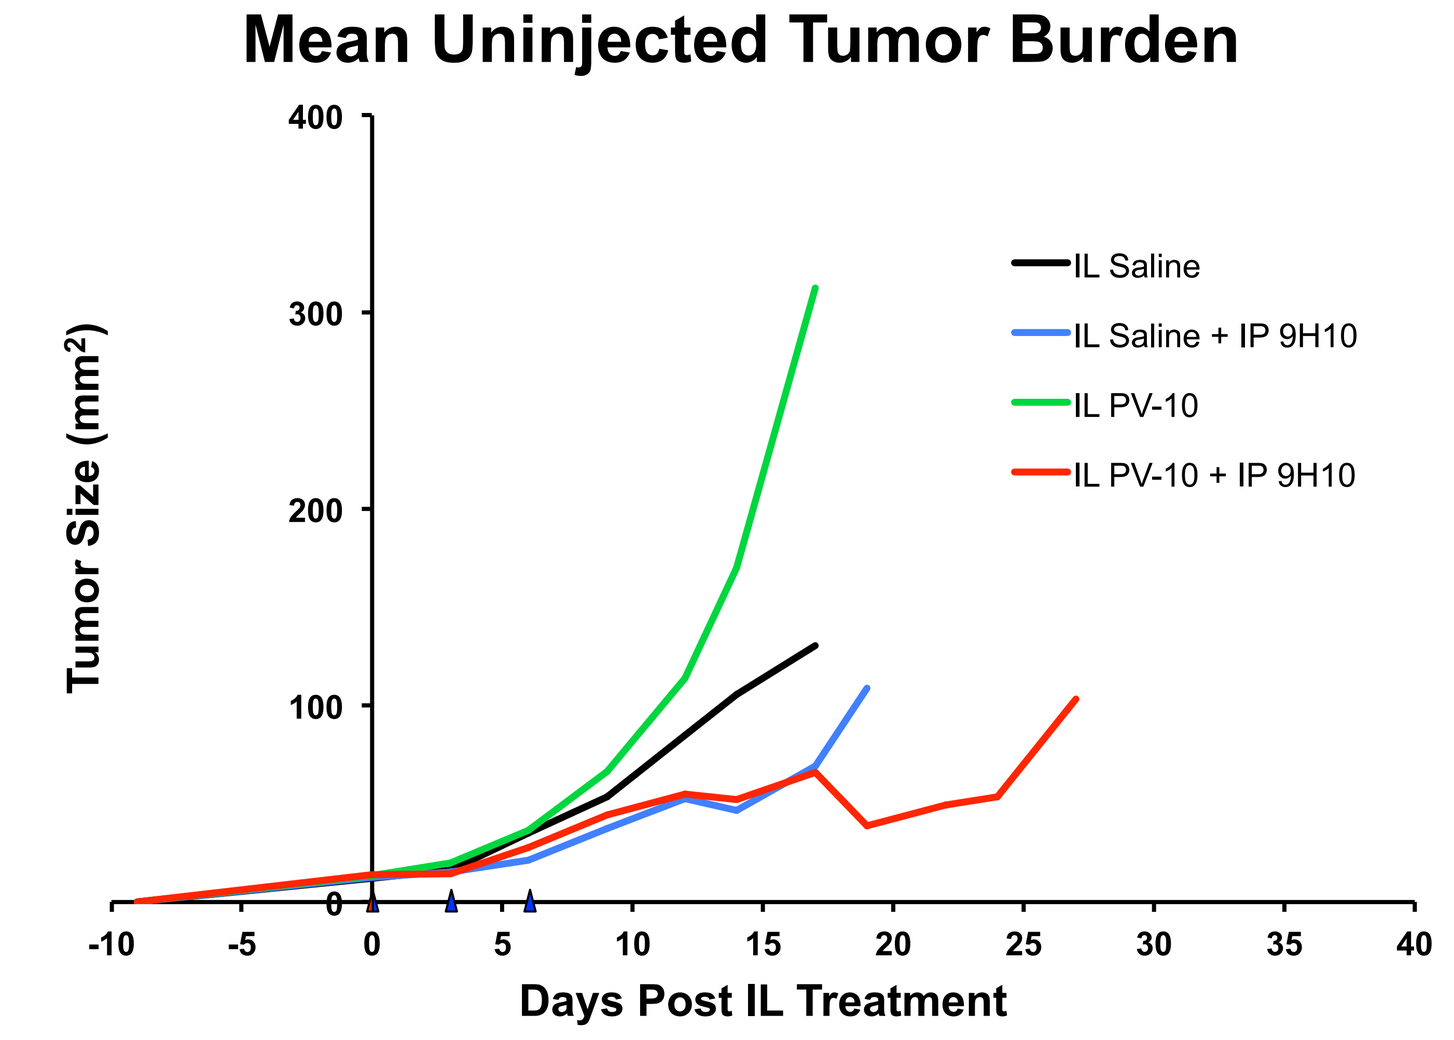
Mean Uninjected Tumor Burden
Tumor Size (mm²)
IL Saline
IL Saline + IP 9H10
IL PV-10
IL PV-10 + IP 9H10
Days Post IL Treatment

Modeled Visceral Disease (Uninjected Flank Tumors): A representation of all groups in the experiment, including 9H10 (blue) and saline (black) controls, is shown above (truncated at survival medians). The combination of PV-10 + 9H10 (red) better controlled un-injected tumor burden than PV-10 alone (green), attaining statistical significance at day 12, 14, 17. (p = 0.008, p = 0.001, p < 0.0001). IL-Saline + 9H10 un-injected tumors were also significantly smaller than PV-10 alone at days 12, 14 and 17.

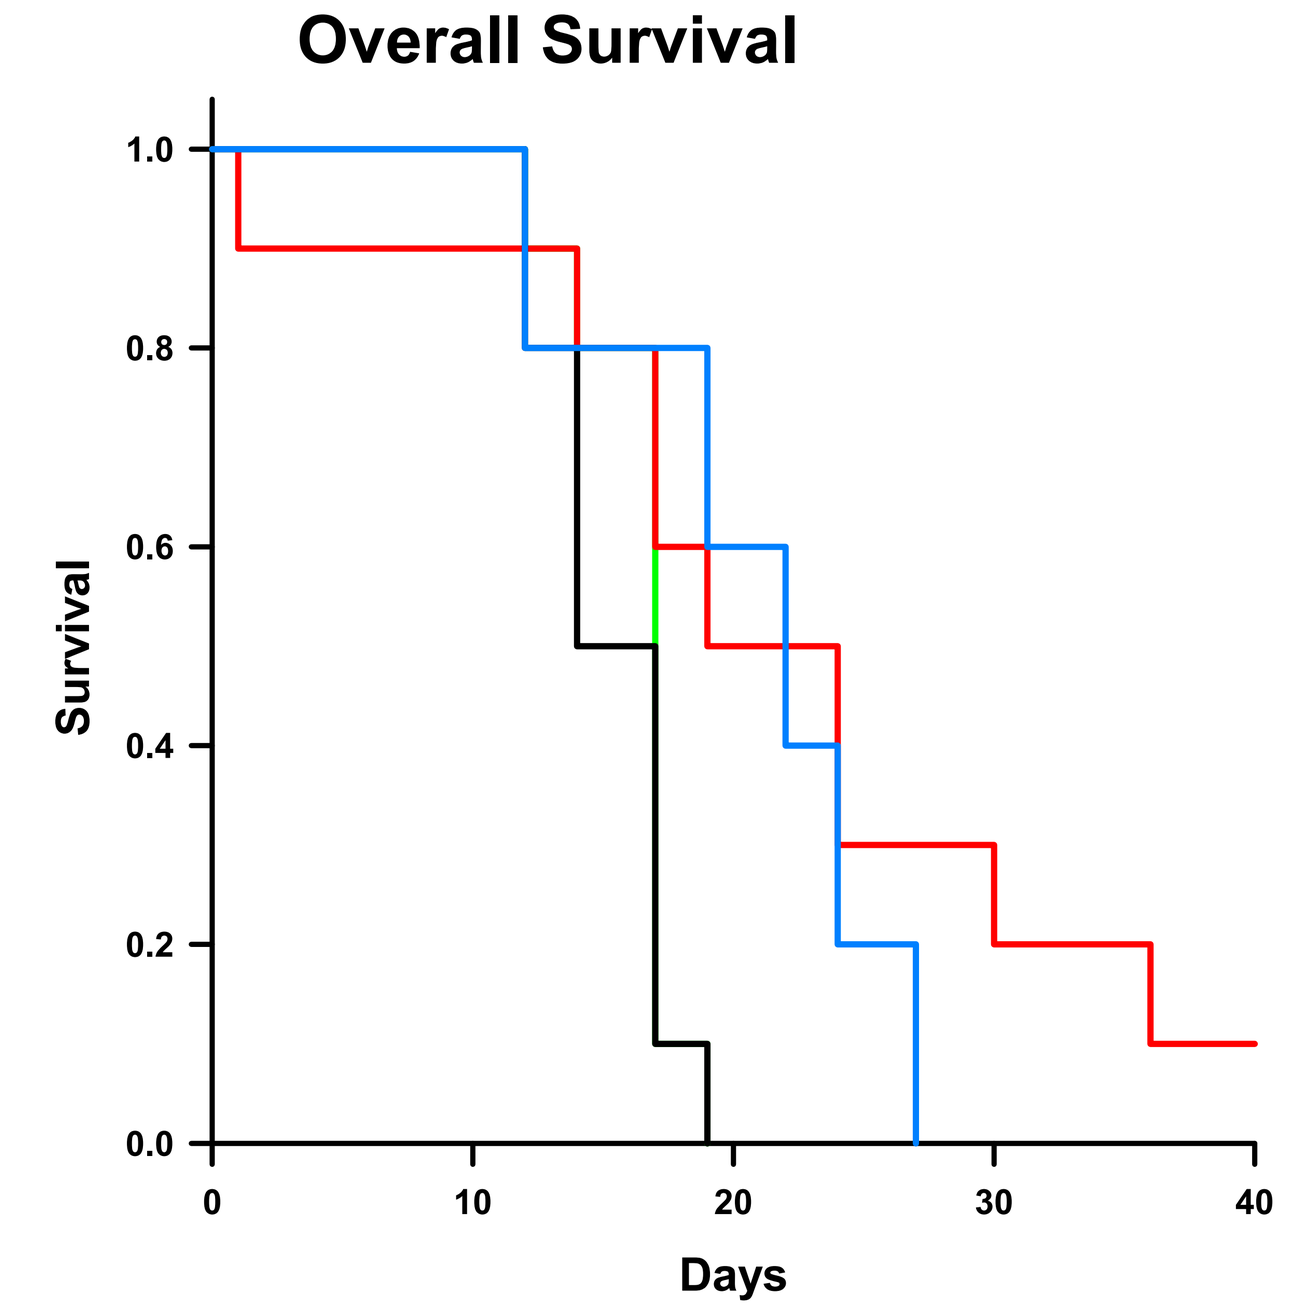
Overall Survival
Survival
Days

Overall Survival: The survival of all groups in the low dose experiment is represented above. Events included animals euthanized for excessive tumor burden. A single animal in the PV-10 + 9H10 arm was censored (sacrificed with no evidence of disease on day 56). The PV-10 + 9H10 combination (red) showed statistically significant survival over PV-10 alone (green, p = 0.052) and saline alone (black, p = 0.01), but not against 9H10 alone (blue, p = 0.13).

Bilateral Flank Model: Mid Dose 9H10 (B16-F10)

Mid Dose 9H10 Experiment: Waitz et al. reported a murine model of prostate cancer where cryoablation of a single TRAMP C2 tumor (5-8 mm in diameter) did not prevent growth of a secondary tumor given on the opposite flank 1 day post ablation. The combination of cryoablation with CTLA-4 blockade was successful at preventing growth of the delayed bystander tumor. The tumor was cryoablated on day 0, and 9H10 regimen administered was 200 µg on day 1 followed by 100 µg each on days 4, 7 and 10 [8].

Mid Dose 9H10 Experiment: Although this delayed tumor challenge model was not aggressive enough for evaluating the PV-10 plus 9H10 combination, the 9H10 dosing regimen was adapted for use in a synchronous B16-F10 bilateral tumor model. The mid dose experiment was identical to the low dose experiment except that intralesional saline or PV-10 was given 8 days after bilateral tumor induction (treatment day 0), and for 9H10 treatment groups 200 µg of 9H10 was administered IP on day 0, and 100 µg each on days 3, 7 and 10. The treatment day selection was necessary to confirm existence of a substantial tumor on both flanks prior to treatment. Differences in tumor burden at day 0 (shown below) were not significant.

Treatment	Mean Total Tumor Burden (mm^2)	Standard Deviation
IL-PV-10	48.7	12.5
IL-PV-10 + 9H10	53.7	14.0
IL-Saline	53.1	26.8
IL-Saline + 9H10	48.3	17.8

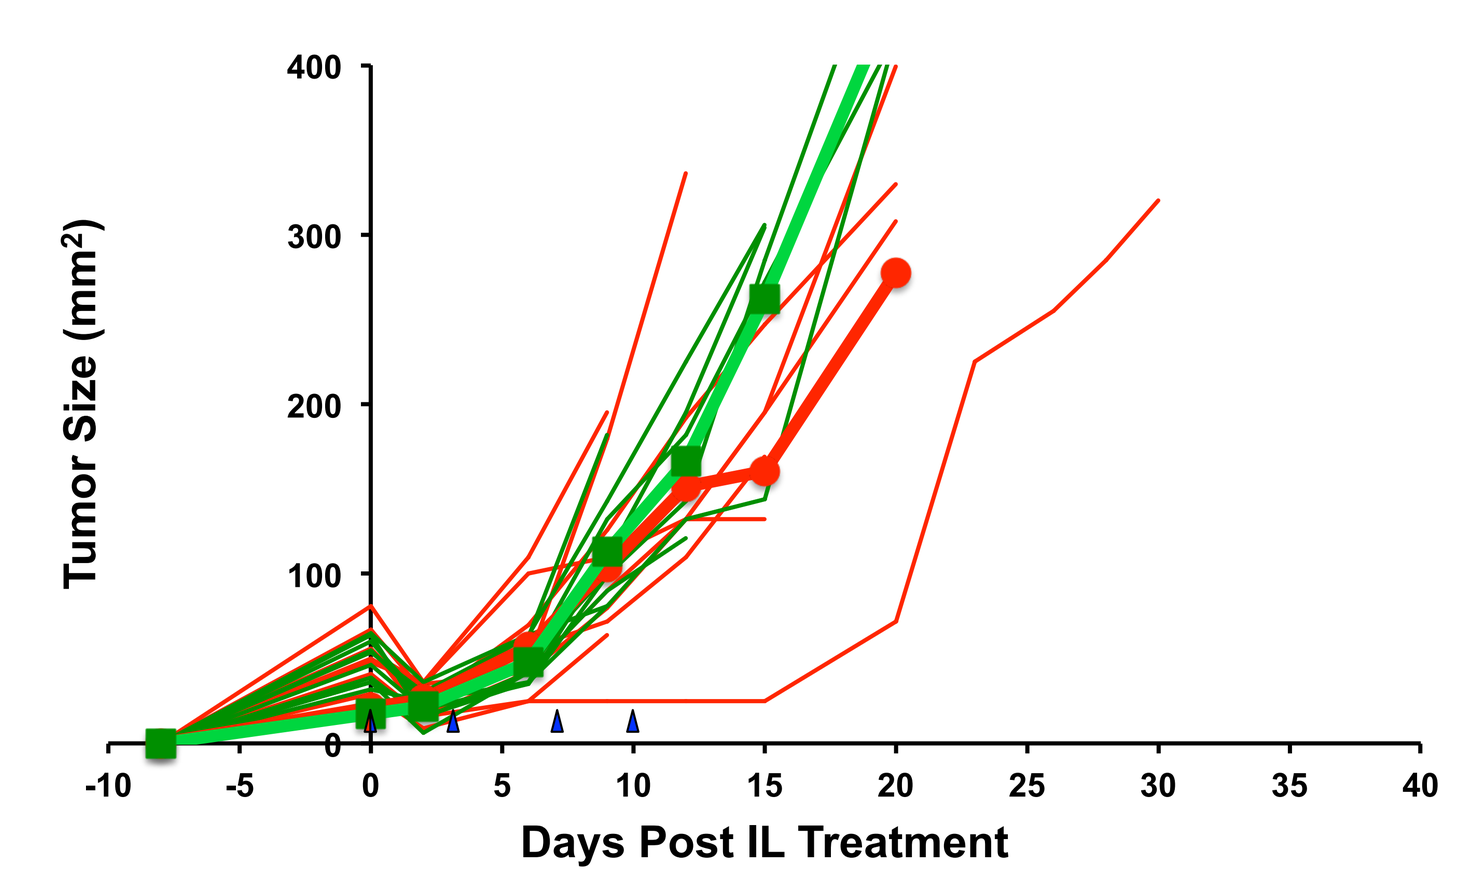
IL PV-10 vs IL PV-10 + IP 9H10
Total Tumor Burden
Tumor Size (mm²)
Days Post IL Treatment

Total Tumor Burden: A representation of PV-10 alone (green) and the combination of PV-10 + 9H10 (red) total tumor burden is shown above. The blue triangles represent 9H10 doses (PV-10 or saline injected IL only on day 0). Once again both these groups are significantly smaller than total tumor burden in the non-ablated saline and 9H10 controls. The PV-10 + 9H10 and PV-10 alone groups aren't statistically different from one another in total tumor burden.

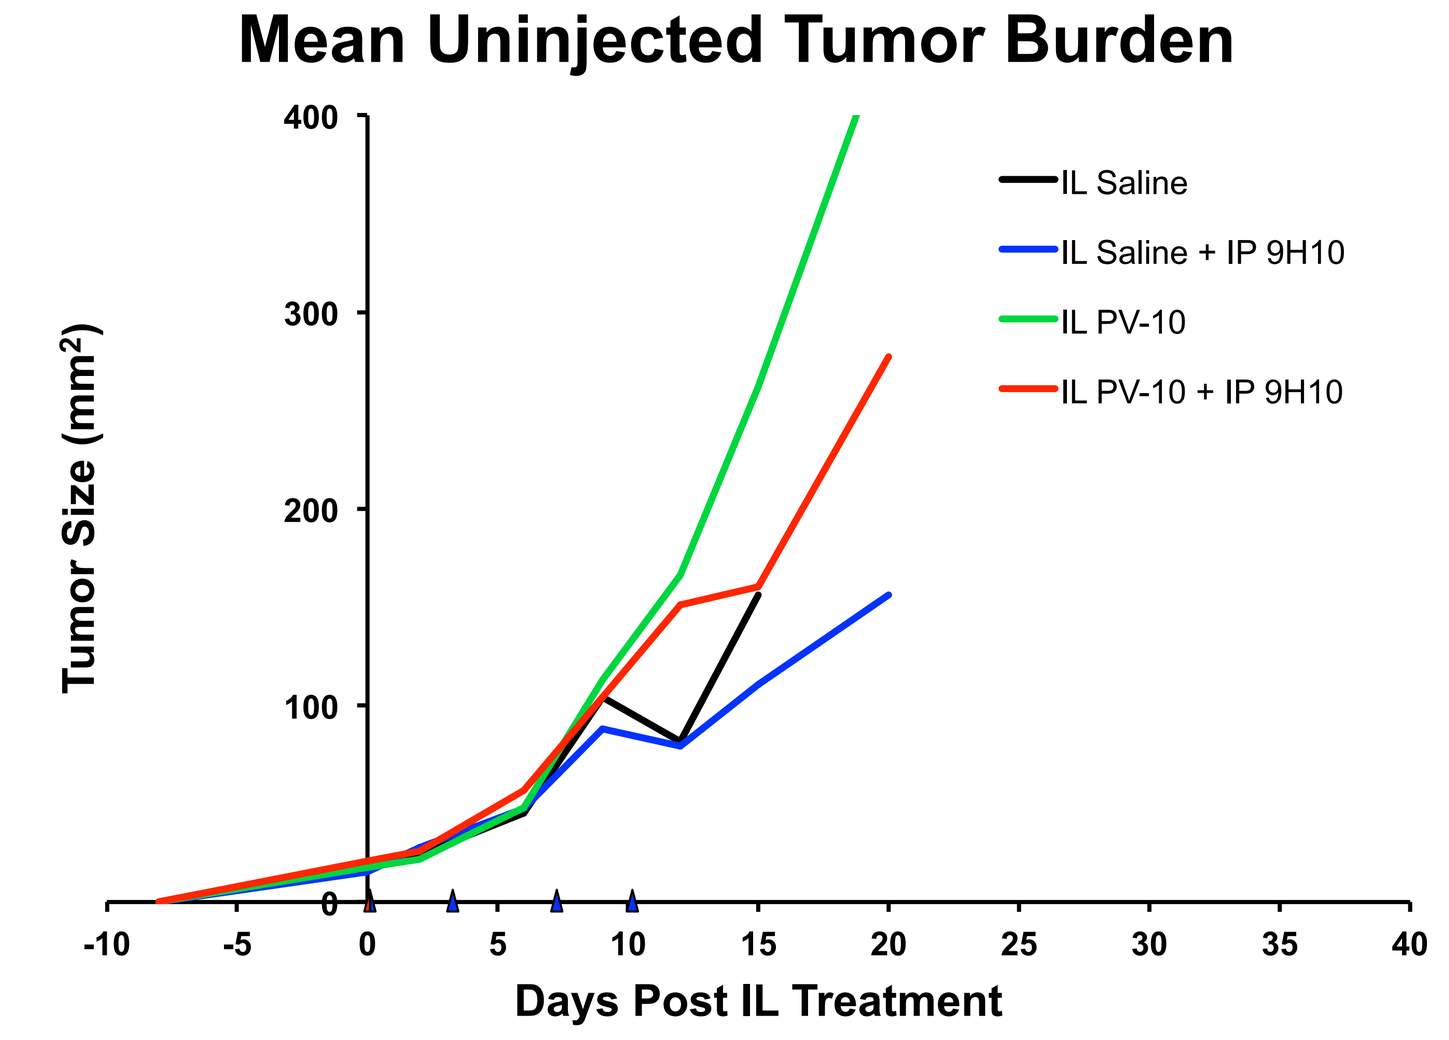
Mean Uninjected Tumor Burden
Tumor Size (mm²)
IL Saline
IL Saline + IP 9H10
IL PV-10
IL PV-10 + IP 9H10
Days Post IL Treatment

Modeled Visceral Disease (Uninjected Flank Tumors): A representation of all groups in the experiment, including 9H10 (blue) and saline (black) controls, is shown above. The combination of PV-10 + 9H10 (red) was no different in un-injected tumor burden than PV-10 alone (green). Perhaps due to the large size of initial tumors relative to the other experiments, this experiment showed no group statistical difference or trend in untreated tumor burden.

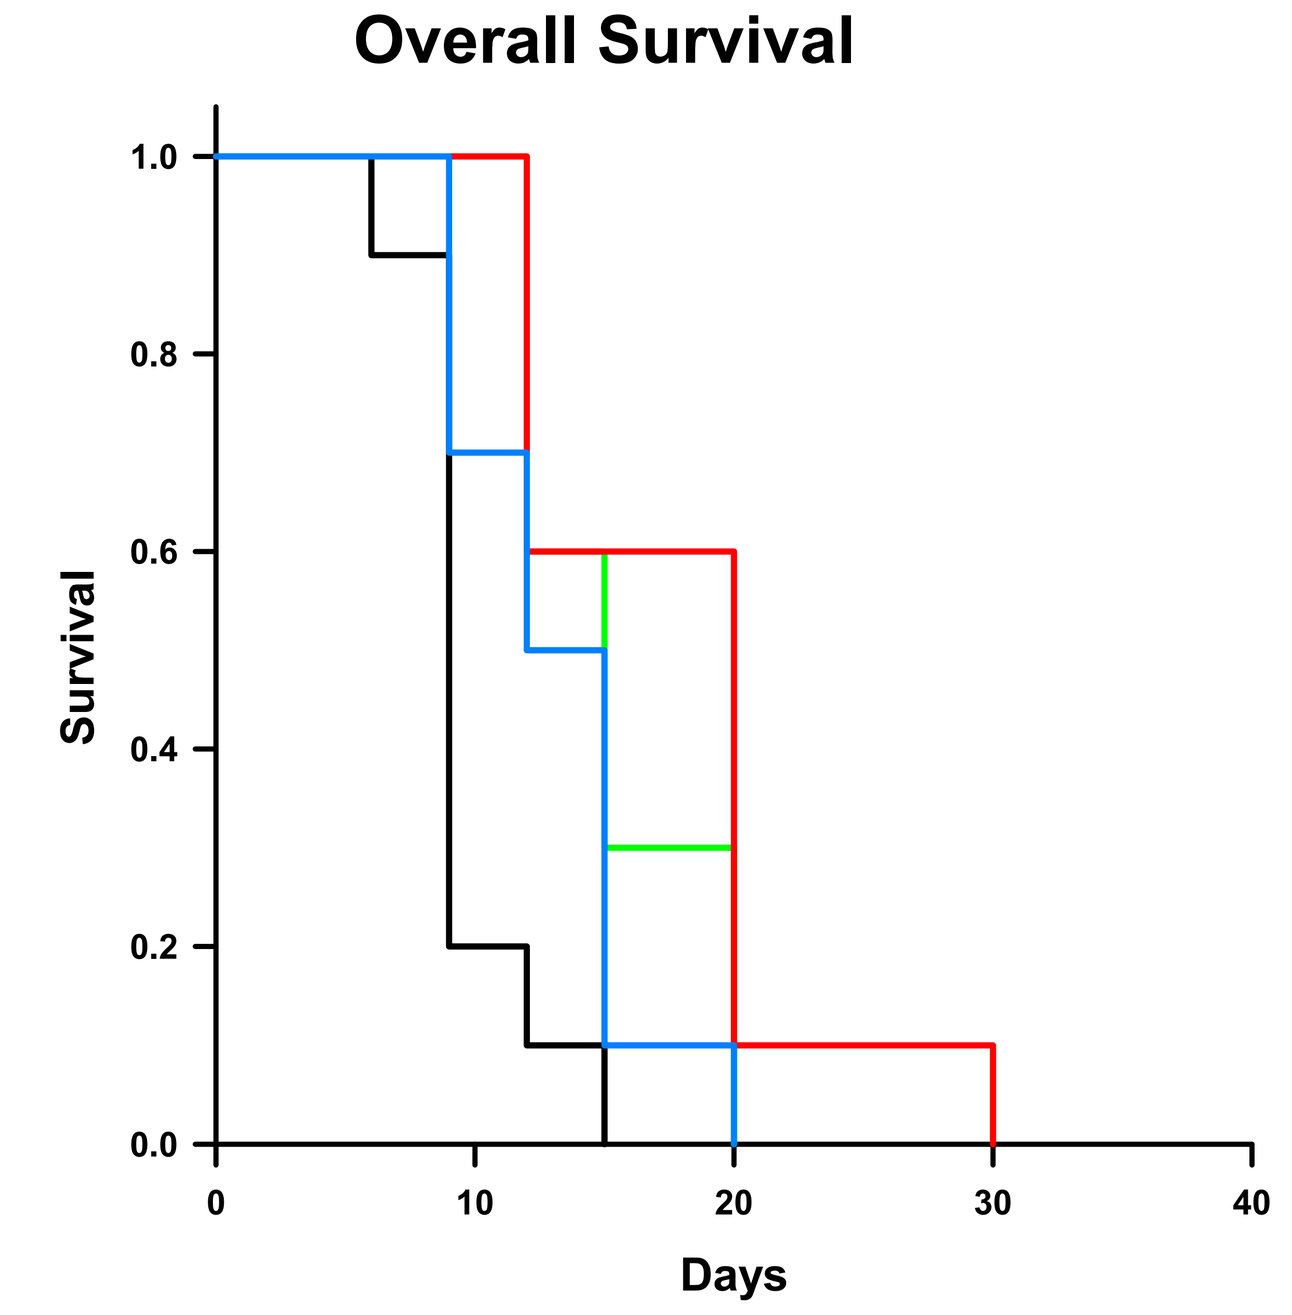
Overall Survival
Survival
Days

Overall Survival: The survival of all groups in the mid dose experiment is represented above. Events included animals euthanized for excessive tumor burden. No animals were censored in this experiment. The PV-10 alone group (green) and the PV-10 + 9H10 combination (red) showed statistically significant survival over saline alone (black, p = 0.02 and 0.0002 respectively) but the PV-10 + 9H10 combination again did not reach significance against 9H10 alone (blue, p = 0.17).

Bilateral Flank Model: High Dose 9H10 (B16-F10)

High Dose 9H10 Experiment: Demaria et al. reported that local radiation in a murine model of breast cancer was only effective at delaying single flank tumor growth in established 4T1 tumors (mean diameter 5 mm) but treatment did not extend overall survival. A combination of local radiation in established solitary tumors with CTLA-4 blockade increased survival in this murine model. An aggressive 9H10 dosing regimen was given, consisting of 200 µg on days 1, 4 and 7 after local radiation. [5]

High Dose 9H10 Experiment: This dose regimen was slightly altered to give the same dose of 9H10, but at days 0, 3, 7 to coincide with IL PV-10 treatment on day 0. Otherwise this experiment was identical to both the low and mid dose experiments with the exception that the IL interventions were given 10 days after tumor cell implantation to ensure all animals had measurable bilateral tumors on treatment day 0. Differences in tumor burden at day 0 (shown below) were not significant.

Treatment	Mean Total Tumor Burden (mm^2)	Standard Deviation
IL-PV-10	38.1	16.7
IL-PV-10 + 9H10	43.7	14.4
IL-Saline	45.0	20.6
IL-Saline + 9H10	37.4	16.5

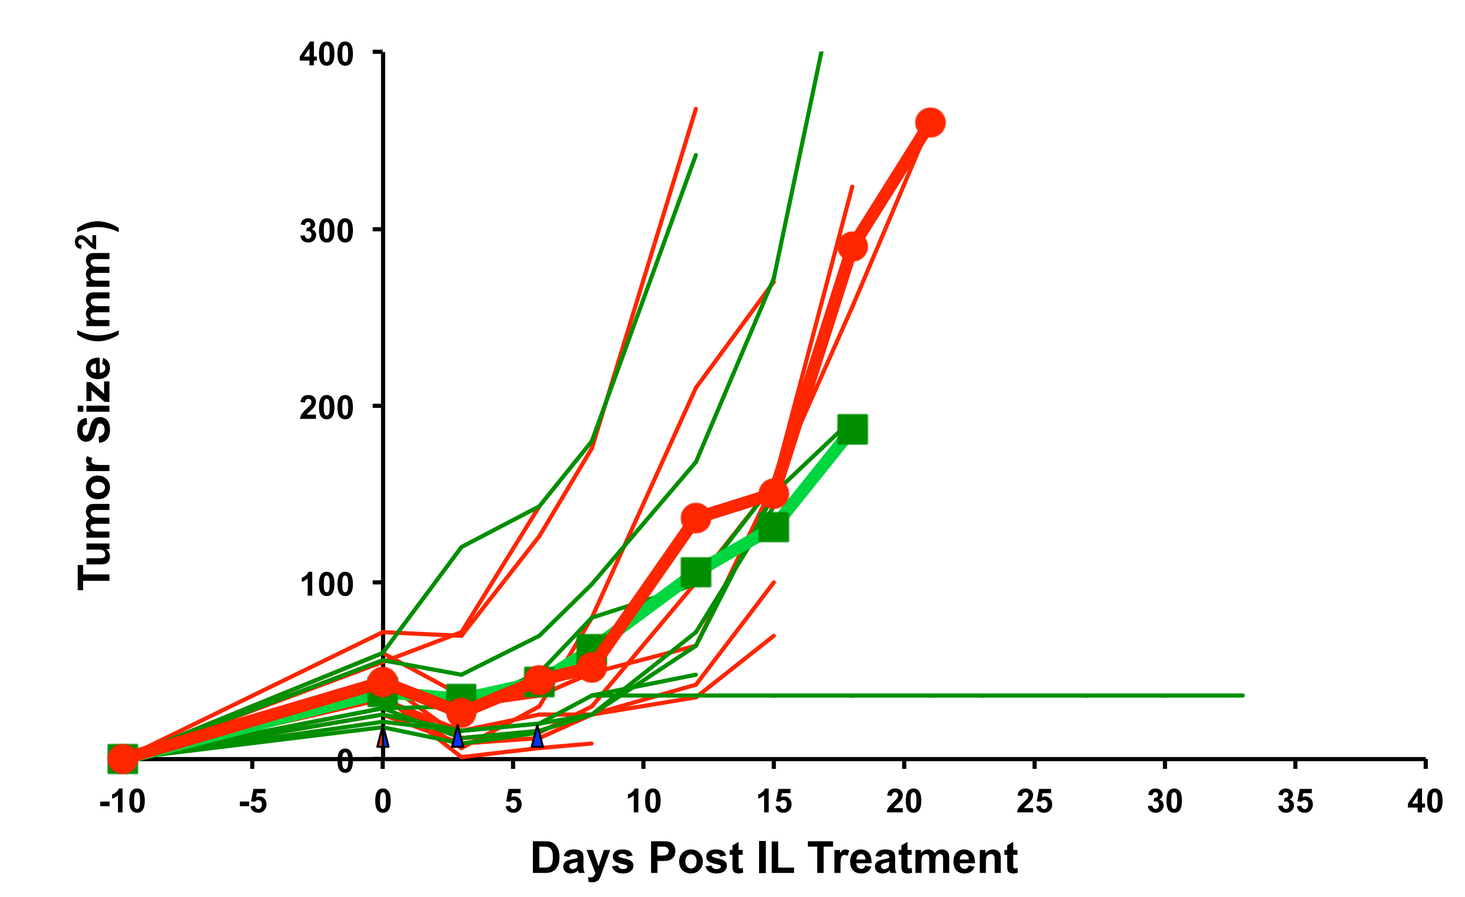
IL PV-10 vs IL PV-10 + IP 9H10
Total Tumor Burden
Tumor Size (mm²)
Days Post IL Treatment

Total Tumor Burden: A representation of PV-10 alone (green) and the combination of PV-10 + 9H10 (red) total tumor burden is shown above. The blue triangles represent 9H10 doses (PV-10 or saline injected IL only on day 0). Both groups are once again smaller than total tumor burden in the non-ablated saline and 9H10 controls. The PV-10 + 9H10 and PV-10 alone groups aren't statistically different from one another in total tumor burden. P-values for the difference between these groups and the 9H10 alone group (given in both groups at the high dose) are < 0.001 for days 3 ,6, 8 and 12.

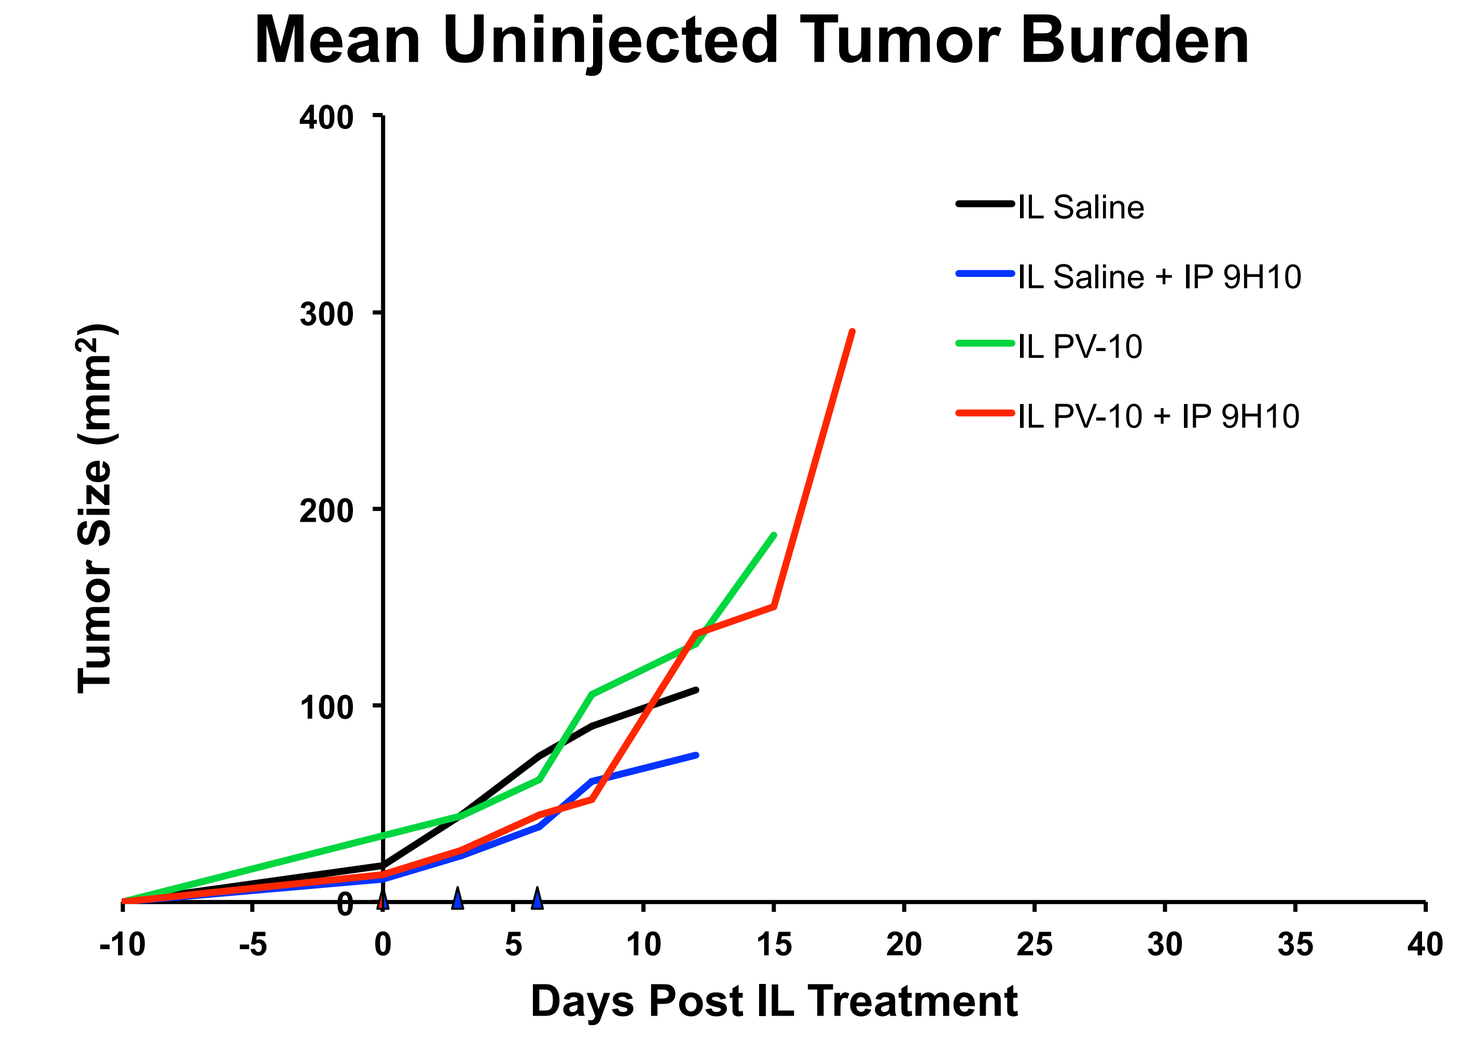
Mean Uninjected Tumor Burden
Tumor Size (mm²)
IL Saline
IL Saline + IP 9H10
IL PV-10
IL PV-10 + IP 9H10
Days Post IL Treatment

Modeled Visceral Disease (Uninjected Flank Tumors): A representation of all groups in the experiment, including 9H10 (blue) and saline (black) controls, is shown above. The combination of PV-10 + 9H10 (red) was no different in un-injected tumor burden than PV-10 alone (green). Perhaps due to the aggressive 9H10 regimen, this experiment showed no group clear difference or trend in untreated tumor burden.

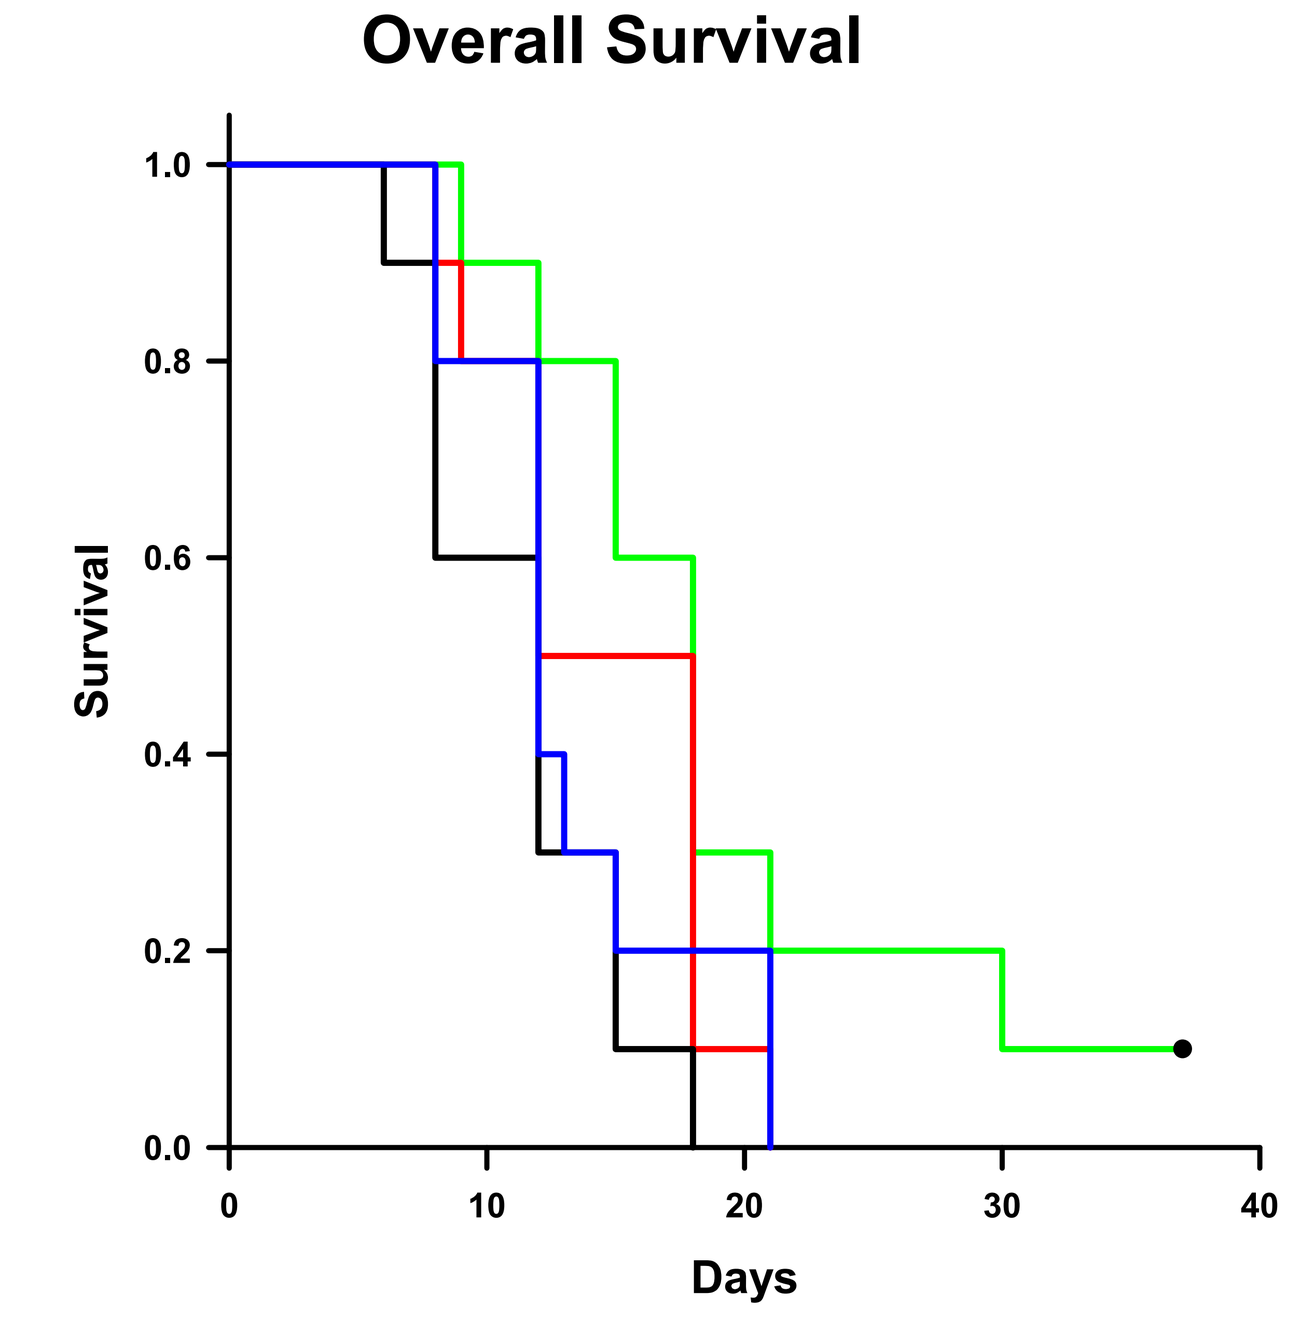
Overall Survival
Survival
Days

Overall Survival: The survival of all groups in the high dose experiment is represented above. Events included animals euthanized for excessive tumor burden. A single animal in the PV-10 alone arm was censored on day 37 (with a slowly regressing tumor on the untreated flank). The PV-10 alone group (green) showed statistically significant survival over saline (black, p = 0.02), but not against 9H10 alone (blue, p = 0.24).

Discussion

Lung Metastasis Results: These results corroborate previously reported data showing robust suppression of synchronous lung metastases upon PV-10 ablation of an established flank tumor [2-3]. While the magnitude of the effect of PV-10 alone makes it impossible to unequivocally conclude whether there is synergy in this model combination, absence of apparent increased toxicity coupled with suppression of lung metastases indicates minimal likelihood of interference between the component therapies.

Comparison of Bilateral Flank Model Results: Despite delay in initiation of therapy until onset of palpable tumors in a very aggressive melanoma model, PV-10 and PV-10 + 9H10 exhibited robust response in both injected and uninjected tumors. As expected, PV-10 + 9H10 combination therapy exhibited maximum benefit. This was most evident in the low dose model, where advantages were pronounced in both tumor growth and survival data. Overlay of curves for the respective combination groups shows that survival in the low dose group (red) was significantly longer than that of the high dose group (blue, p = 0.03).

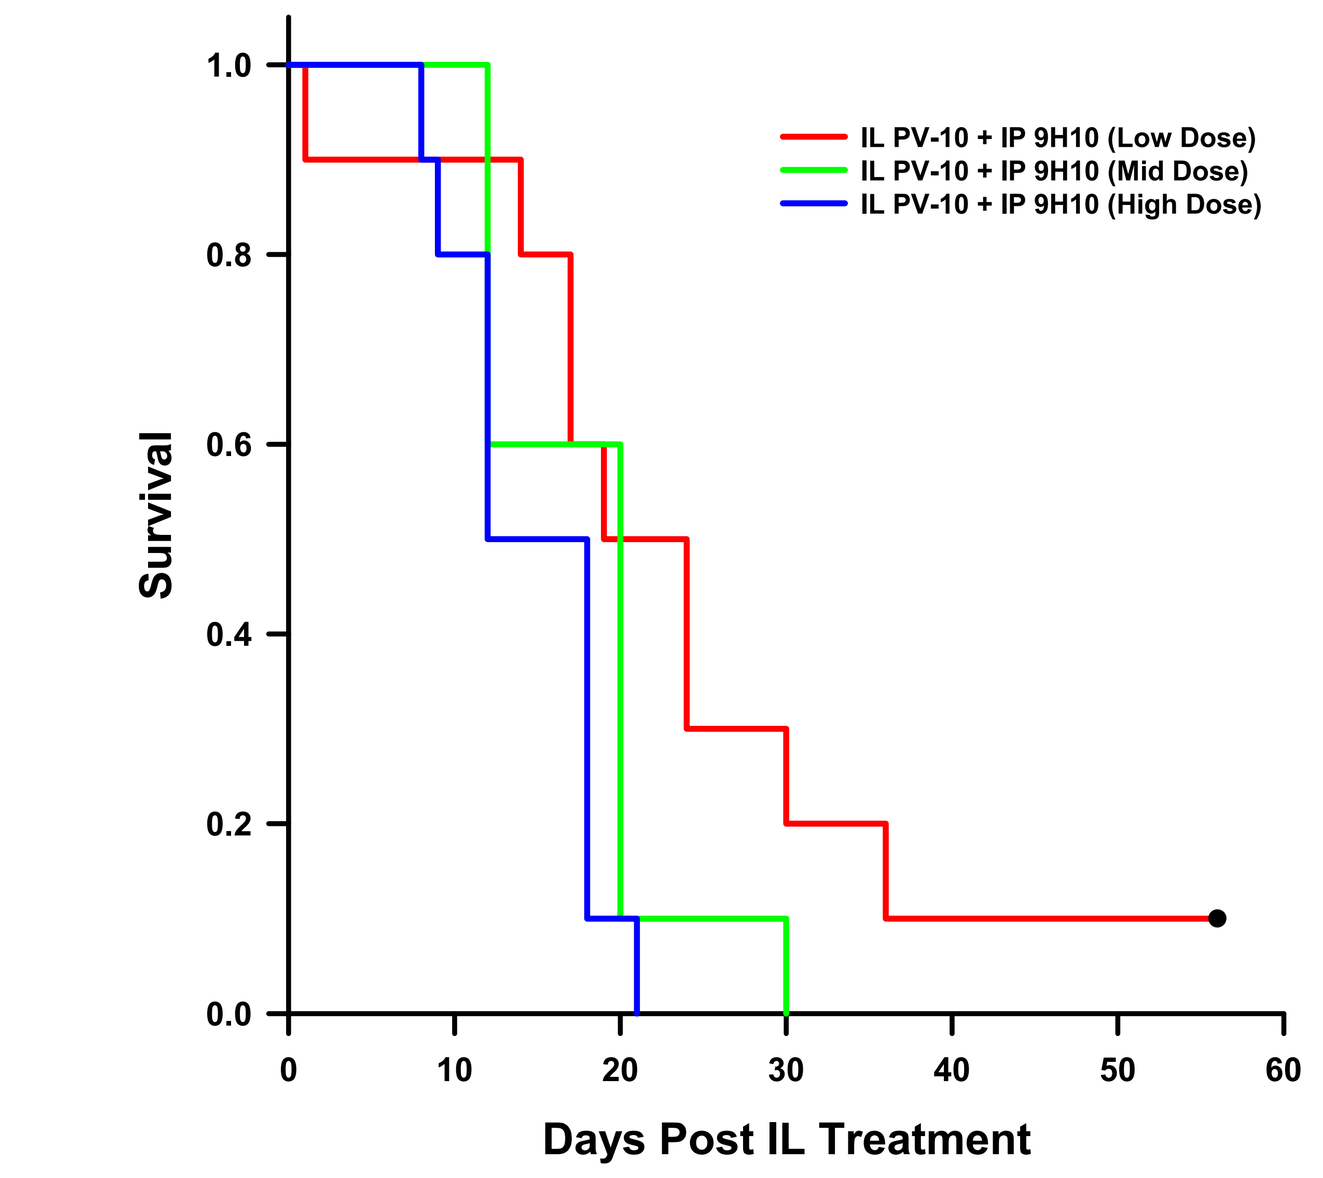
IL PV-10 + IP 9H10 (Low Dose)
IL PV-10 + IP 9H10 (Mid Dose)
IL PV-10 + IP 9H10 (High Dose)
Survival
Days Post IL Treatment

In general, the combination of PV-10 + 9H10 was favored, while PV-10 yielded increased survival over saline in each case. At the high dose level of 9H10, PV-10 proved to be more effective than the combination.

Mean Survival	Low Dose	Mid Dose	High Dose
PV-10	16.4	15.3	19.2
PV-10 + 9H10	21.8	17.8	14.6
Saline	15.3	9.6	11.4
Saline + 9H10	20.8	13.1	13.4

Median Survival	Low Dose	Mid Dose	High Dose
PV-10	17.0	15.0	18.0
PV-10 + 9H10	21.5	20.0	15.0
Saline	15.5	9.0	12.0
Saline + 9H10	22.0	13.5	12.0

Variability in response across dose levels may be due to toxicity of 9H10 at elevated doses, with additional differences arising from the effect of tumor burden. In these experiments, uninjected tumor burden and dose of 9H10 varied between studies, and the impact of this is illustrated below (similar results were noted when data were plotted against total tumor burden). Saline alone (black) and PV-10 alone (green) exhibited a linear relationship between tumor burden and survival whereas 9H10 (blue) had a highly nonlinear response that featured a marked reduction in survival at the high dose. Combination of PV-10 with 9H10 (red) dramatically improved survival at the mid dose despite higher tumor burden. This was repeated at the high dose, although apparent toxicity of 9H10 in these groups negated much of the advantage of the combination.

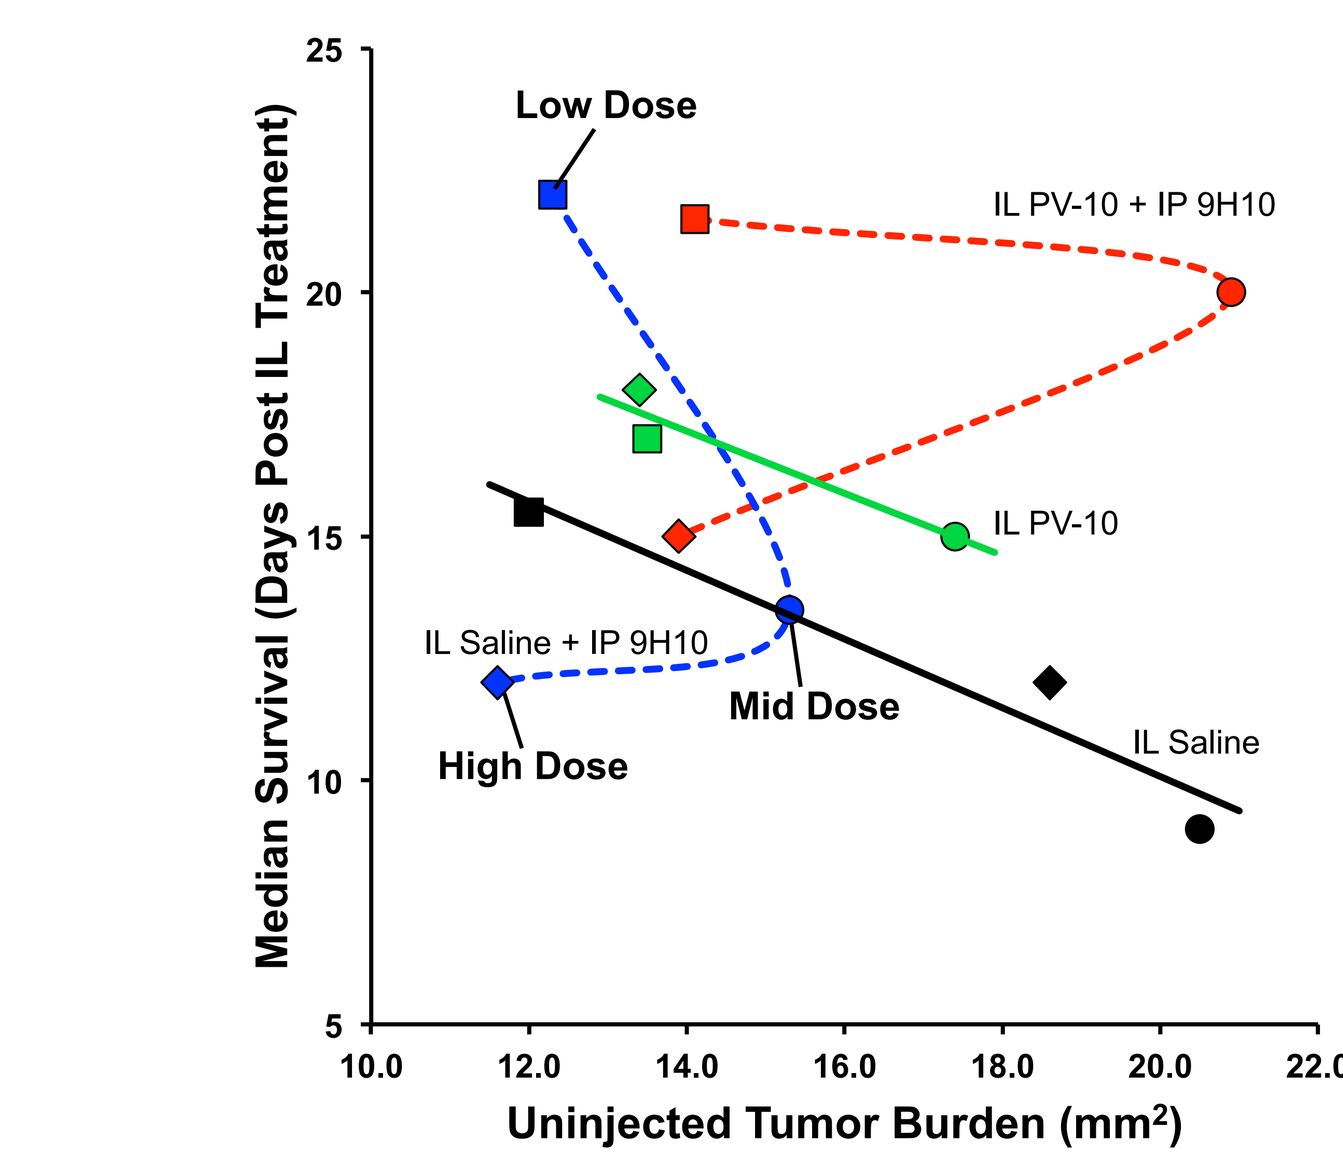
Low Dose
IL PV-10 + IP 9H10
IL PV-10
IL Saline + IP 9H10
Mid Dose
High Dose
IL Saline
Median Survival (Days Post IL Treatment)
Uninjected Tumor Burden (mm²)

Conclusions

Examples of successful treatment of established B16 tumors in murine models are rare. As a single agent, a single dose of IL PV-10 can be highly effective in reducing overall tumor burden in both murine models and clinical trial patients. However, when substantial inaccessible tumor burden is present, single injection may be insufficient to rapidly control uninjectable disease. It is in these cases that systemic therapy in combination with PV-10 may be indicated.

Overall, these results demonstrate:

- PV-10 is highly effective when all existing tumor is accessible for injection, providing rapid reduction in tumor burden (both in animal models and clinically in cancer patients)
- Tumor ablation with PV-10 induces tumor-specific immunity
- Rapid, permanent local ablation in combination with induction of tumor-specific immunity afforded by PV-10 comprises a uniquely powerful approach that may be complementary to many systemic therapies, while its distinctive adverse effect profile and pharmacology minimizes potential for drug interaction
- For visceral or other inaccessible disease, the combination of PV-10 with CTLA-4 blockade has important potential for synergy
- Apparent toxicity of high dose 9H10 markedly reduced the effect of both 9H10 alone and the combination of PV-10 + 9H10, although the combination remained more effective than 9H10 alone at the high dose
- Further studies are underway to confirm the apparent synergy of PV-10 with CTLA-4 blockade
- A phase 1/2 anti-CTLA-4 dose escalation trial with PV-10 is warranted

References

1. Wachter et al. Functional imaging of photosensitizers using multiphoton microscopy. SPIE 2002; 4630:143-7.
2. Pilon-Thomas et al. Intralesional injection with PV-10 induces a systemic anti-tumor immune response in murine models of breast cancer and melanoma. 2013 AACR Poster 1248.
3. Dees et al. Generation of an antitumor response and immunity using a small molecule drug (PV-10). SITC Meeting 26-28 October 2012, Poster 1452582.
4. Waitz et al. CTLA-4 blockade synergizes with cryoablation to mediate tumor rejection. Oncoimmunology 2012; 1: 544-546.
5. Demaria et al. Immune-mediated inhibition of metastases after treatment with local radiation and CTLA-4 blockade in mouse model of breast cancer. Clin Cancer Res 2005; 11: 728-734.
6. den Brok et al. Efficient loading of dendritic cells following cryo and radiofrequency ablation in combination with immune modulation induces anti-tumor immunity. Br. J. of Cancer 2006; 95: 896-905.
7. van Elsas et al. Combination immuno-therapy of B16 melanoma using anti-cytotoxic T lymphocyte- associated antigen 4 (CTLA-4) and granulocyte/macrophage colony-stimulating factor (GM-CSF)-producing vaccines induces rejection of subcutaneous and metastatic tumors accompanied by autoimmune depigmentation. J Exp Med 1999; 190: 355–366.
8. Waitz et al. Potent induction of tumor immunity by combining tumor cryoablation with anti-CTLA-4 therapy. Cancer Research 2012; 72: 430-439.