Phase 1 Study of PV-10 for Chemoablation of Hepatocellular Cancer and Cancer Metastatic to the Liver

Paul Goldfarb, MD[1], Russell Low, MD[1], James Lyon, MD[1], Sanjiv S. Agarwala, MD[2], Alexander Rosemurgy, MD FACS[3], and Eric A. Wachter, PhD[4]

Sharp Clinical Oncology Research, San Diego, CA USA[1], St. Luke's Hospital and Health Network and Temple University, Bethlehem, PA USA[2], Florida Hospital Tampa, Tampa, FL USA[3], and Provectus Biopharmaceuticals, Inc., Knoxville, TN USA[4]

Abstract Number P-116
ESMO 17th World Congress on Gastrointestinal Cancer
Barcelona, Spain 1-4 July 2015

For additional information:
conference@pvct.com

Abstract

BACKGROUND: Intralesional PV-10, a 10% solution of rose bengal, has recently demonstrated high rates of complete response and durable local control in metastatic melanoma [1]. The current Phase 1 study is assessing safety, pharmacokinetics, and preliminary efficacy of PV-10 in subjects with non-resectable hepatocellular carcinoma (HCC) or cancer metastatic to the liver (NCT 00986661).

METHOD: Subjects having at least one liver tumor ≥ 1 cm are administered a single percutaneous intralesional injection of PV-10 to one Target Lesion at dose of 0.25 or 0.50 mL per cm^3 lesion volume. Plasma concentrations of PV-10 from 1 hour to 28 days after injection are measured. Radiologic assessments of the injected Target Lesion are performed to determine response over initial 28 day and long-term 9-15 month periods. Serum levels of potential liver injury markers are measured, and adverse events recorded.

RESULTS: In an initial study cohort, six subjects received PV-10. Significant adverse events were limited to injection site and photosensitivity reactions, and resolved without sequelae. All injected tumors were stable in size at 28 days, and of 4 that had long-term assessment, 2 had partial response, for a long-term tumor-specific objective response rate of 50%. PV-10 plasma levels decreased rapidly in a bi-exponential pattern, with initial and terminal phase half-lives of 4.5 and 100 hours, respectively. Elevated liver enzymes levels subsided within a week of treatment.

CONCLUSIONS: Preliminary efficacy in treatment of liver tumors with PV-10 was observed. Toxicity was transient, and treatment had acceptable tolerability. The study is continuing at three study centers with two expansion cohorts to assess response in hepatocellular carcinoma and other cancers metastatic to the liver.

[1] Thompson et al., Annals Surg Oncol 2015; 22: 2135-2142.

Background

PV-10 is a sterile, non-pyrogenic solution of Rose Bengal disodium (10% RB) for intralesional injection

- **Small molecule** Fluorescein derivative attributed to Gnehm (1882)
- **Prior human use** of RB
 - IV hepatic diagnostic, ^{131}I radiolabeled RB: Robengatope®
 - Topical ophthalmic diagnostic: Rosettes® and Minims®
- **Established safety history**
 - Not metabolized
 - Short circulatory half-life (ca 30 min)
 - Excretion via bile
- **Radiopaque** with prolonged retention in tumors

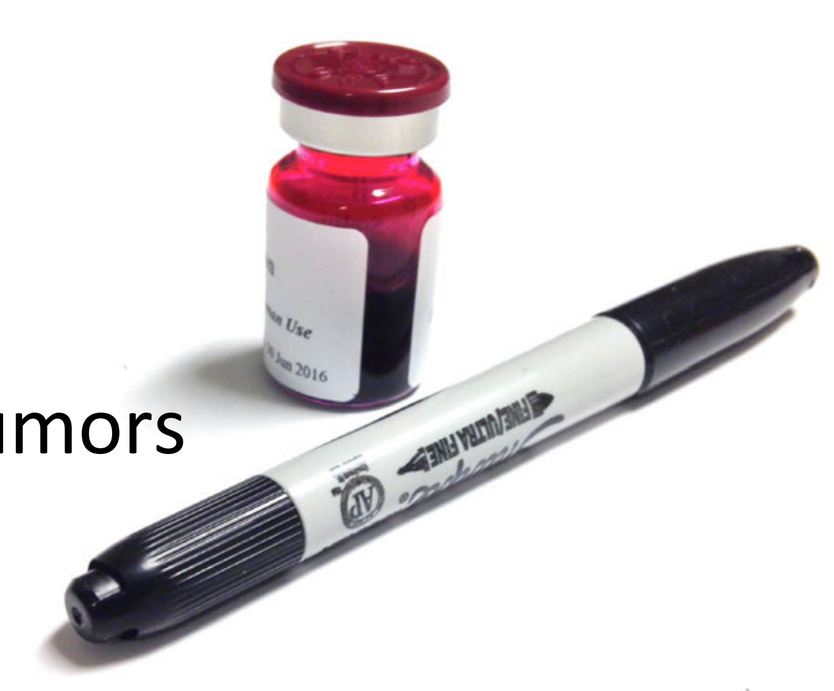

Primary Ablative Mechanism [2]

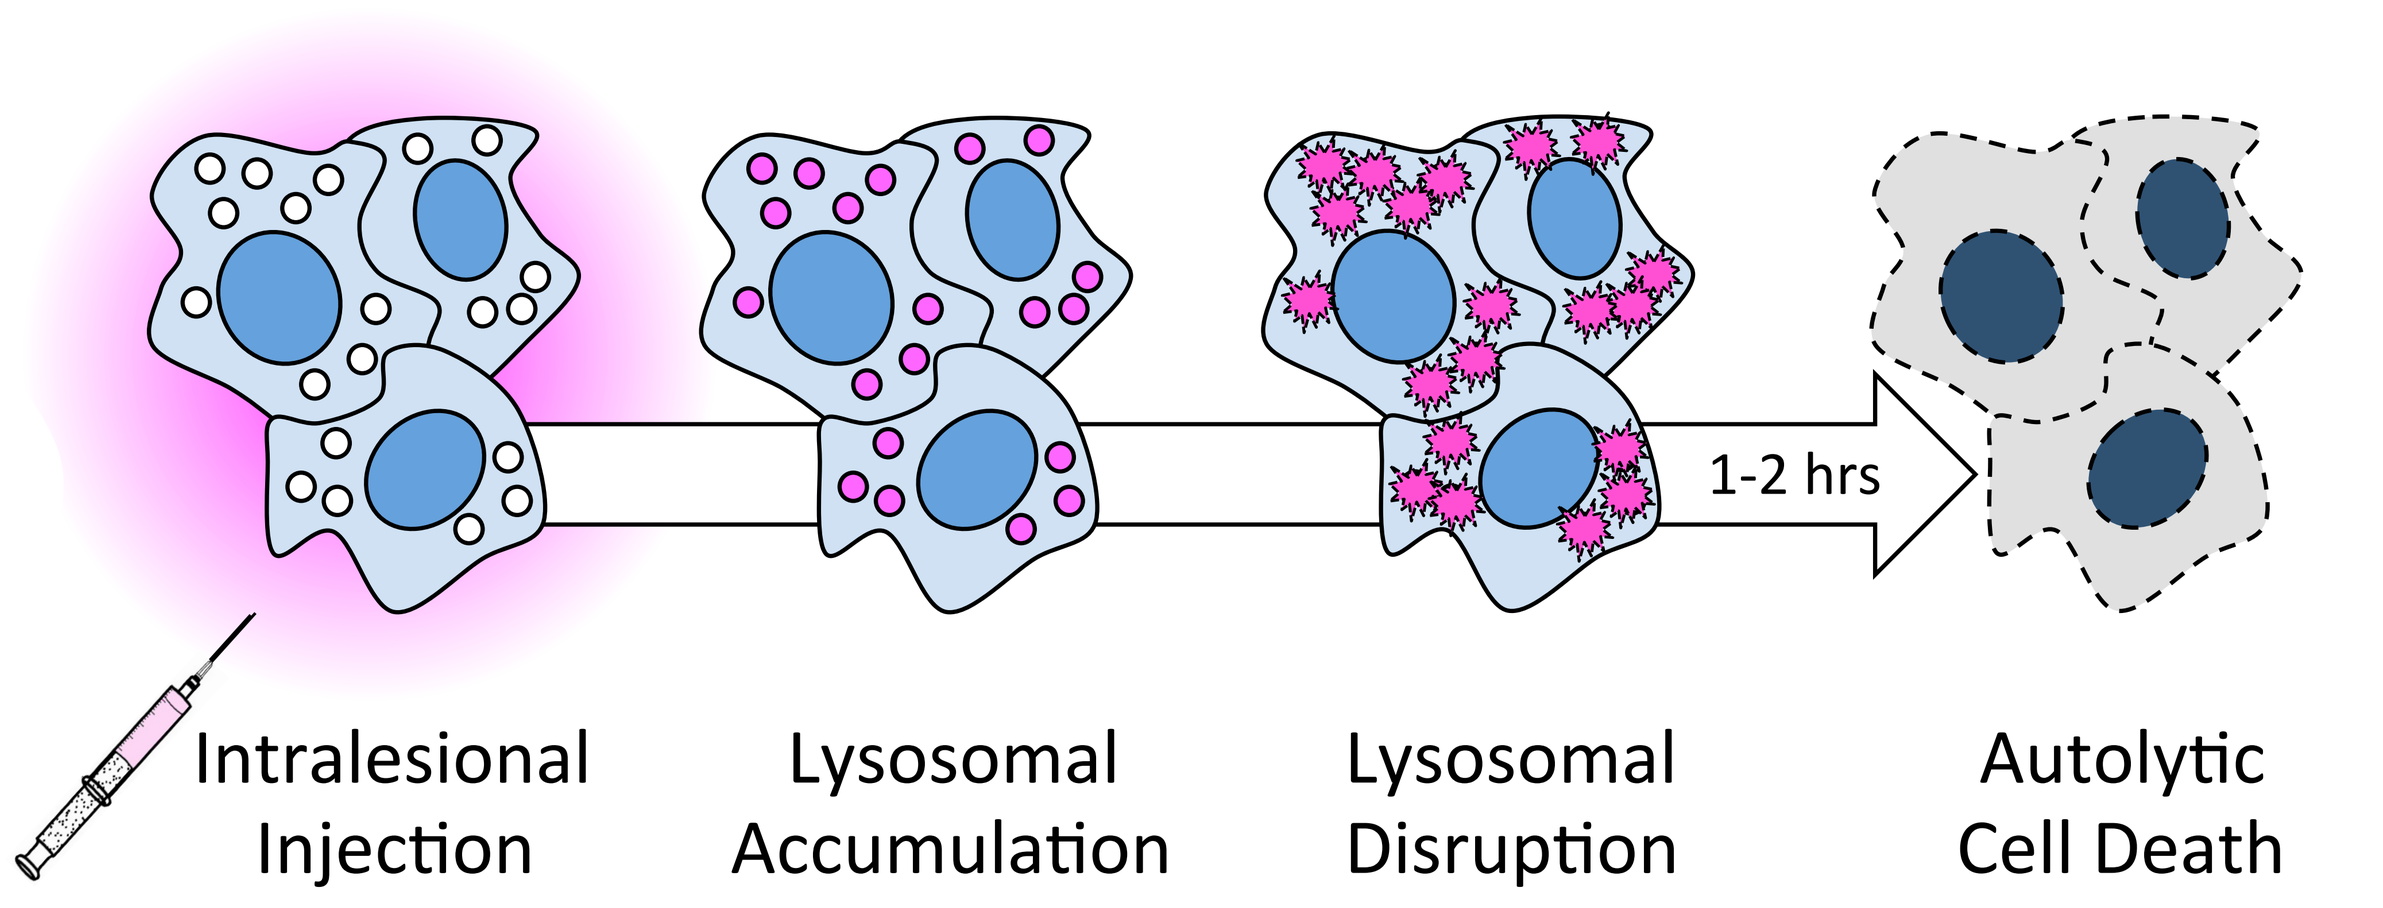
1-2 hrs
Intralesional Injection
Lysosomal Accumulation
Lysosomal Disruption
Autolytic Cell Death

Secondary Immunologic Activation [3]

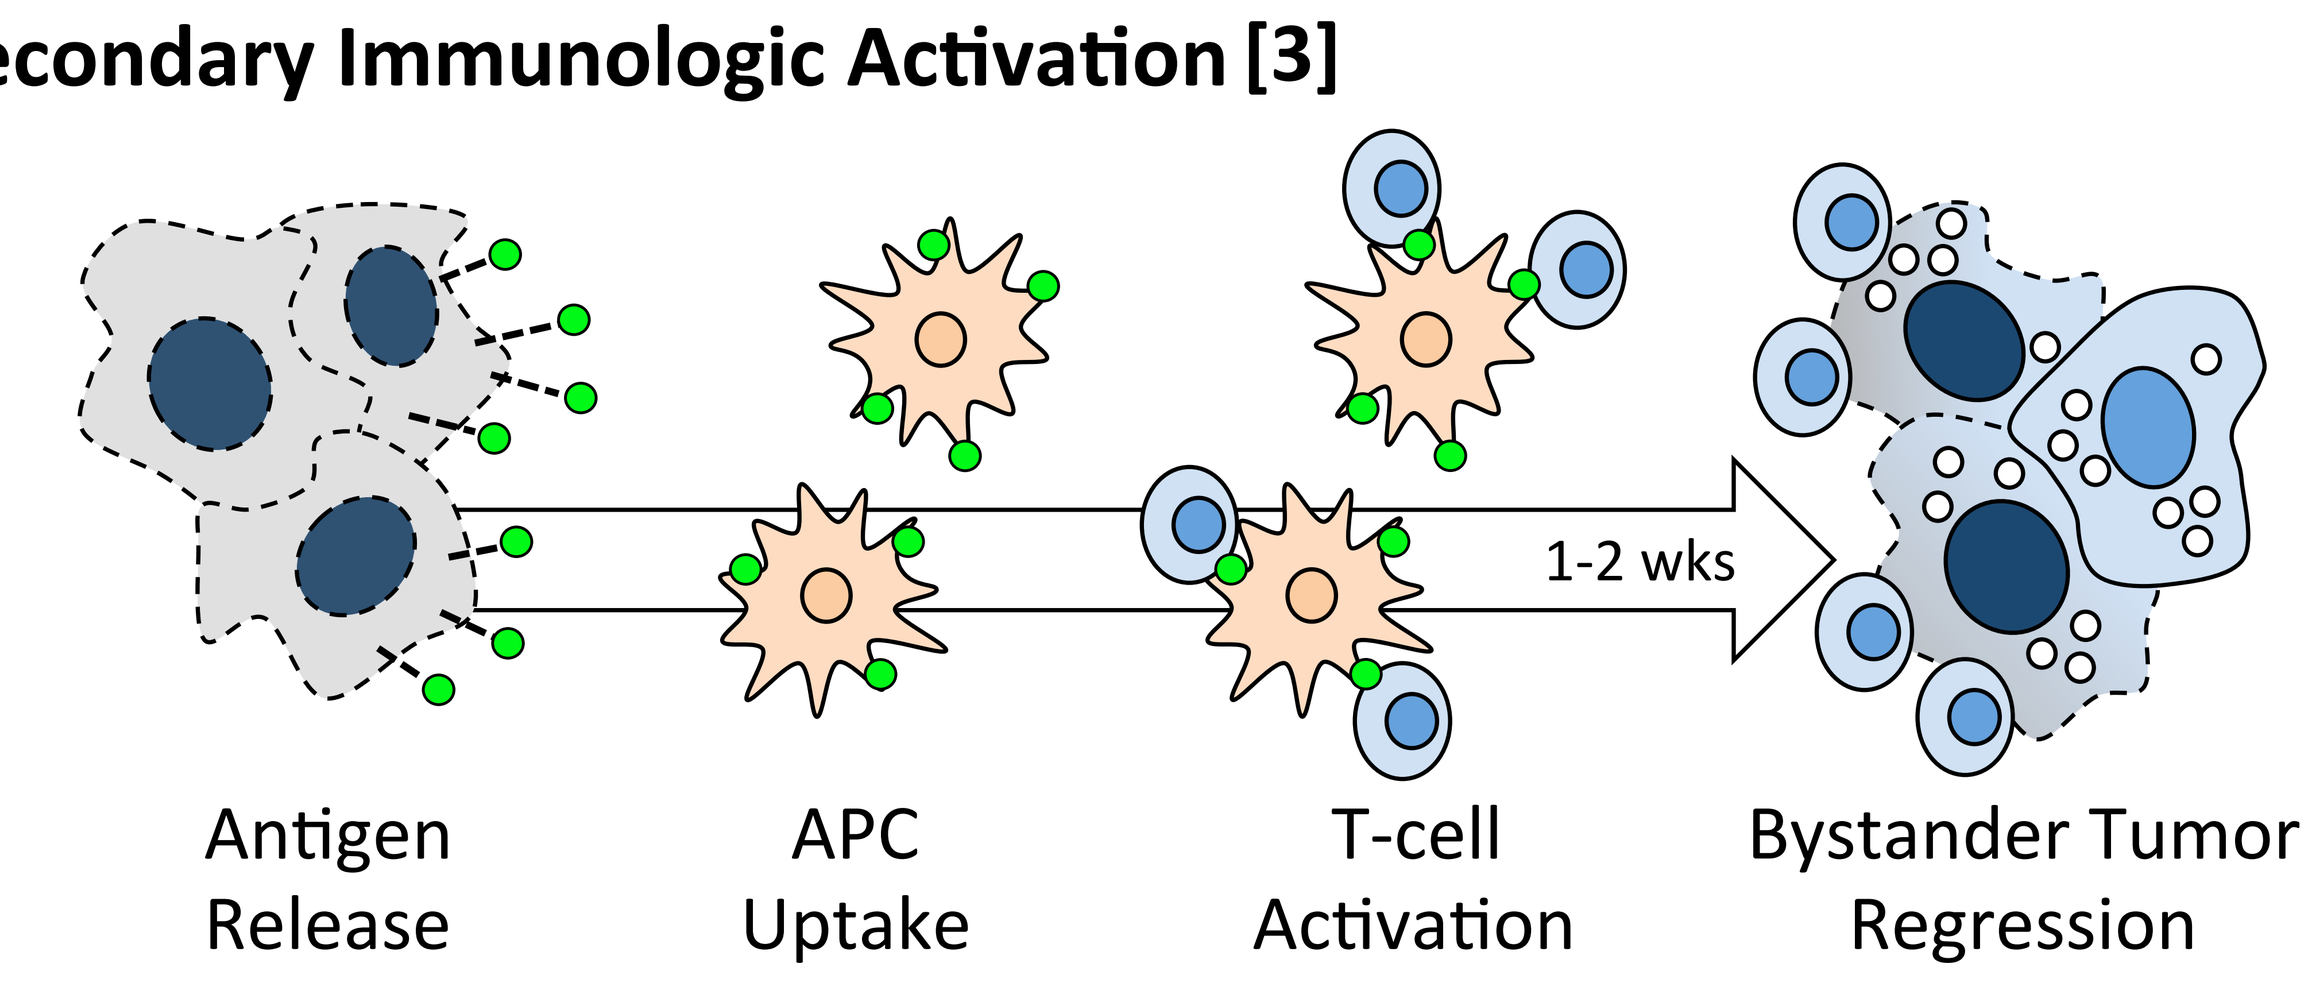
1-2 wks
Antigen Release
APC Uptake
T-cell Activation
Bystander Tumor Regression

[2] Wachter et al., SPIE Proceedings 2012; 4620: 143-147.
[3] Toomey et al., PLoS1 2013; 8: e68561.

Phase 1 Patients

- 6 Male, 7 Female, median age 68 years (range 51 – 89)
- Hepatocellular Carcinoma – 6 patients
- Metastases to Liver – 7 patients: 3 colorectal mets (CRLM), 2 non-small cell lung (NSCL), 2 melanoma (Mel), 1 ovarian (Ova)
 - Injected HCCs: median diameter 3.8 cm (range 1.9 – 8.9 cm)
 - Injected CRLM: 2.5 cm
 - Injected NSCL: 3.2 – 3.6 cm
 - Injected Mel: 1.1 – 1.9 cm
 - Injected Ova: 1.4 cm
 - One HCC and one Mel patient with multiple tumors enrolled twice to allow sequential treatment of additional tumors

Example Response

Subject 0001, female, age 71
3.4 cm HCC lesion injected once with 5.1 mL PV-10
(second 3.8 cm HCC lesion injected once with 7.2 mL PV-10 3 months later)

CT Follow-up

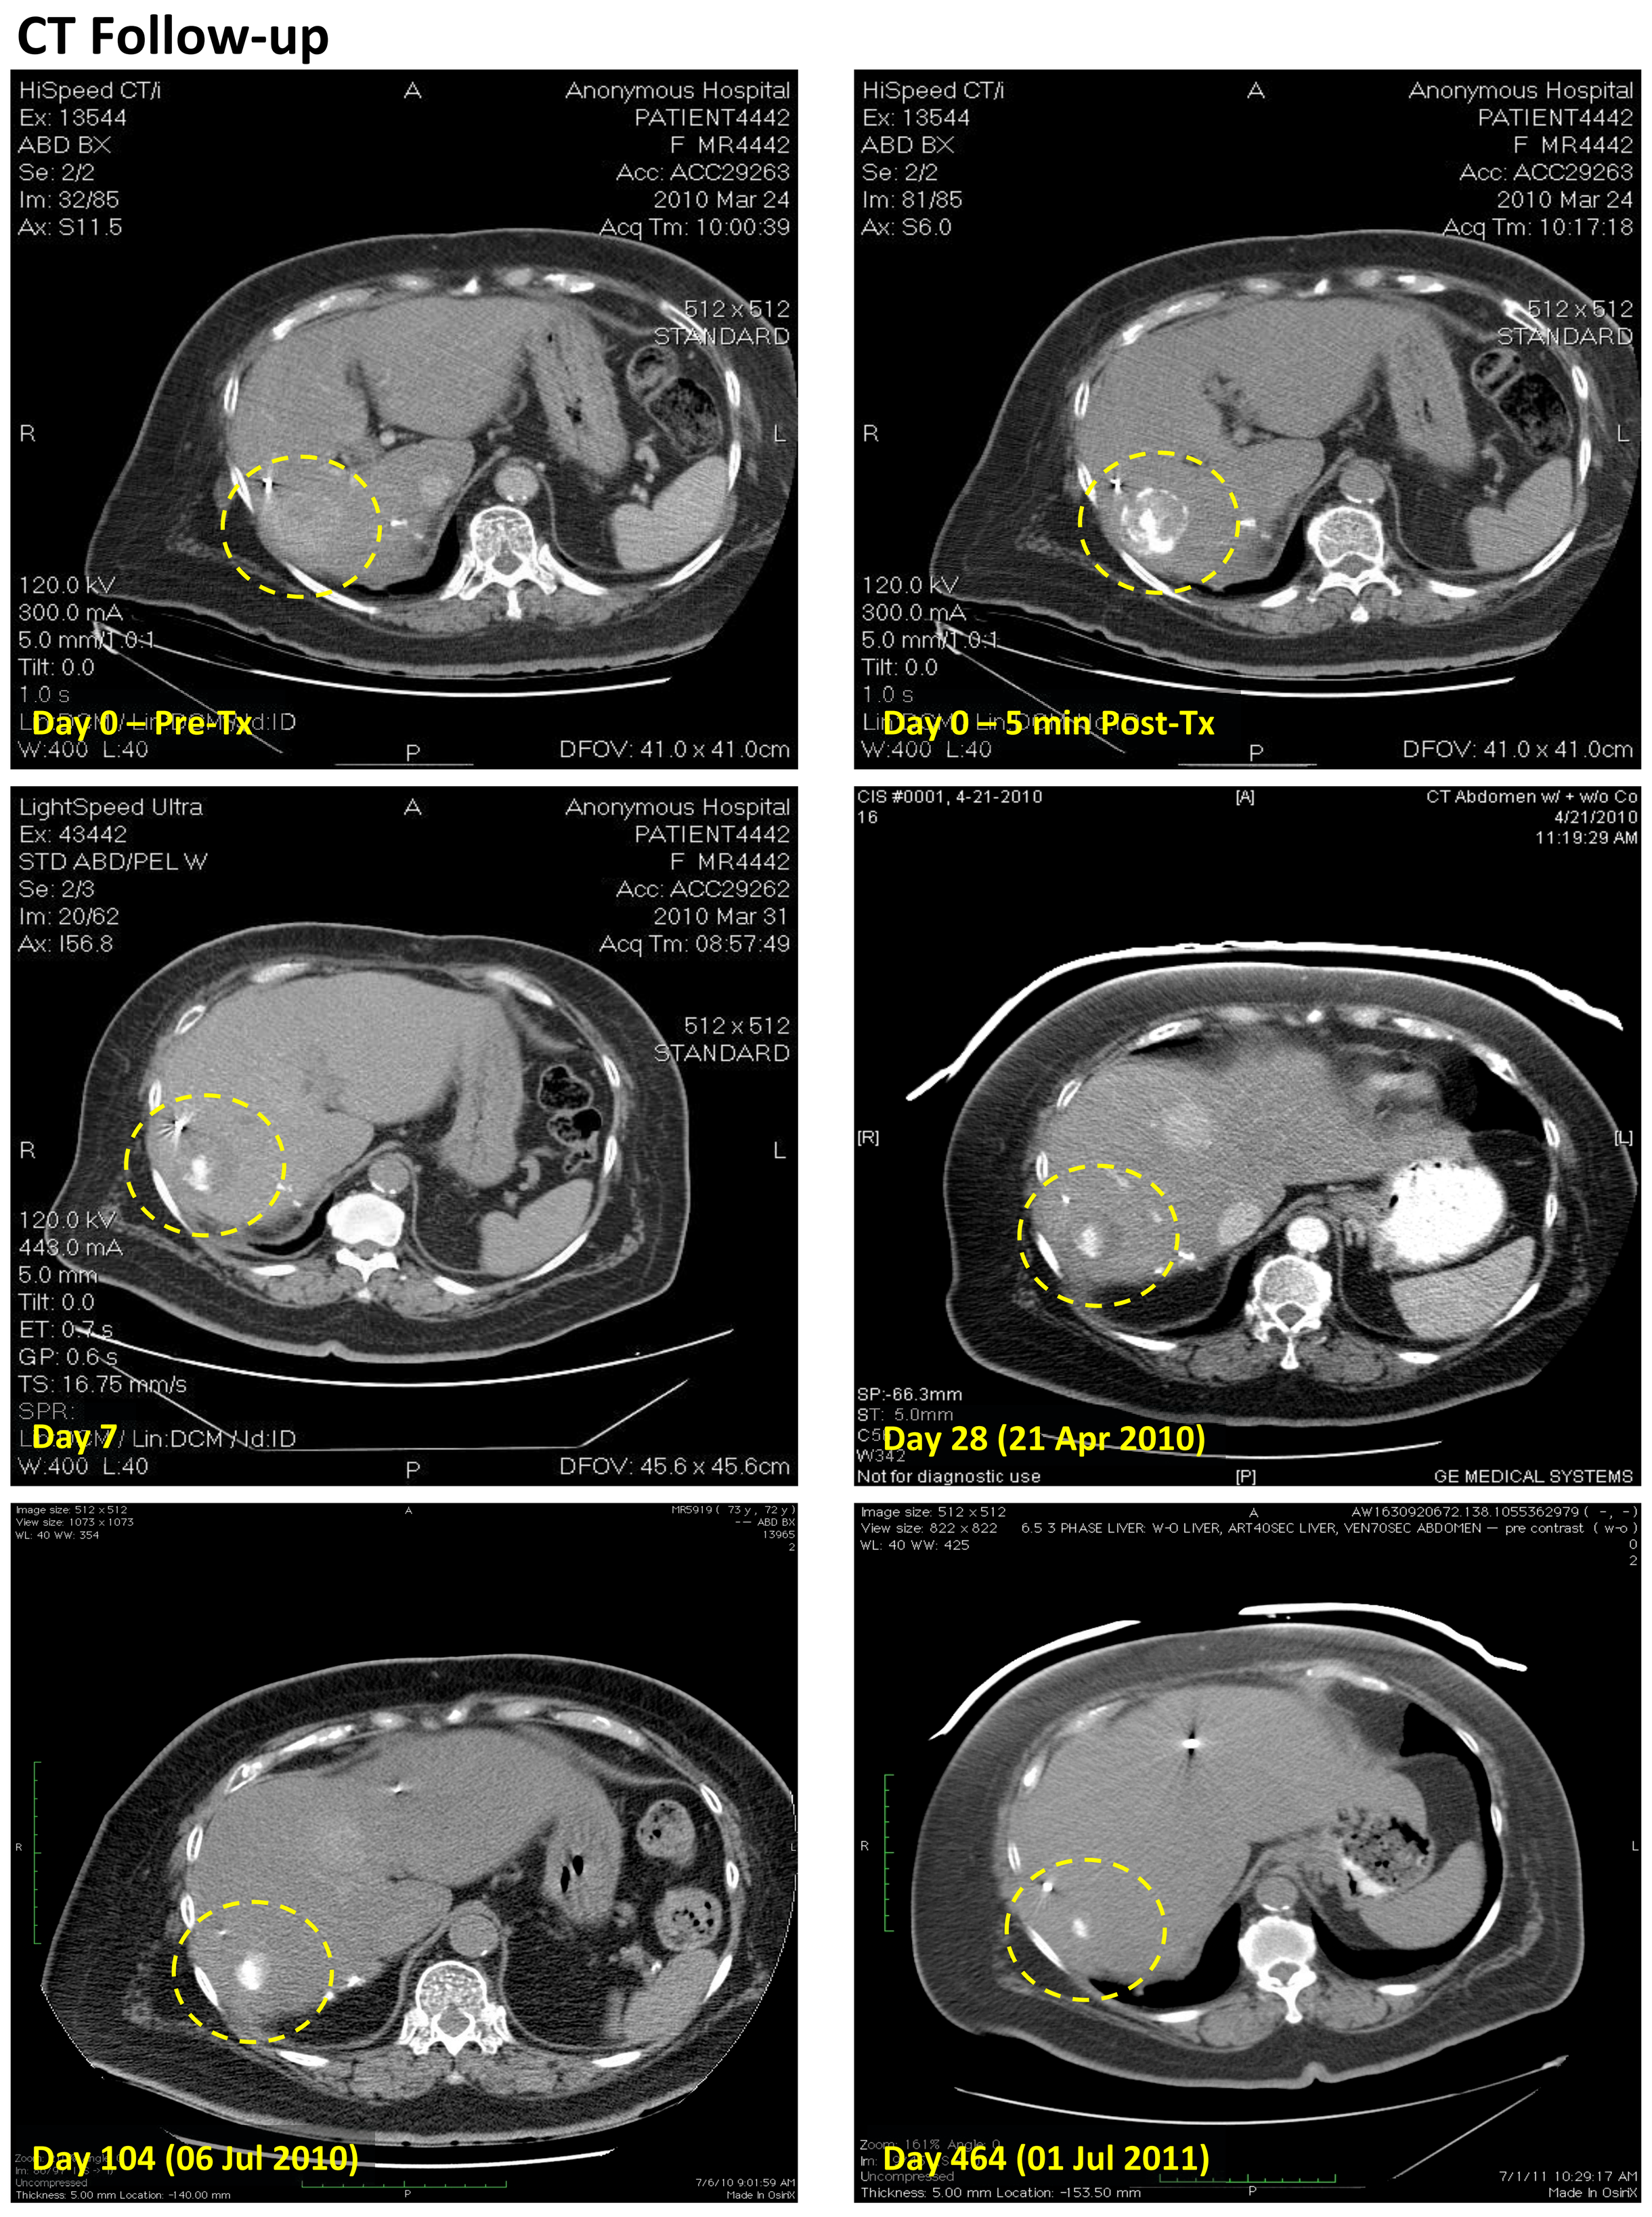
Day 0 – Pre-Tx
Day 0 – 5 min Post-Tx
Day 7
Day 28 (21 Apr 2010)
Day 104 (06 Jul 2010)
Day 464 (01 Jul 2011)

MRI Follow-up

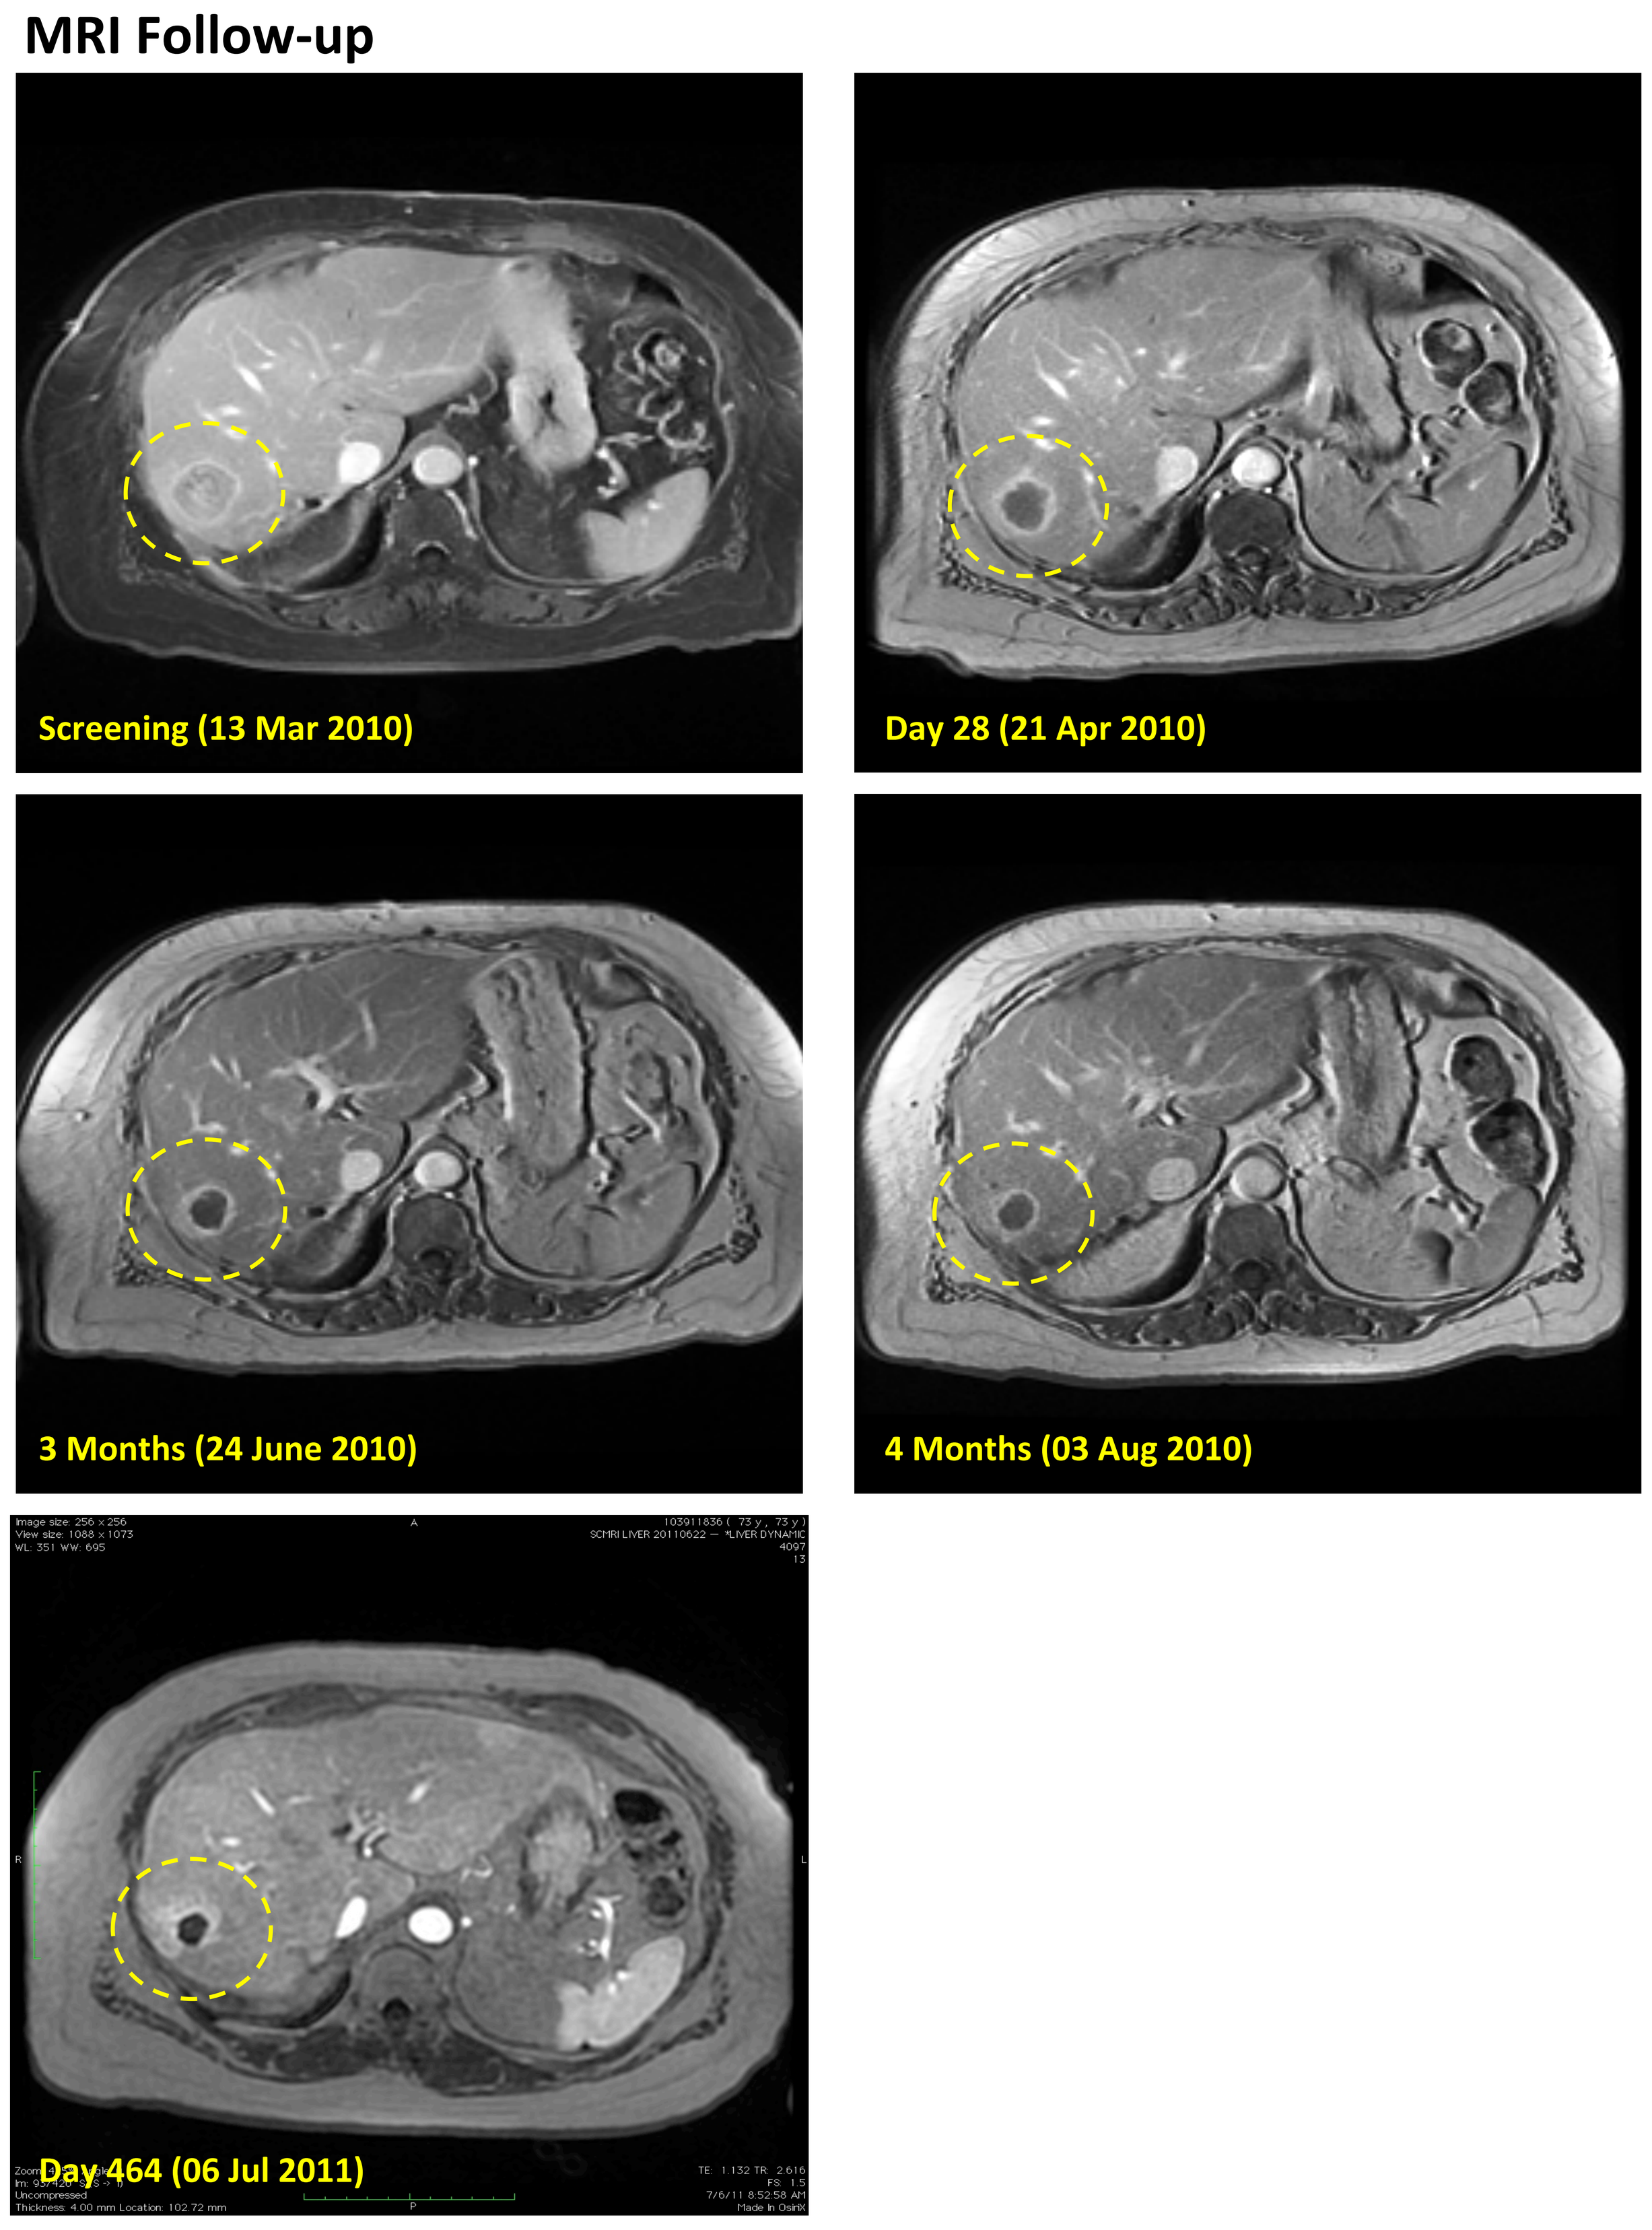
Screening (13 Mar 2010)
Day 28 (21 Apr 2010)
3 Months (24 June 2010)
4 Months (03 Aug 2010)
Day 464 (06 Jul 2011)

Outcome

Subject / Demographics		Disease	Status
0001	F, age 71	HCC	Alive (with Disease, 51 mo)
0004	F, age 73	HCC (HepC, Cirrhosis, Portal Hypertension)	Expired (DP, 48 mo)
0005	M, age 68	HCC (HepB and Cirrhosis)	Alive (NED, 54 mo)
0006	M, age 61	mCRC	Alive (NED, 42 mo)
0007	M, age 67	HCC	Expired (Cardiac Comorbidity, 2 mo)

- 10 of first 13 patients alive after up to 54 months follow-up
 - 1 death due to cardiac comorbidity
 - 1 death due to SAE (89 yo, Karnofsky 60, 8.9 cm HCC, possible thromboembolism)
 - 1 death due to HCC progression
- No long-terms AEs

PV-10 Pharmacokinetics

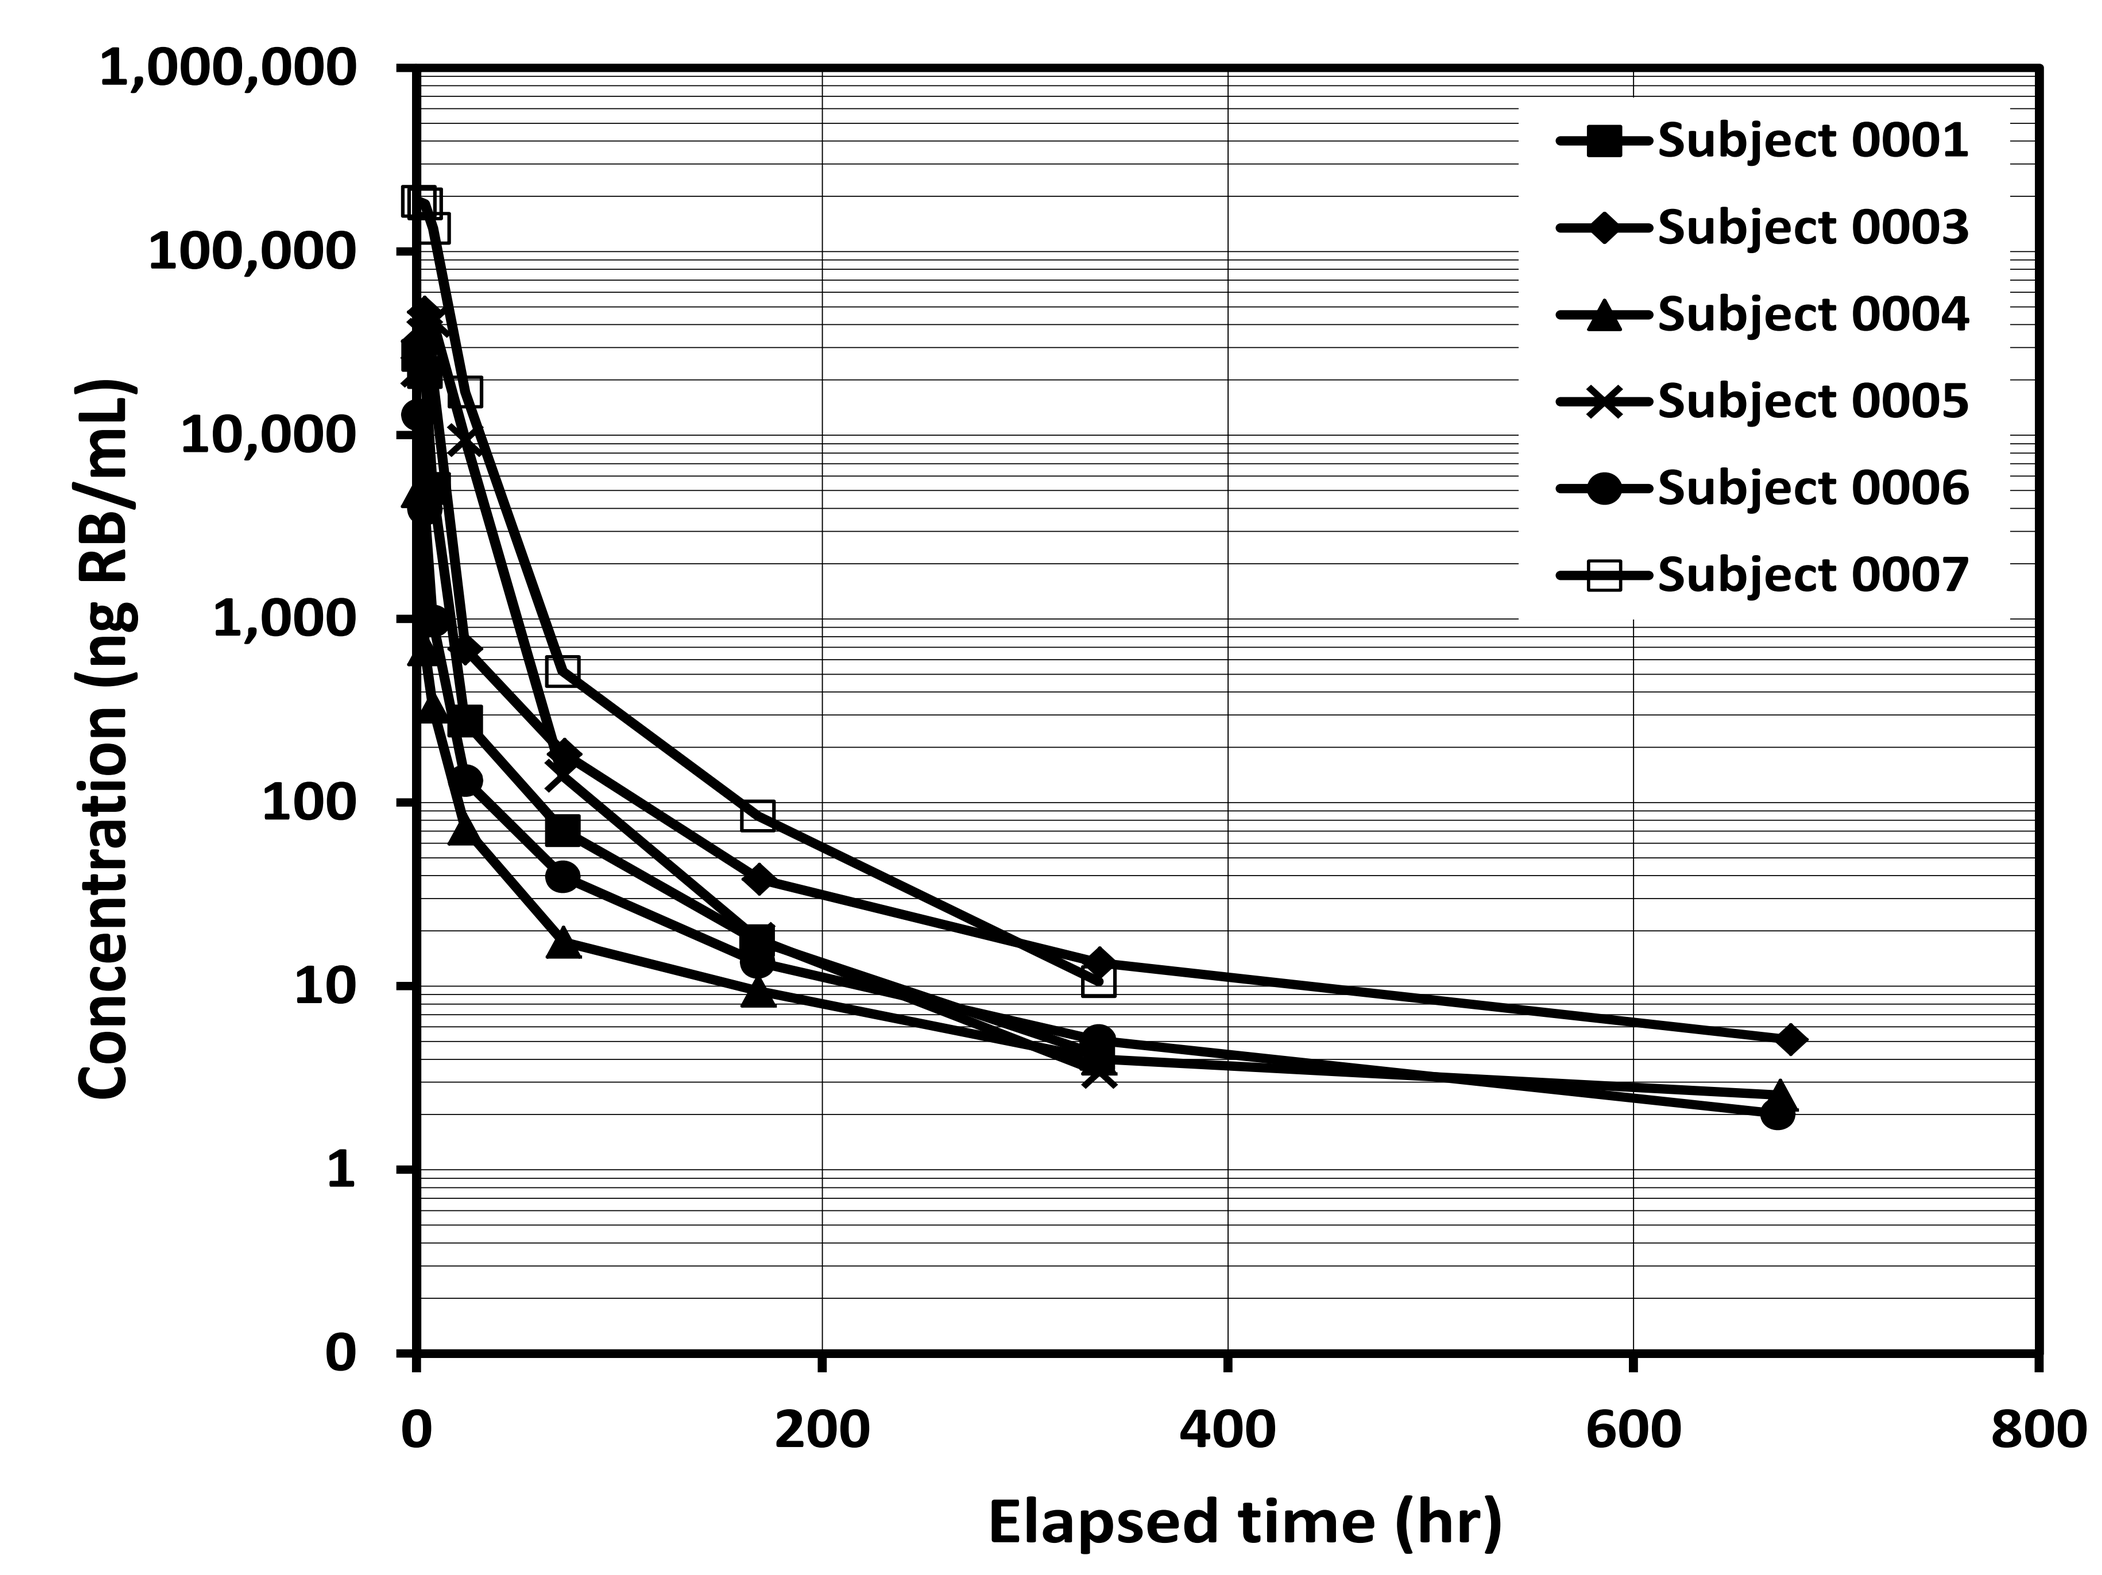
1,000,000
100,000
10,000
1,000
100
10
1
0
Concentration (ng RB/mL)
0
200
400
600
800
Elapsed time (hr)
Subject 0001
Subject 0003
Subject 0004
Subject 0005
Subject 0006
Subject 0007

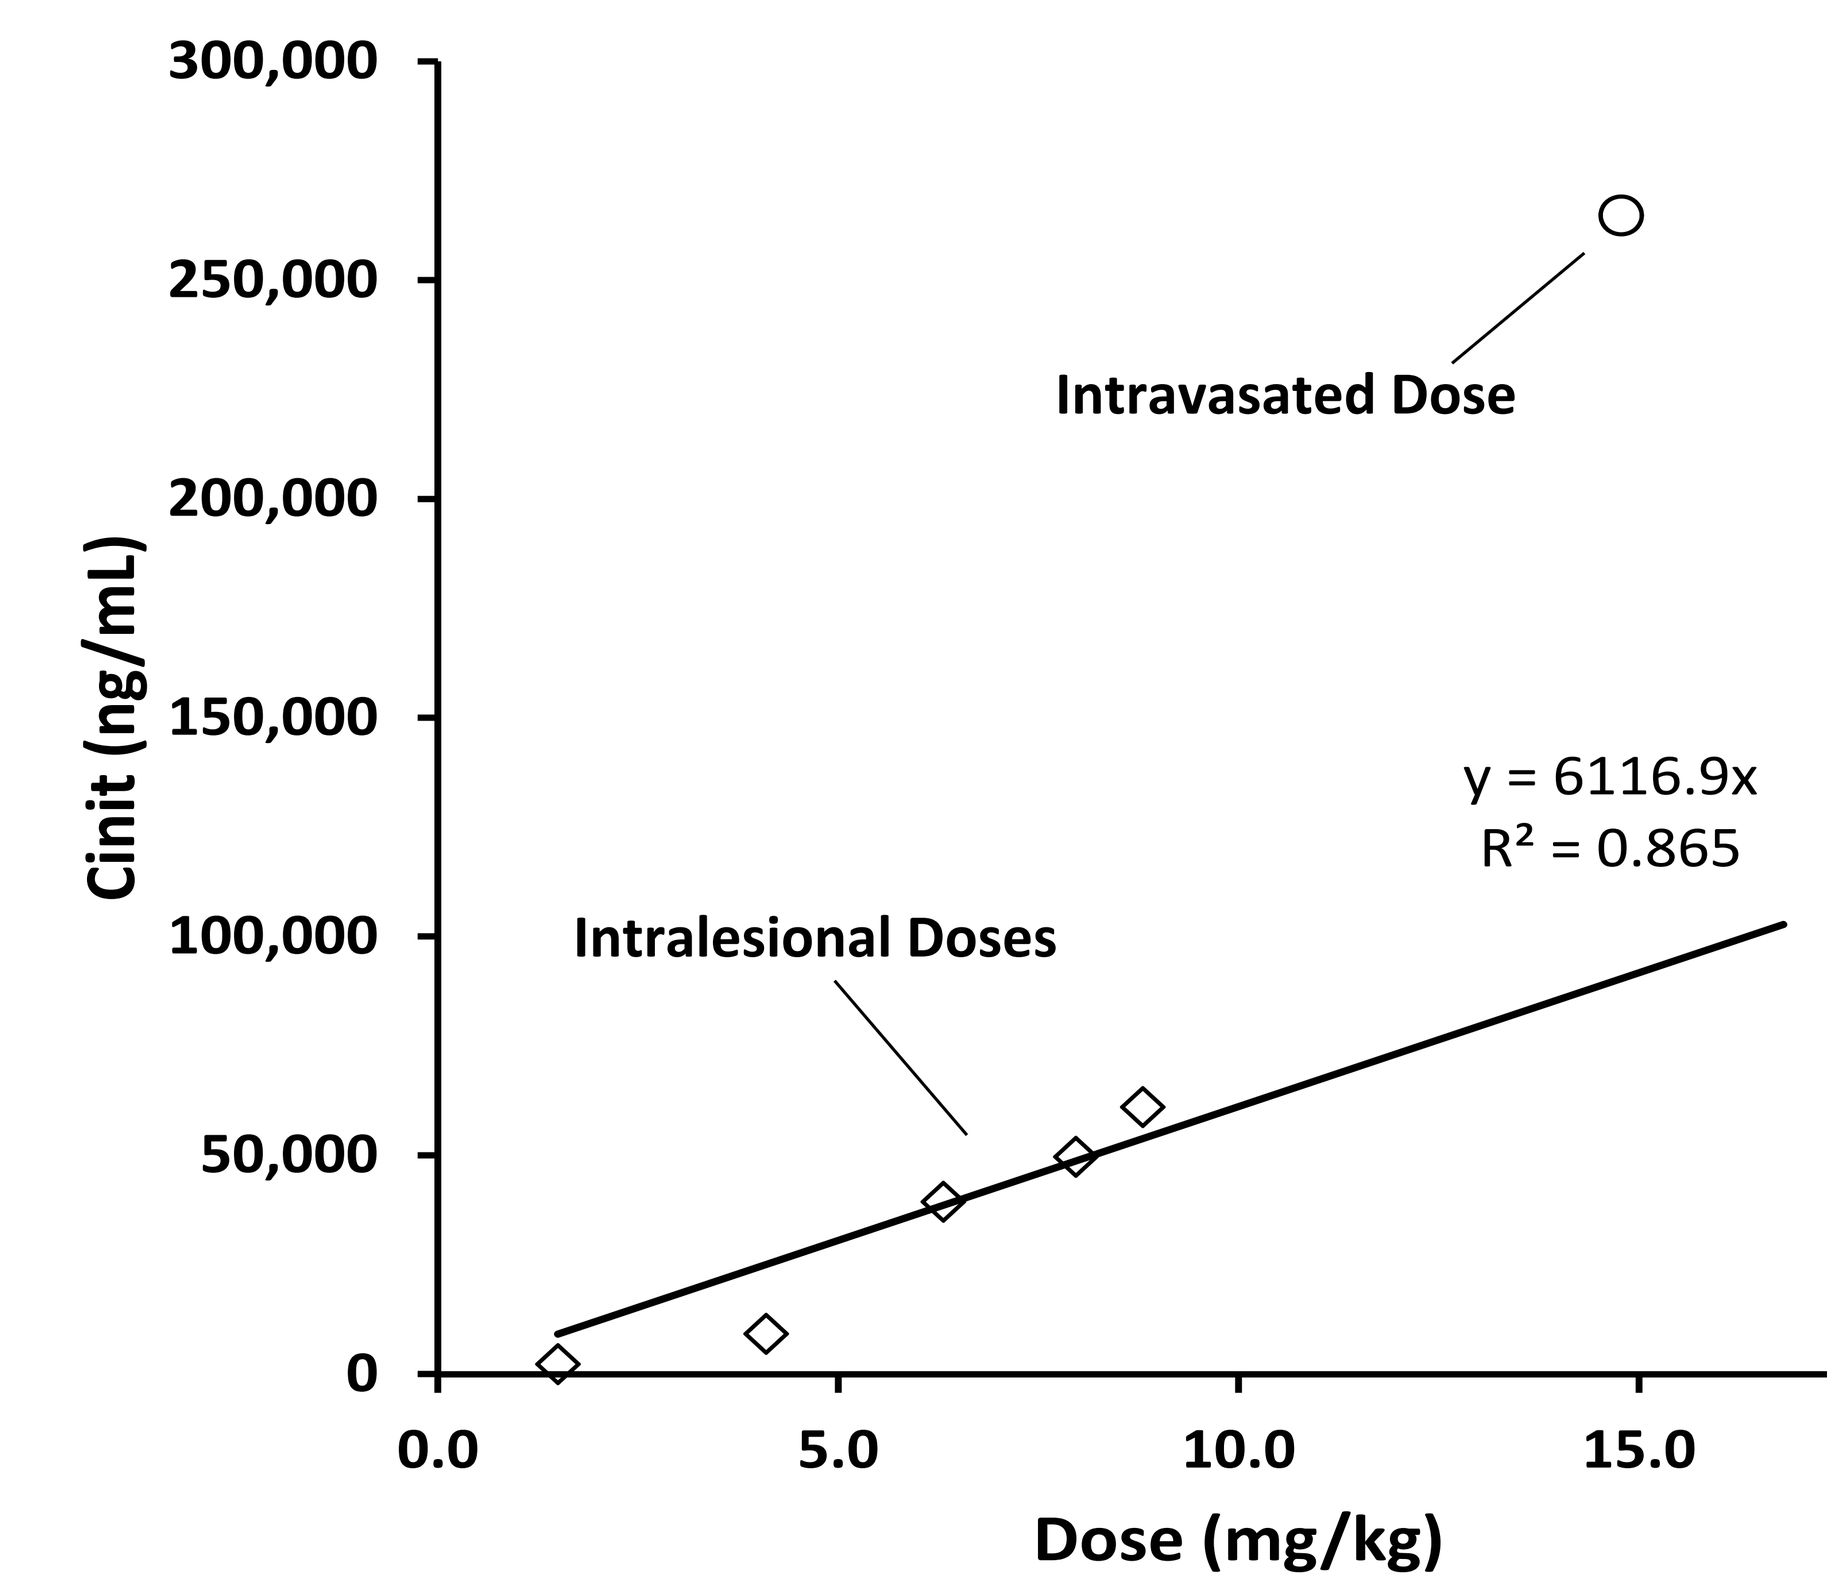
300,000
250,000
200,000
150,000
100,000
50,000
0
Cinit (ng/mL)
Intravasated Dose
y = 6116.9x
R² = 0.865
Intralesional Doses
0.0
5.0
10.0
15.0
Dose (mg/kg)

RB extravasate rapidly cleared with prolonged retention in tumor

- PK samples from initial 6 participants over 28 days following 1st injection
- Bi-exponential clearance (extravasate followed by tumor depot):

$C_{initial}$	29,500 ng RB/mL		
$k_{D/A}$	0.155 hr^{-1}	k_E	0.0070 hr^{-1}
$t_{1/2, D/A}$	4.5 hours	$t_{1/2, E}$	100 hours (4.2 days)
$\%AUC\infty_{D/A}$	89.1%	$\%AUC\infty_E$	7.0%

- Rapid clearance consistent with observed safety profile of PV-10
- Systemic uptake and clearance compare favorably with Robengatope® bolus ($C_{initial}$ ≈ 20,000 ng/mL and k_E = 0.234 hr^{-1})
- Levels decrease to < 5% of $C_{initial}$ within 7 days, making cumulative plasma levels unlikely upon repeat treatment at intervals of 1 week or greater

Liver Enzymes

Transient Elevation of Transaminases

- Samples at screening and 1, 2, 3, 7, 14, 28 and 274 days after injection
- Marked elevation of ALT / AST upon ablation
- Return to baseline within 7-14 days
- Similar trends reported for EtOH attributed to "technical success" of ablation

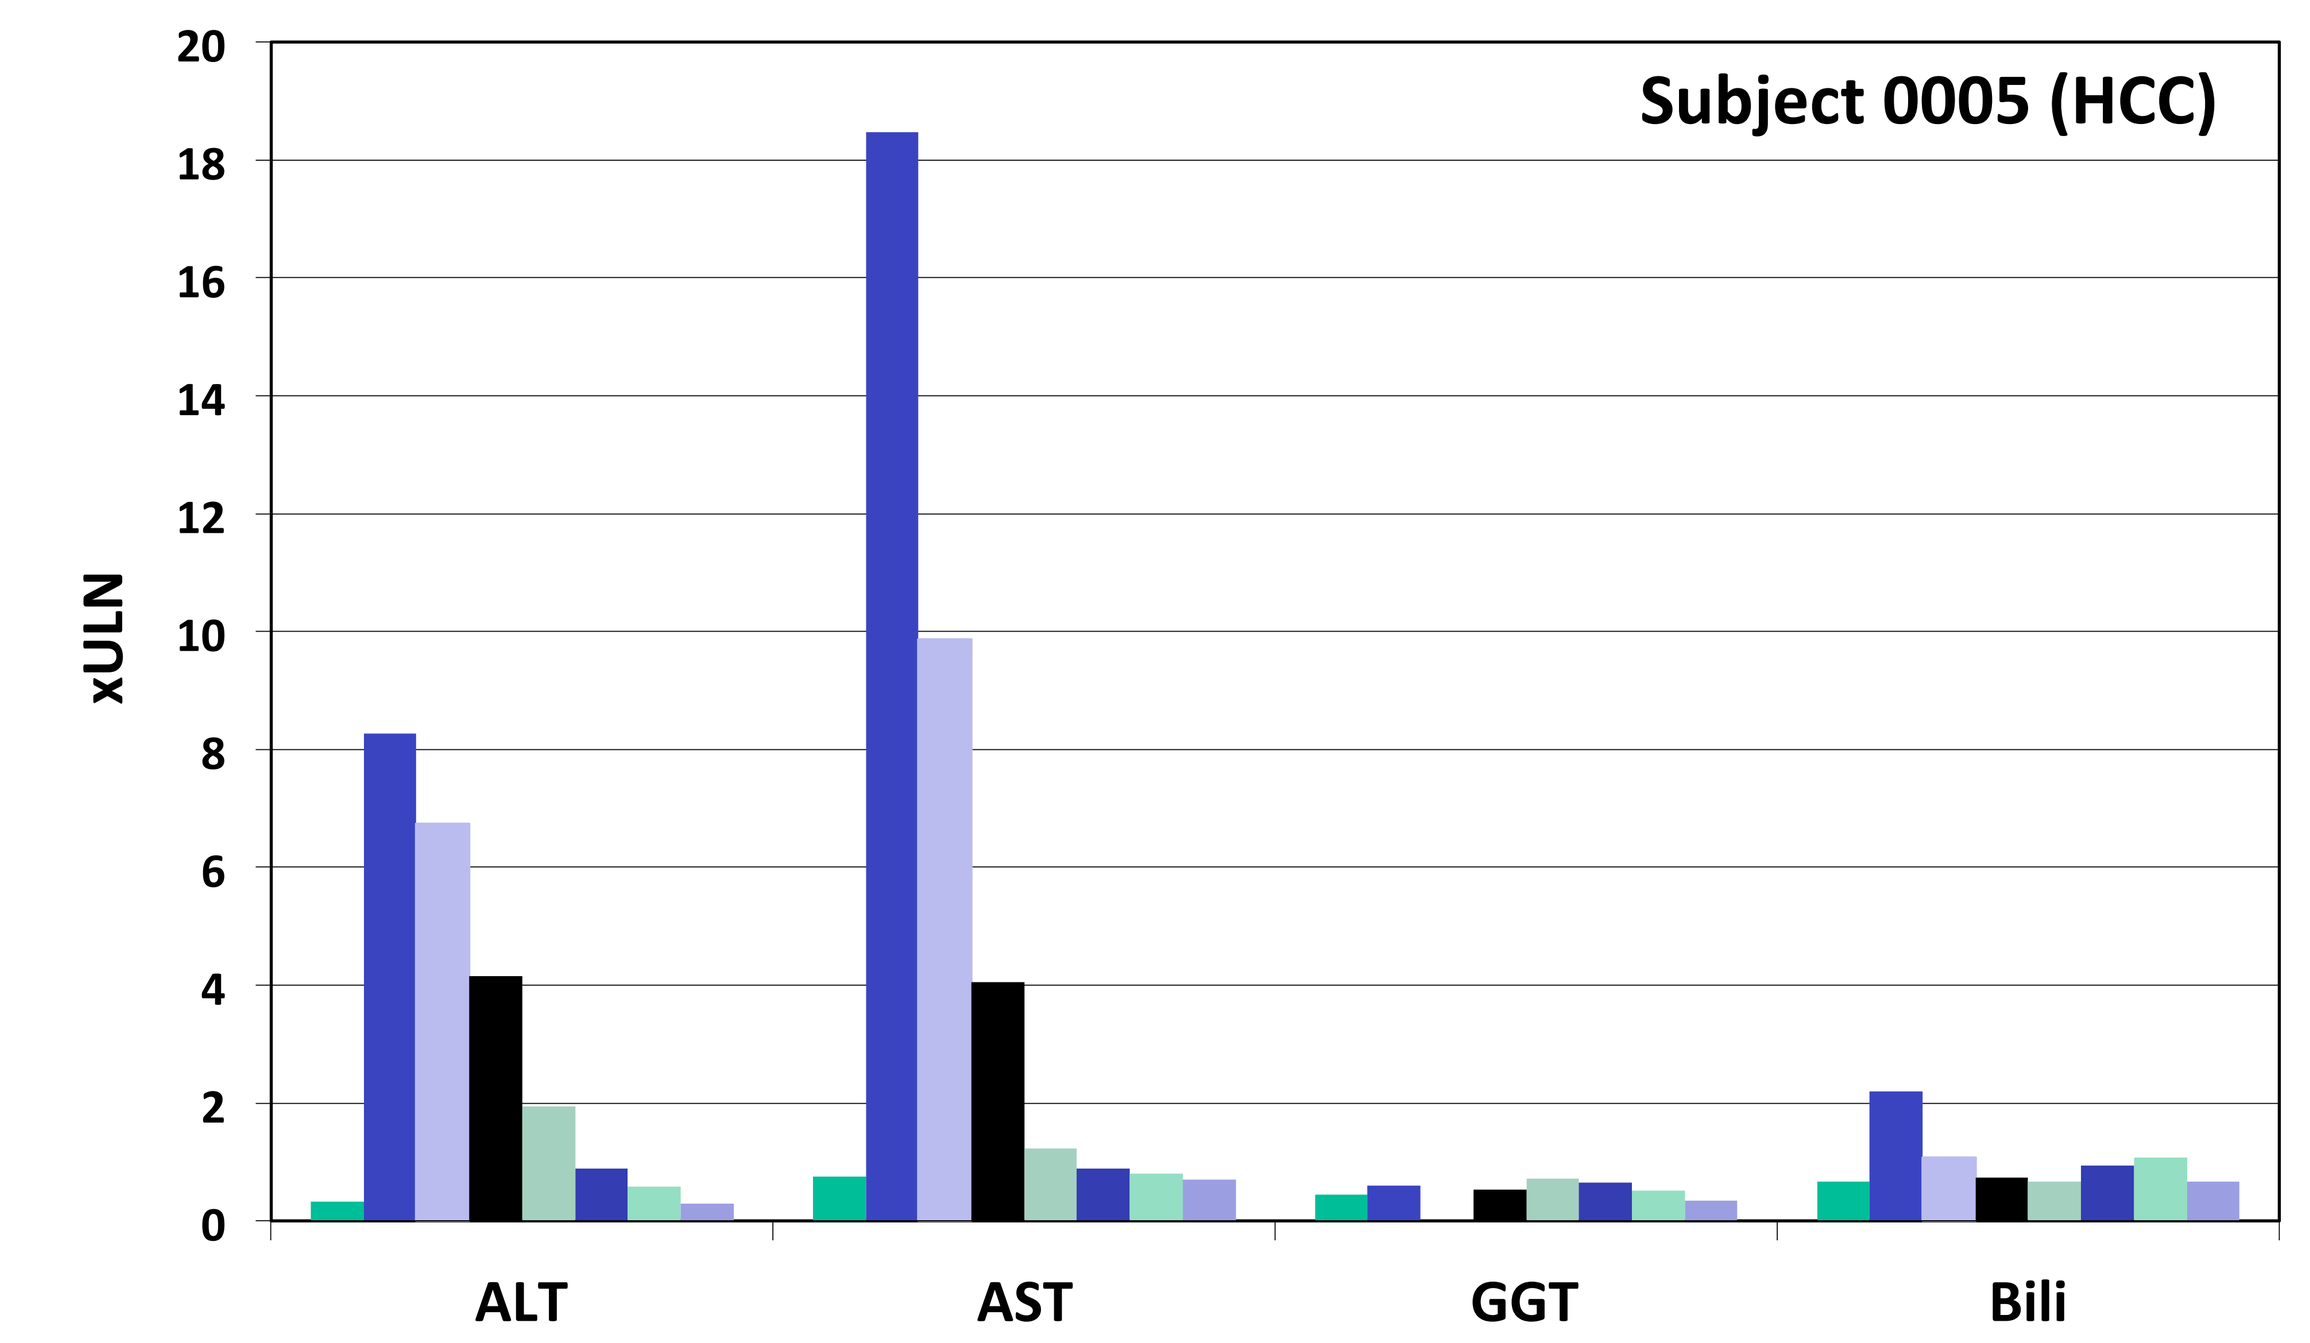
Subject 0005 (HCC)
20
18
16
14
12
10
8
6
4
2
0
xULN
ALT
AST
GGT
Bili

Summary and Conclusions

- Study Ongoing at Three Centers in the USA
 - Expansion Cohort 1 – Additional HCC and Liver Metastases
 - 24 Subjects, Single Treatment, Re-enroll for Multiple Lesions
 - Expansion Cohort 2 – HCC Patients on Sorafenib
 - PV-10 Dose Escalation (6 + 6 Subjects)
- Objective Response Observed in Injected Tumors
- Toxicity was Transient
- Upcoming: Asia/Pacific Phase 1b/2 Combination Study for HCC
 - SAT: SOC + PV-10
 - RCT: SOC ± PV-10