Trials in Progress: Intralesional PV-10 vs Systemic Chemotherapy for Treatment of Locally Advanced Cutaneous Melanoma

John F. Thompson[1], Sanjiv S. Agarwala[2], B. Mark Smithers[3], Merrick I. Ross[4], Charles R. Scoggins[5], Brendon J. Coventry[6], Susan J. Neuhaus[6], David R. Minor[7], and Eric A. Wachter[8]

Melanoma Institute Australia and the University of Sydney, Sydney, NSW AUS[1], St. Luke's Hospital and Health Network and Temple University, Bethlehem, PA USA[2], Queensland Melanoma Project, Princess Alexandra Hospital, University of Queensland, Woolloongabba, QLD AUS[3], M.D. Anderson Cancer Center, Houston, TX USA[4], University of Louisville, Louisville, KY USA[5], University of Adelaide and Royal Adelaide Hospital, Adelaide, SA AUS[6], California Pacific Medical Center, San Francisco, CA USA[7], and Provectus Biopharmaceuticals, Inc., Knoxville, TN USA[8]

HemOnc Today Melanoma and Cutaneous Malignancies 2015
New York City 10-11 April 2015

For additional information:
conference@pvct.com

Abstract

Study PV-10-MM-31 is an international multicenter, open-label, pivotal randomized controlled trial (RCT) of single-agent intralesional (IL) PV-10 versus systemic chemotherapy with dacarbazine (DTIC) or temozolomide (TMZ) in patients with locally advanced cutaneous melanoma (ClinicalTrials.gov identifier NCT02288897). The primary endpoint of progression free survival (PFS) will be assessed according to RECIST 1.1 (based on blinded independent review of study photography and radiology data by an Independent Review Committee (IRC) to compare outcome in the study arms). A total of 225 eligible subjects are being randomized in a 2:1 ratio to the two treatment arms. Comprehensive disease assessments, including review of clinical photography of lesions and radiologic assessment for the presence of visceral and nodal metastases, are performed at screening and every 12 weeks thereafter, and at final follow-up by CT imaging. Clinical assessment of progression status will be performed at 28-day intervals during the treatment phase of the study. Patients must have locally advanced cutaneous melanoma (i.e., Stage IIIB or Stage IIIC recurrent, satellite or in-transit cutaneous or subcutaneous melanoma) with at least 1 Target Lesion with longest diameter >10 mm; no lesion > 30 mm in longest diameter; and no more than 20 total lesions at enrollment. Patients must have failed at least one immune checkpoint inhibitor or not otherwise be candidates for immune checkpoint inhibitors and must be BRAF wild-type.

Background

Primary Ablation and Secondary Tumor-Specific Immune Response

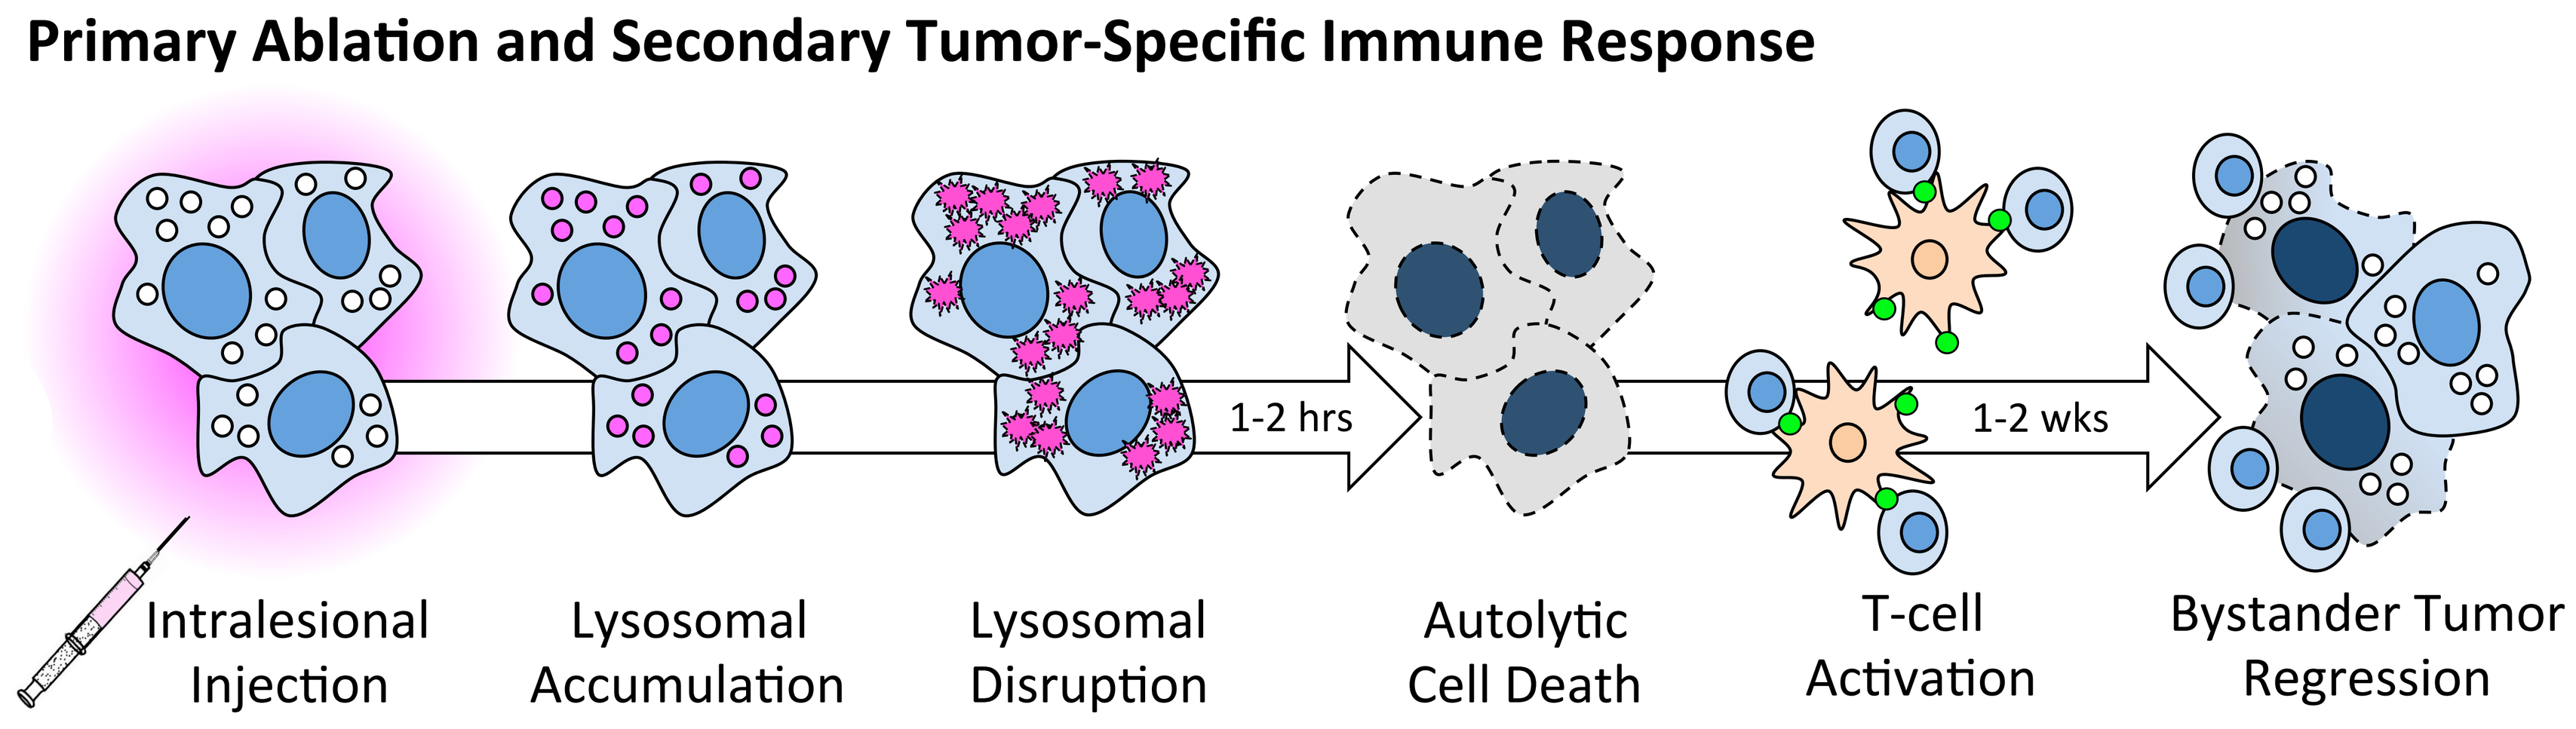
1-2 hrs
1-2 wks
Intralesional Injection
Lysosomal Accumulation
Lysosomal Disruption
Autolytic Cell Death
T-cell Activation
Bystander Tumor Regression

- Wachter et al., SPIE 4630, 147, 2002 (lysosomal accumulation and rupture in tissue culture)
- Thompson et al., Mel Res 18, 405, 2008 (ablation of injected tumors and bystander regression in recurrent patients)
- Toomey et al., PLoS One 8, e68561, 2013 (tumor-specific immune response in mice)
- Liu et al., AACR 2014 Abstract 0630 (DC recruitment/activation and T-cell activation in mice)
- Sarnaik et al., ASCO 2014 Abstract 9028 (pCR and T-cell activation in refractory patients)

Phase 2 Experience

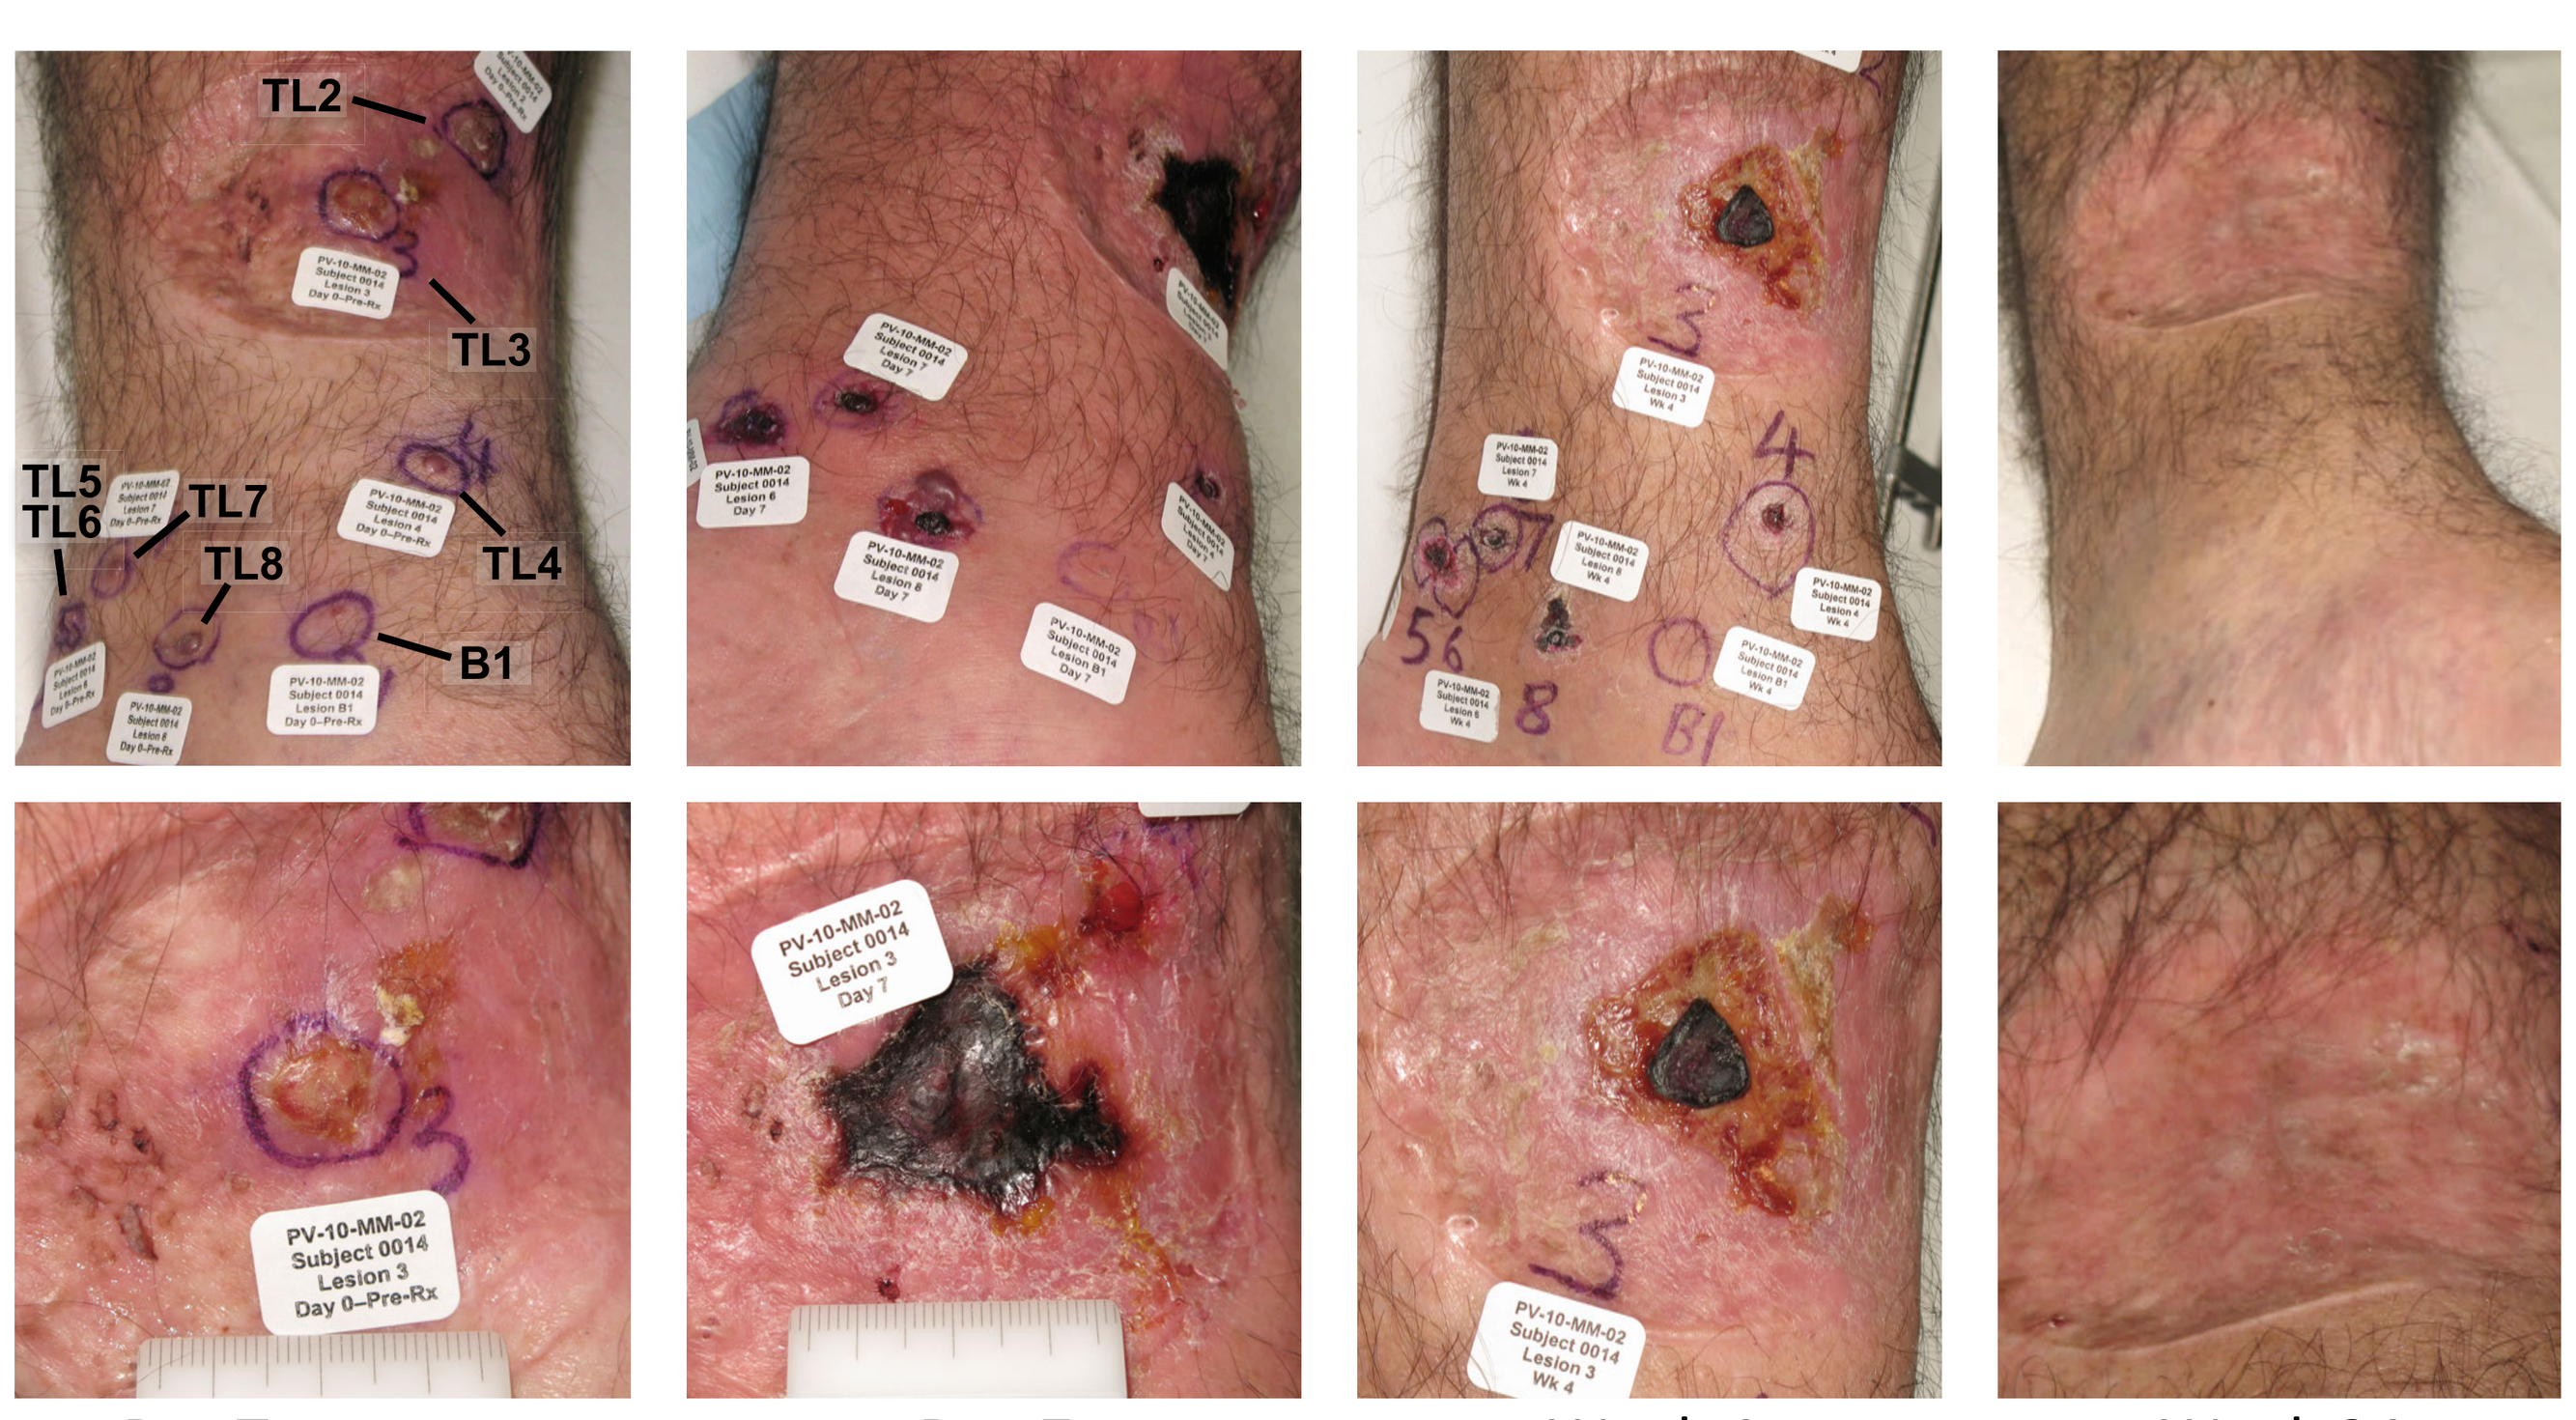
TL2
TL3
TL5
TL6
TL7
TL8
TL4
B1
PV-10-MM-02 Subject 0014 Lesion 3 Day 0–Pre-Rx
PV-10-MM-02 Subject 0014 Lesion 3 Day 7
PV-10-MM-02 Subject 0014 Lesion 3 Wk 4

Pre-Treatment | Day 7 | Week 4 | Week 24

Response of Target Lesions	ITT Population		All Lesions Treated		Bystanders Untreated	
N (Patients)	80	%	28	%	26	%
CR	21	26%	14	50%	6	23%
PR	20	25%	6	21%	8	31%
SD	14	18%	3	11%	8	31%
PD (PD + NEV)[a]	25	31%	5	18%	4	15%
CR + PR	41	51%	20	71%[b]	14	54%
CR + PR + SD (Locoregional Disease Control)	55	69%	23	82%	22	85%
Mean PFS (months)[c]	8.4		9.8[d]		9.6[e]	

[a] Patients that were non-evaluable due to progression prior to week 8 were tracked separately but combined with PD for tabulation of outcome.
[b] P = 0.006 vs. TNTC or Stage IV subgroup, BORR by Chi-square test.
[c] PFS by mRECIST, maximum follow-up duration 12 months.
[d] P = 0.001 vs. TNTC or Stage IV subgroup, by log-rank test.
[e] P = 0.01 vs. TNTC or Stage IV subgroup, by log-rank test.

System Organ Class[a] / Preferred Term[a]	Adverse Events[b,c] (ITT Population, N = 80) CTCAE Grade 1	2	3	Total	%
General Disorders and Administration Site Conditions					
Injection Site Pain	29	25	10	64	80%
Injection Site Edema	19	14	0	33	41%
Injection Site Vesicles	17	13	1	31	39%
Injection Site Discoloration[d]	13	12	0	25	31%
Injection Site Swelling	14	7	1	22	28%
Injection Site Pruritus	14	3	0	17	21%
Injection Site Erythema	6	4	1	11	14%
Injection Site Infection	3	2	1	6	8%
Peripheral Edema	3	0	1	4	5%
Injection Site Cellulitis	0	2	1	3	4%
Gastrointestinal Disorders					
Dysphagia	0	0	1	1	1%
Nervous System Disorders					
Headache	11	2	0	13	16%
Skin and Subcutaneous Tissue Disorders					
Photosensitivity Reaction	0	0	1	1	1%

[a] System Organ Class and Preferred Term are based on the MedDRA® version 13.0 terminology dictionary. Locoregional adverse events were coded to "injection site" Preferred Terms to differentiate these from systemic events.
[b] Includes all AEs with an incidence of 10% or higher and all CTCAE Grade 3 and higher AEs; there were no treatment-related Grade 4 or 5 AEs reported.
[c] If a patient experienced an AE more than once during the study the greatest severity is presented.
[d] Discoloration locoregional to injected lesions.

- Thompson et al., Ann Surg Oncol 2014 (Phase 2 study of intralesional PV-10 in refractory metastatic melanoma)

Why Address Locoregional Disease with a Focal Treatment?

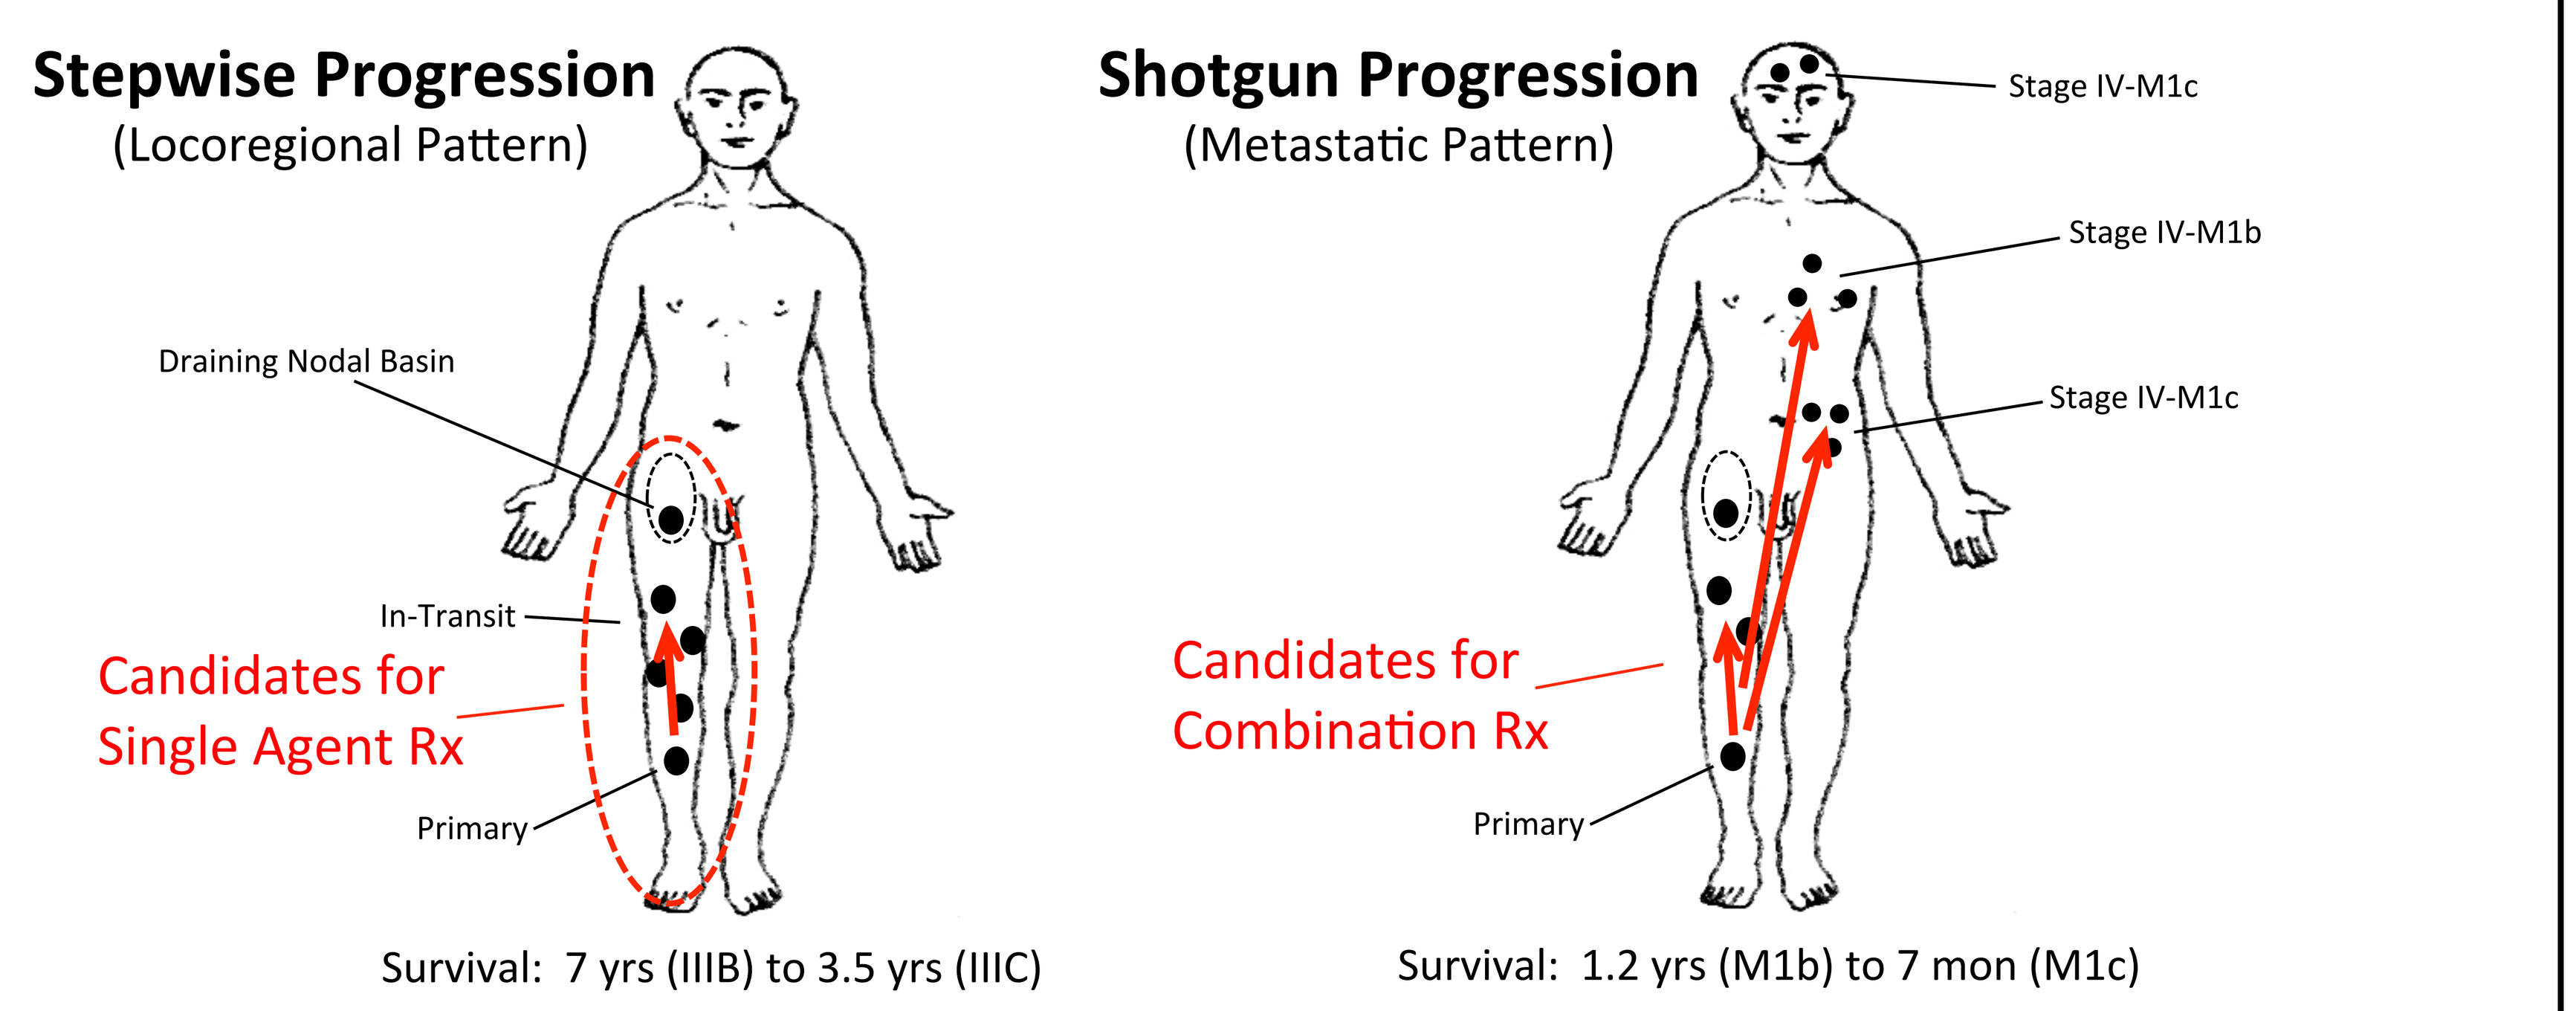
Stepwise Progression
(Locoregional Pattern)
Draining Nodal Basin
In-Transit
Candidates for Single Agent Rx
Primary
Survival: 7 yrs (IIIB) to 3.5 yrs (IIIC)
Shotgun Progression
(Metastatic Pattern)
Stage IV-M1c
Stage IV-M1b
Stage IV-M1c
Candidates for Combination Rx
Primary
Survival: 1.2 yrs (M1b) to 7 mon (M1c)

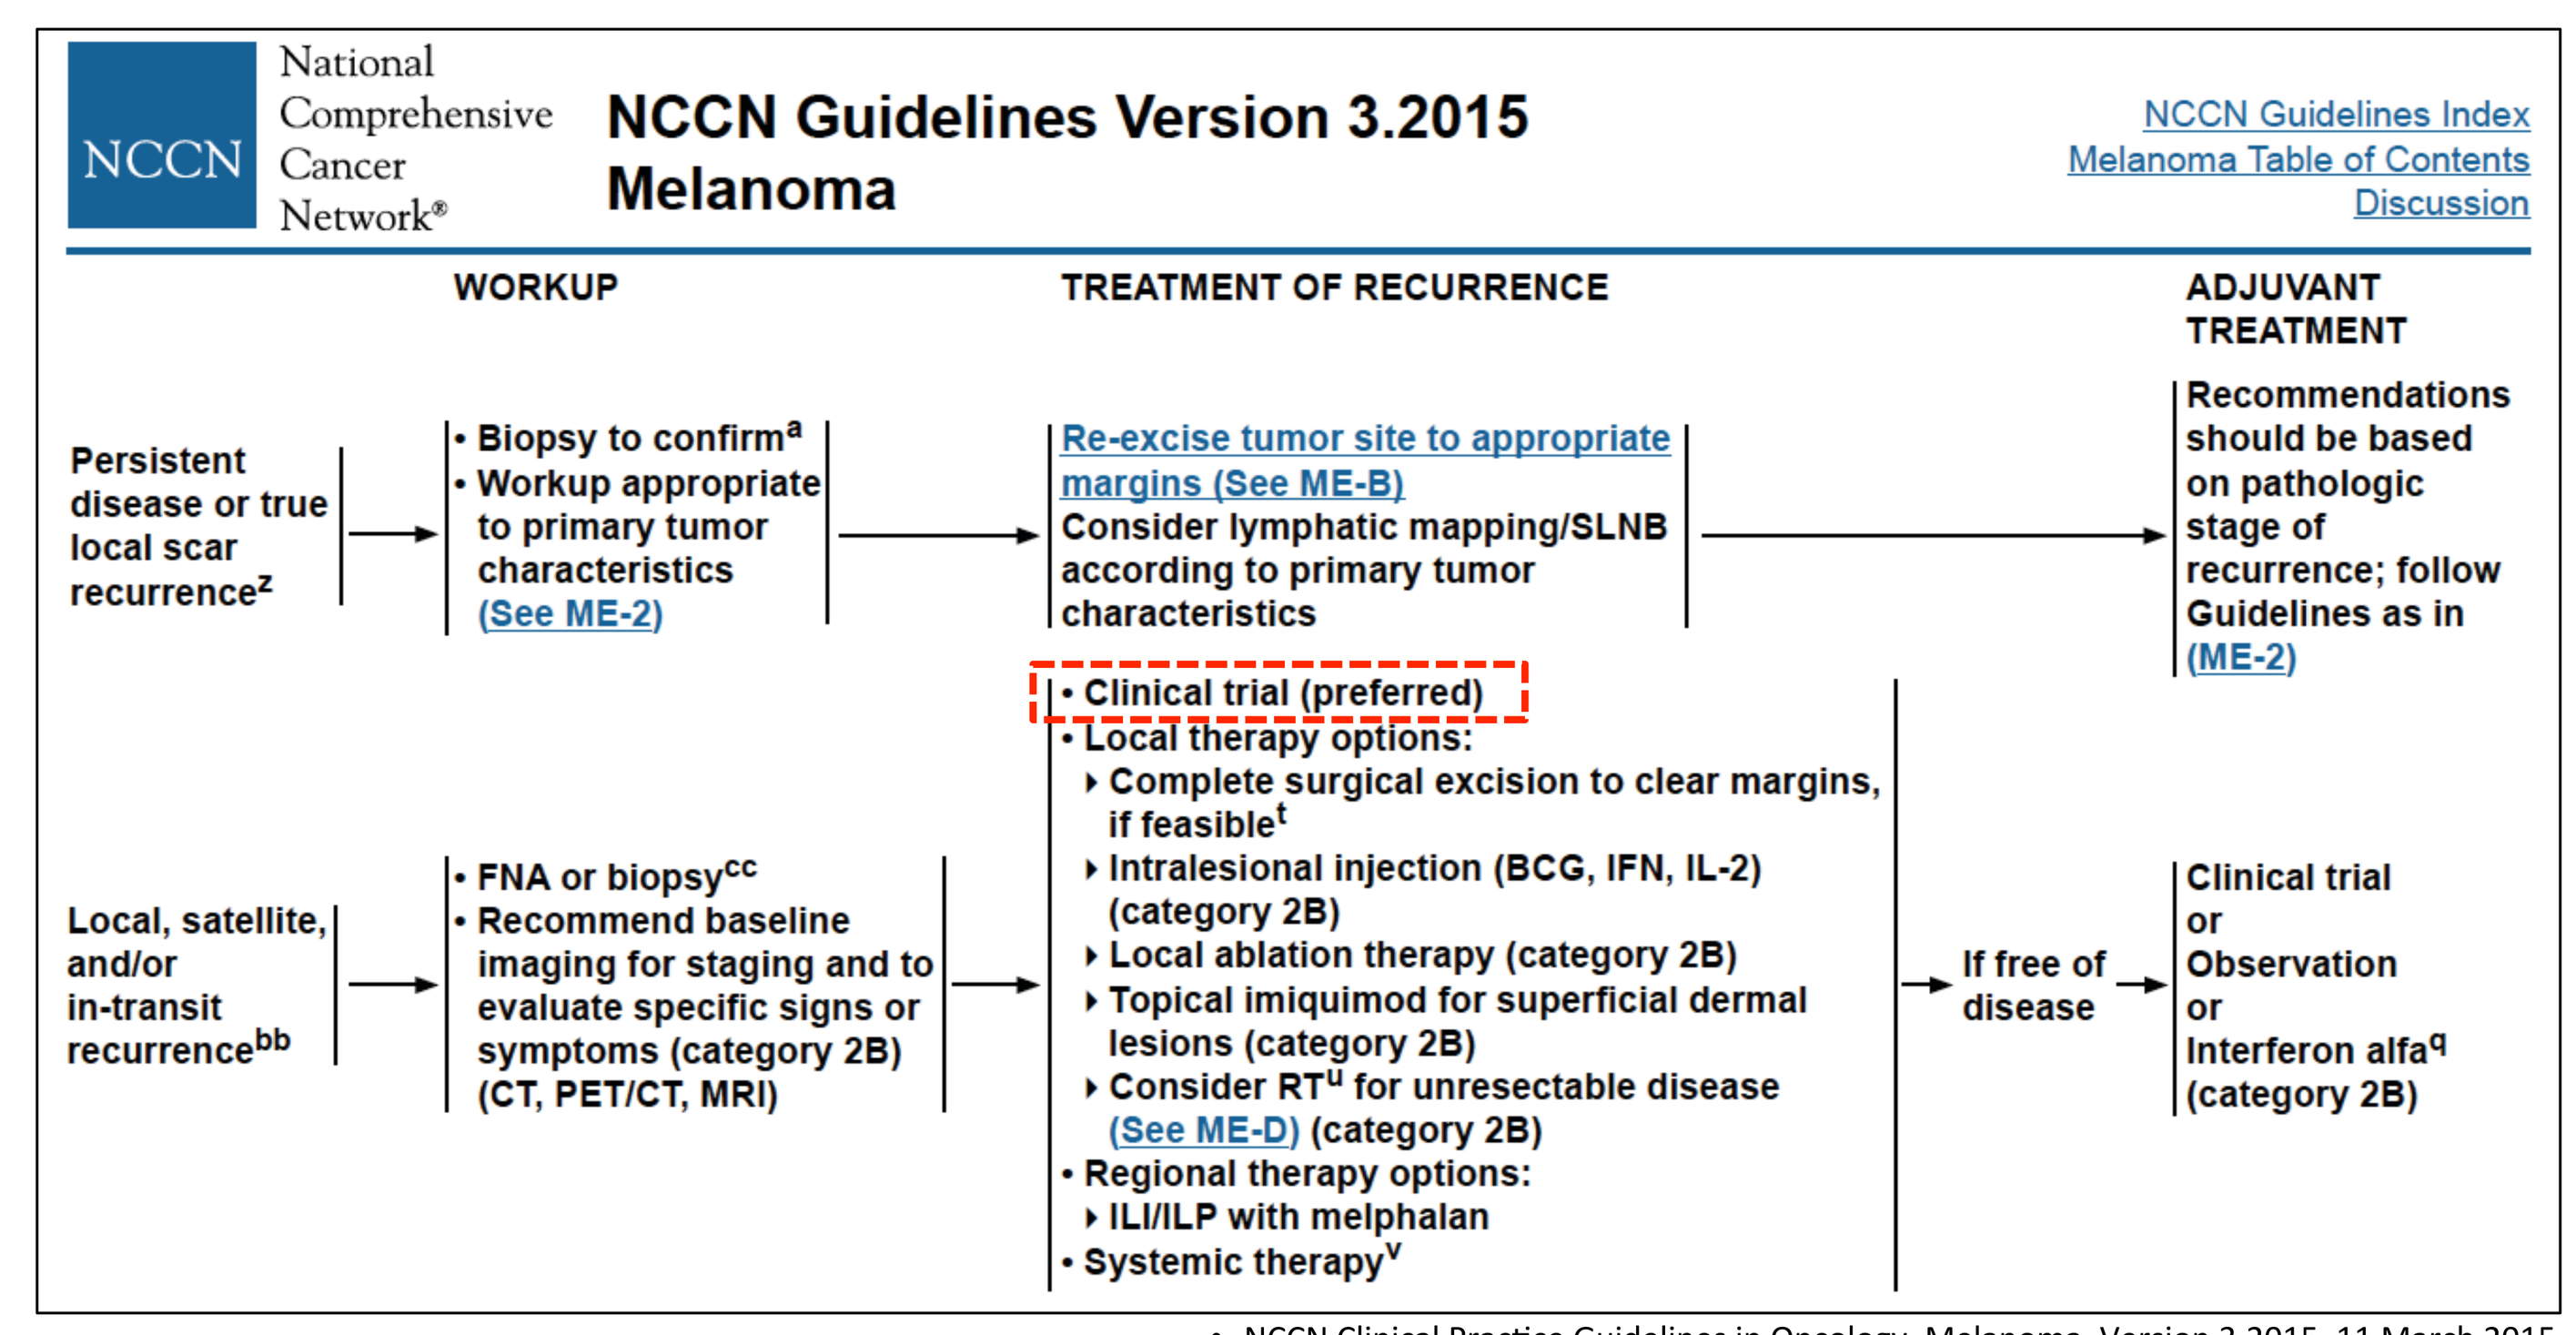
NCCN National Comprehensive Cancer Network®
NCCN Guidelines Version 3.2015
Melanoma
NCCN Guidelines Index
Melanoma Table of Contents
Discussion
WORKUP
TREATMENT OF RECURRENCE
ADJUVANT TREATMENT
Persistent disease or true local scar recurrence[z]
• Biopsy to confirm[a]
• Workup appropriate to primary tumor characteristics (See ME-2)
Re-excise tumor site to appropriate margins (See ME-B)
Consider lymphatic mapping/SLNB according to primary tumor characteristics
Recommendations should be based on pathologic stage of recurrence; follow Guidelines as in (ME-2)
• Clinical trial (preferred)
• Local therapy options:
▸ Complete surgical excision to clear margins, if feasible[t]
▸ Intralesional injection (BCG, IFN, IL-2) (category 2B)
▸ Local ablation therapy (category 2B)
▸ Topical imiquimod for superficial dermal lesions (category 2B)
▸ Consider RT[u] for unresectable disease (See ME-D) (category 2B)
• Regional therapy options:
▸ ILI/ILP with melphalan
• Systemic therapy[v]
Local, satellite, and/or in-transit recurrence[bb]
• FNA or biopsy[cc]
• Recommend baseline imaging for staging and to evaluate specific signs or symptoms (category 2B) (CT, PET/CT, MRI)
If free of disease
Clinical trial or Observation or Interferon alfa[q] (category 2B)

- NCCN Clinical Practice Guidelines in Oncology, Melanoma, Version 3.2015, 11 March 2015

Demographics and Response Data for Recent Melanoma Immunotherapy Studies

Study	N	Stage III	CR
Ipilimumab plus dacarbazine[a]	250	2.4%	1.6%
Ipilimumab + gp100 and ipilimumab alone[b]	540[b]	1.1%	0.6%
Pembrolizumab in ipilimumab refractory patients[c]	173	1%	1%
Nivolumab in previously untreated melanoma[d]	210	NR[d]	7.6%
Nivolumab plus ipilimumab in advanced melanoma[e]	53	NR[e]	9.6%
PV-10 phase 2[f]	80	77.5%	26%

[a] Robert et al. Ipilimumab plus dacarbazine for previously untreated metastatic melanoma. NEJM 2011.
[b] Hodi et al. Improved survival with ipilimumab in patients with metastatic melanoma. NEJM 2010. Combined results for ipilimumab + gp100 and ipilimumab alone arms.
[c] Robert et al. Anti-programmed-death-receptor-1 treatment with pembrolizumab in ipilimumab-refractory advanced melanoma: a randomised dose-comparison cohort of a phase 1 trial. Lancet. 2014.
[d] Robert et al. Nivolumab in Previously Untreated Melanoma without BRAF Mutation. NEJM 2014. Percentage of Stage III patients not reported, Stage III-IVM1b = 39%.
[e] Wolchok et al. Nivolumab plus ipilimumab in advanced melanoma. NEJM 2013. Percentage of Stage III patients not reported, Stage IV = 92%. Data for concurrent treatment cohorts.
[f] Thompson et al. Phase 2 study of intralesional PV-10 in refractory metastatic melanoma. Ann Surg Oncol 2014.

Phase 3 Study in Locally Advanced Cutaneous Melanoma

RCT
- PV-10 vs DTIC or TMZ in 225 patients randomized 2 : 1
- Cross-over on documented progression
- Interim assessment

Patients
- Cutaneous and subcutaneous disease (Stage IIIB or IIIC) with no active nodal disease
- Failed or not candidates for at least one immune checkpoint inhibitor
- BRAF wild-type
- Cutaneous only target disease (critical for photodocumentation and IRC review)
- All disease must be injectable on Day 1
- No lesion > 3 cm, no more than 20 lesions at baseline
- 1-5 cutaneous target lesions at least 1 cm in longest diameter

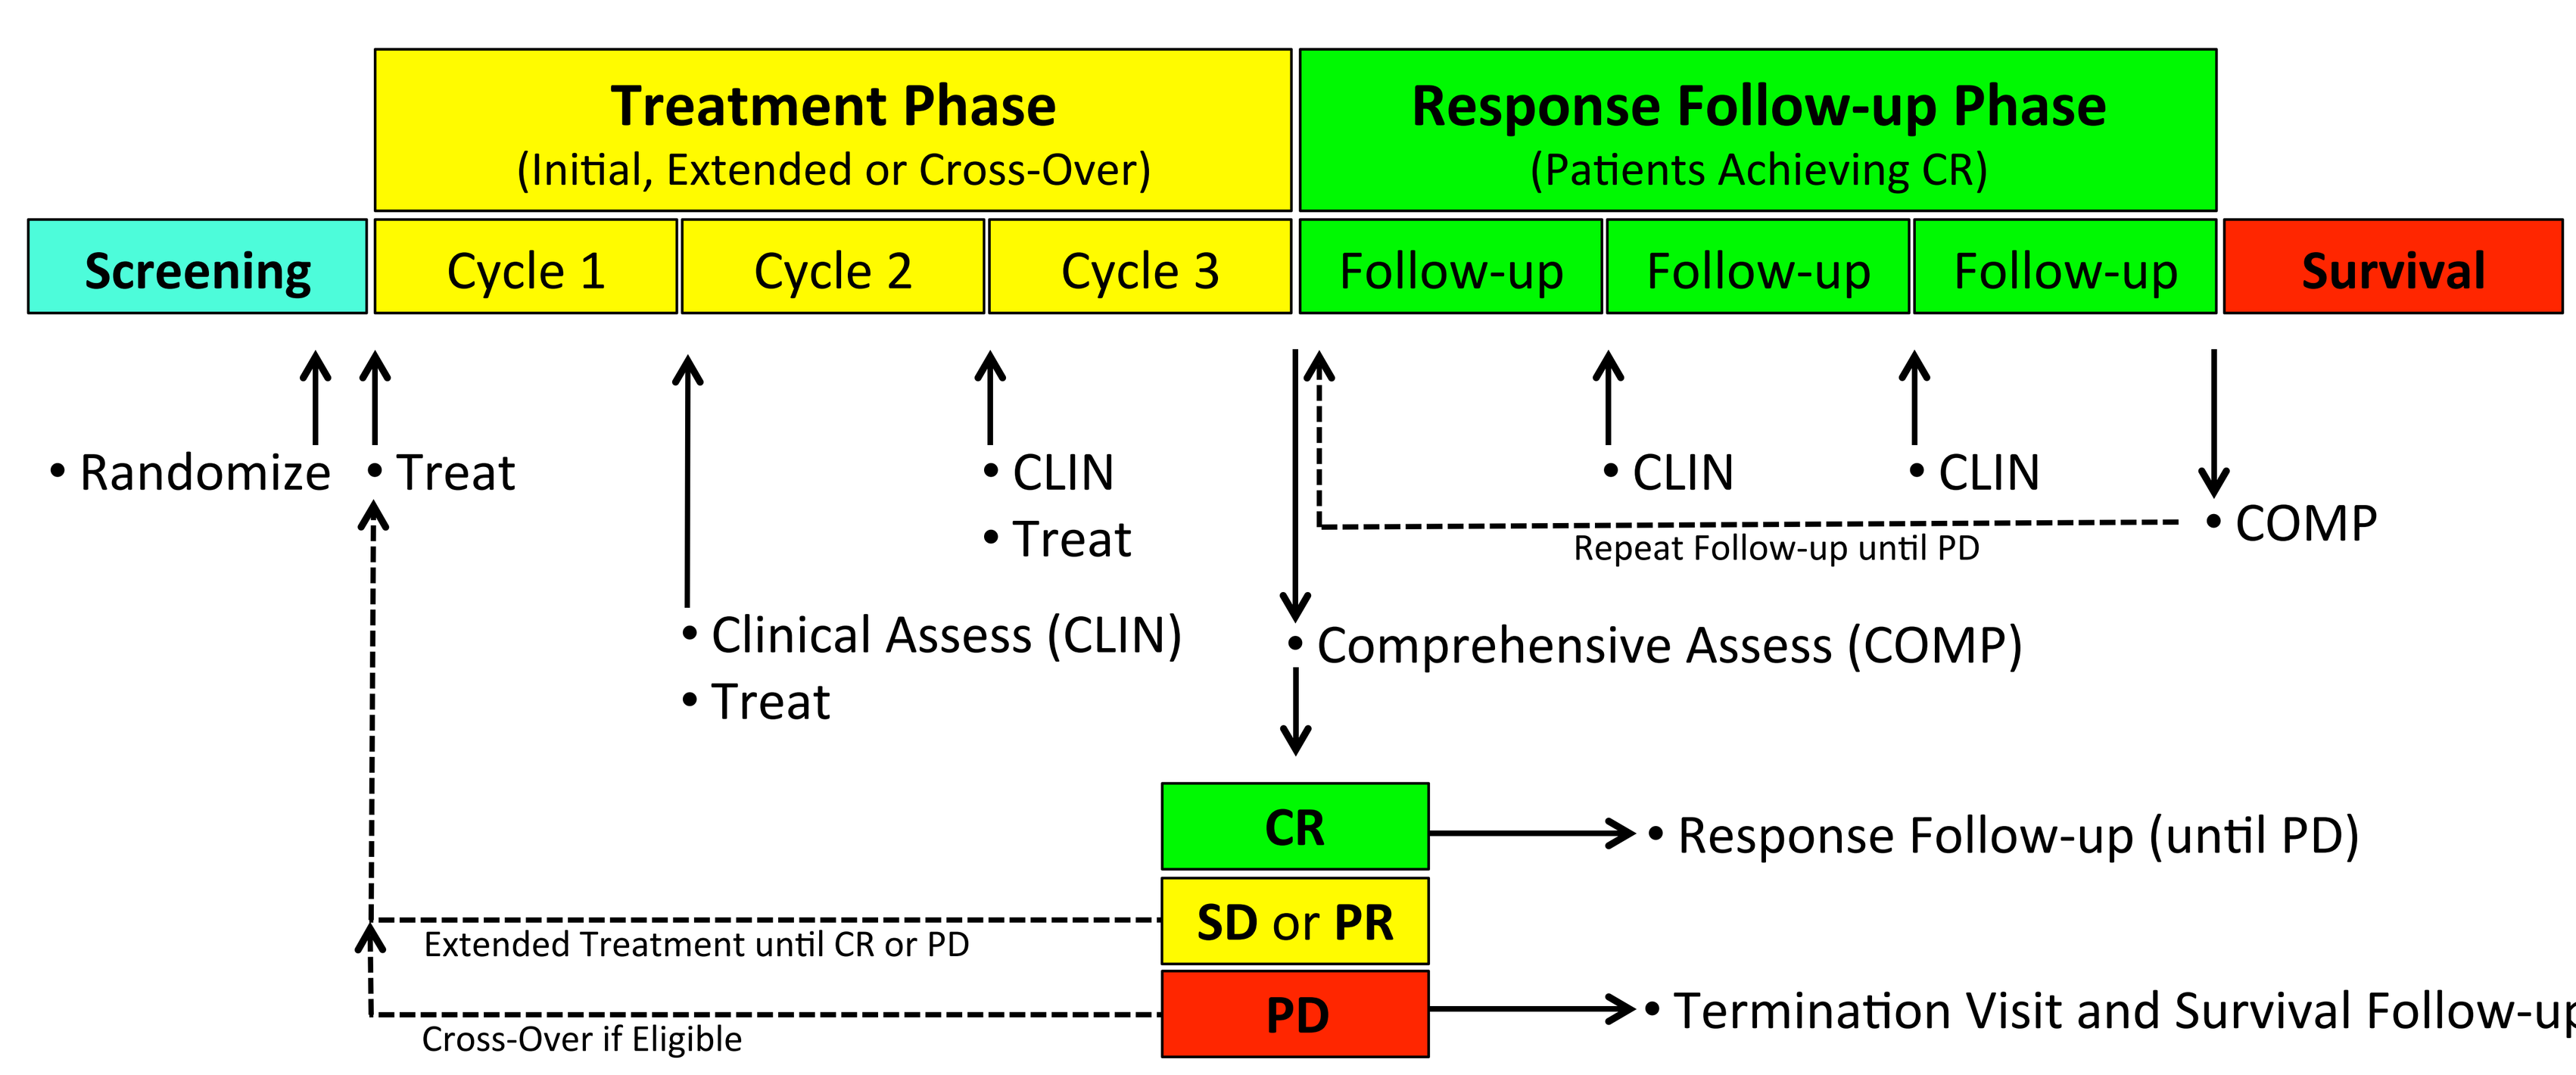
Treatment Phase
(Initial, Extended or Cross-Over)
Response Follow-up Phase
(Patients Achieving CR)
Screening
Cycle 1
Cycle 2
Cycle 3
Follow-up
Follow-up
Follow-up
Survival
• Randomize
• Treat
• Clinical Assess (CLIN)
• Treat
• CLIN
• Treat
• Comprehensive Assess (COMP)
• CLIN
• CLIN
• COMP
Repeat Follow-up until PD
CR
SD or PR
PD
• Response Follow-up (until PD)
• Termination Visit and Survival Follow-up
Extended Treatment until CR or PD
Cross-Over if Eligible

Primary EP
- Progression Free Survival by RECIST 1.1

Secondary
- Complete Response Rate (CRR)
- Duration of Complete Response
- Overall Survival
- Safety

Exploratory
- Change in Domain Scores on Skindex-16
- Change in Patient Reported Pain and Pain Medication Use
- Change in Investigator Assessed Lesion Bleeding, Ulceration and Infection (CTCAE)

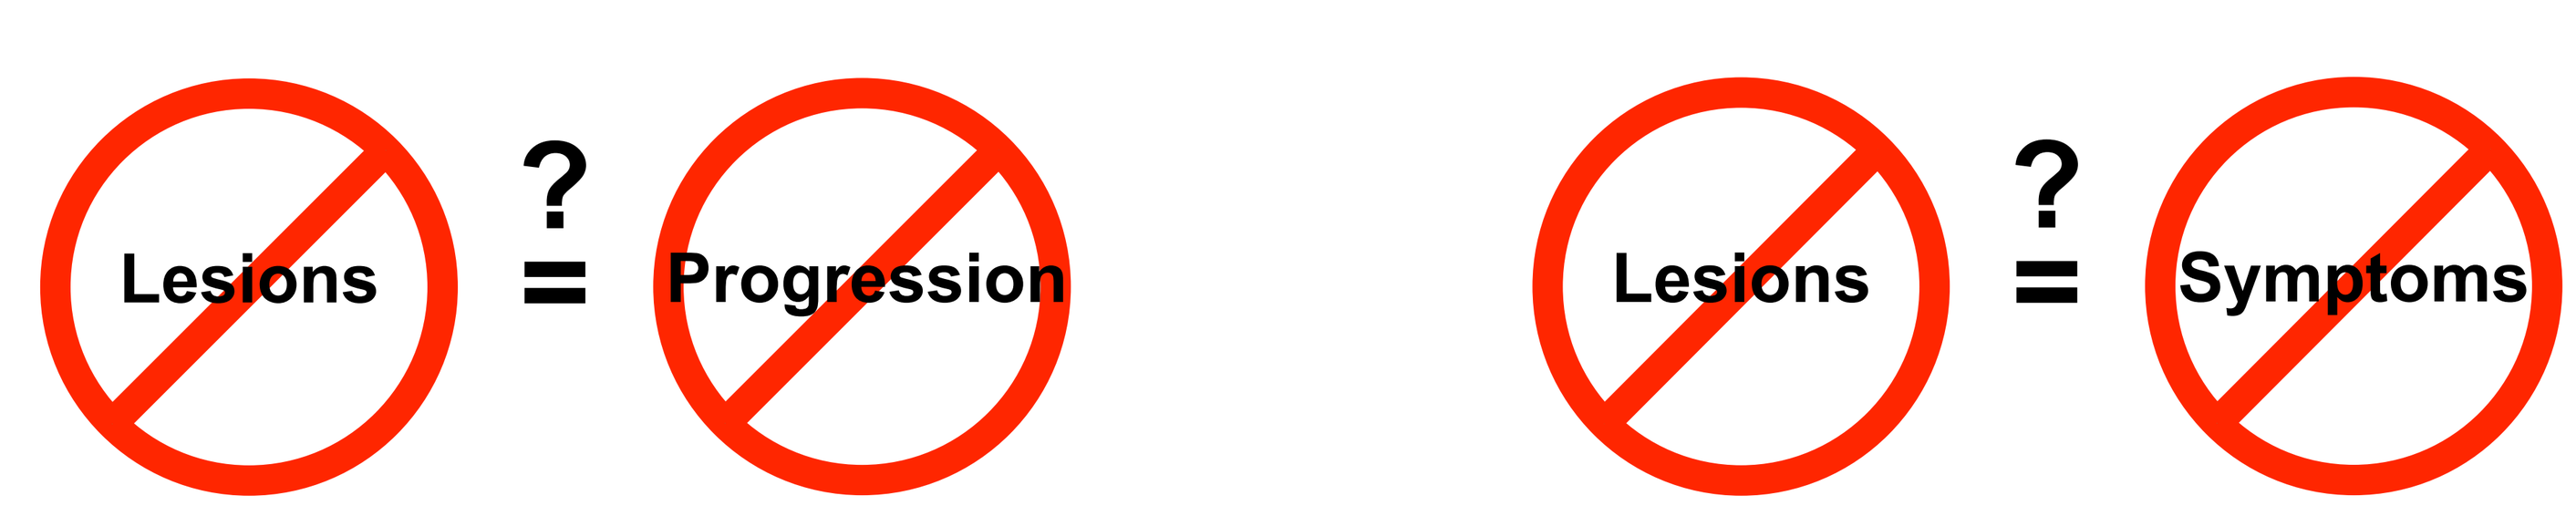
Lesions
?
=
Progression
Lesions
?
=
Symptoms